70

Inhibition of Fatty Acid Oxidation by the Peroxisome Proliferator Activated Receptor-α Antagonist TPST-1120 Elicits Tumor Regression Concomitant with Immune Activation

Nathan Standifer, Valerie Chen, Yonchu Jenkins, Mandy Cheng, David Freund, Henry Johnson, Dara Burdette, and Sam Whiting
Tempest Therapeutics, Brisbane, CA

OBJECTIVE

To assess TPST-1120 activities on tumor progression, FAO, and immune activation in preclinical models and clinical samples from patients with advanced solid tumors receiving TPST-1120 as monotherapy or in combination with immune checkpoint inhibitors

BACKGROUND

Peroxisome Proliferator Activated Receptor-α (PPARα)

- Nuclear hormone receptor that functions as master regulator of fatty acid oxidation (FAO), upregulated in multiple cancers, and expressed by immune cells (**Figure 1A**)[1]
- Represses multiple proinflammatory transcription factors (e.g., NF-κB, STAT-1, STAT3, and AP-1), thereby limiting anti-tumor immunity (**Figure 1B**)[1]
- Numerous tumors utilize FAO as a primary metabolic pathway, including renal cell carcinoma (RCC), cholangiocarcinoma (CCA), and hepatocellular carcinoma (HCC), making PPARα an attractive target for therapeutic intervention[2]

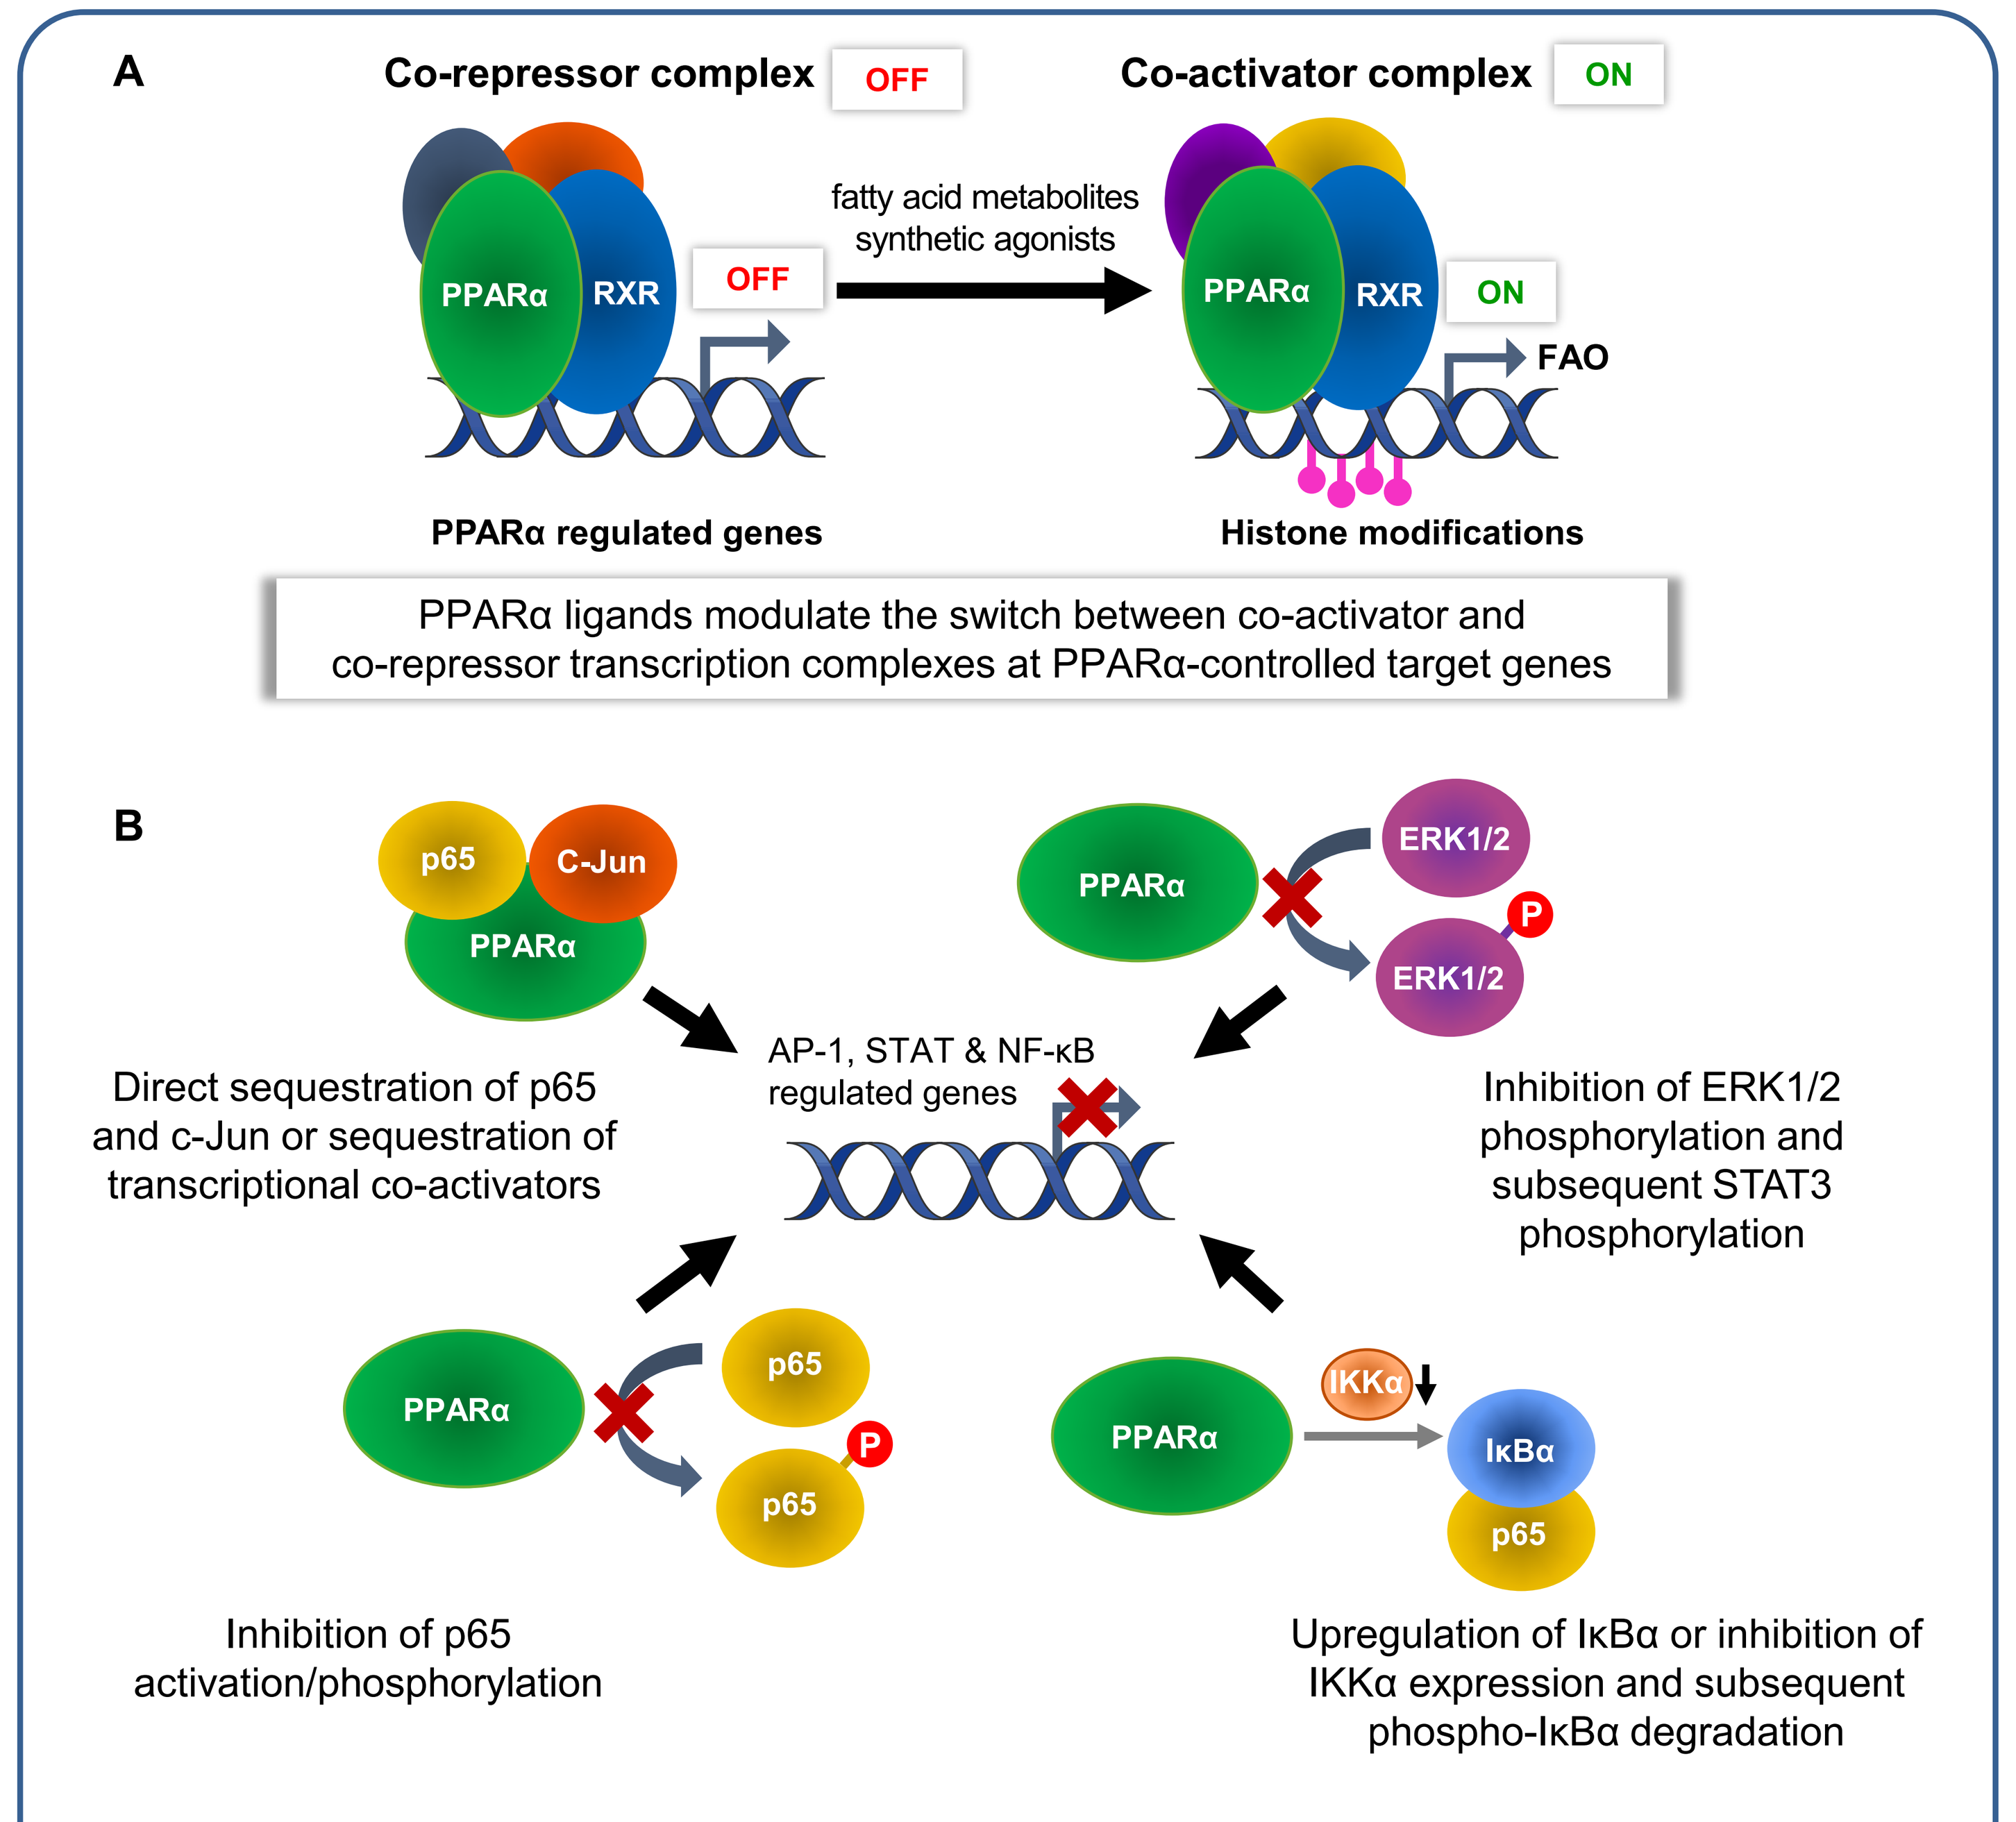
A
Co-repressor complex OFF
Co-activator complex ON
PPARα
RXR
OFF
fatty acid metabolites
synthetic agonists
PPARα
RXR
ON
FAO
PPARα regulated genes
Histone modifications
PPARα ligands modulate the switch between co-activator and co-repressor transcription complexes at PPARα-controlled target genes
B
p65
C-Jun
PPARα
PPARα
ERK1/2
P
ERK1/2
AP-1, STAT & NF-κB regulated genes
Direct sequestration of p65 and c-Jun or sequestration of transcriptional co-activators
Inhibition of ERK1/2 phosphorylation and subsequent STAT3 phosphorylation
PPARα
p65
P
p65
PPARα
IKKα
IκBα
p65
Inhibition of p65 activation/phosphorylation
Upregulation of IκBα or inhibition of IKKα expression and subsequent phospho-IκBα degradation

Figure 1. (A) PPARα-Regulated Transcription; (B) PPARα Transrepressive Activities

TPST-1120

- First-in-class, orally-bioavailable, potent, highly selective competitive antagonist of PPARα
- Hypothesized to block tumor-promoting FAO and alleviate PPARα-induced immune suppression
- Clinical experience:
 - ***Phase 1 trial in advanced solid tumors (NCT03829436):*** 53% disease control rate as single agent and 30% objective response rate (ORR) in combination with nivolumab at the two highest TPST-1120 doses tested[3]
 - ***Phase 1b/2 randomized trial in 1L metastatic HCC (NCT04524871):*** 30% confirmed ORR with TPST-1120 plus atezolizumab + bevacizumab vs. 13.3% with atezolizumab + bevacizumab alone; favorable PFS and OS hazard ratio in TPST-1120 arm vs. control arm[4]
 - Tolerable safety profile alone and in combination w/ nivolumab or atezolizumab + bevacizumab[3,4]

METHODS

X-Ray Crystallography

- The ligand binding domain (LBD) of human PPARα (amino acids 198-468) was expressed and purified from *E. coli*. The PPARα/TPST-1120 co-crystals were grown at 4°C by sitting drop vapor diffusion. Data collection was performed at Spring8 BL45XU beamline, and the structure was determined by molecular replacement with existing structure PDB ID: 1K7L

In Vitro PPARα and Gene Expression Assays

- TPST-1120 inhibitory activity against PPARα was determined in a CHO cell-based reporter cell line with a PPARα response element (PPRE) linked to a luciferase gene
- TPST-1120-mediated effects on gene expression and lipid content in THP-1 and U937 myeloid cell lines were assessed using qPCR and flow cytometry

In Vivo Experiments: Pancreatic, Colon, and Liver Tumor Models

- BALB/c or C57BL/6 mice bearing PancOH7 (pancreatic), MC38 (colon), or Hepa 1-6 (liver) murine tumors were treated with TPST-1120 as monotherapy or in combination with mouse anti-PD-1
- Tumors were allowed to establish for 8 days (average of 171 ± 5 mm³) prior to treatment with vehicle (n = 10), TPST-1120 at 30 mg/kg orally BID (n = 7), anti-PD1 200 µg IP Q3D (n = 10), or TPST-1120 + anti-PD-1 (n = 15). Animals were followed for tumor growth
- Animals in vehicle group (n = 10) were sacrificed on treatment Day 16 due to tumor growth (2024 ± 153 mm³)

TPST-1120-01 Phase 1 Clinical Study Design (NCT03829436)

Key Eligibility Criteria	TPST-1120 Dose Escalation	Endpoints / Translational Readouts
Inclusion • Advanced/metastatic solid tumor • ECOG PS 0-1 • Adequate organ function • No standard therapy available ***Exclusion*** • Immunosuppressive meds • Autoimmune disease • Fibrates within 28 days of enrollment	**Part 1:** TPST-1120 Monotherapy Dose Escalation **Solid Tumors** *3+3 Design* *TPST-1120 100 mg up to 600 mg BID* **Part 2:** TPST-1120 Combination with anti-PD-1 Dose Escalation **HCC, RCC, Cholangiocarcinoma** *3+3 Design* *TPST-1120 200 mg up to 600 mg BID* *Full dose nivolumab (480 mg Q4W)*	**Endpoints** • Safety • MTD and/or OBD of TPST-1120 • Pharmacokinetics • Preliminary efficacy **Translational Readouts** • Baseline tumor mutational status • Whole blood gene expression • Lipidomics

BID, twice daily; ECOG PS, Eastern Cooperative Oncology Group performance status; HCC, hepatocellular carcinoma; MTD, maximum tolerated dose; OBD, optimal biological dose; RCC, renal cell carcinoma; Q4W, once every 4 weeks.

Clinical Biomarker Assays

- Changes in whole blood gene expression and plasma lipid levels in patients enrolled in a phase 1 clinical trial (NCT03829436) were assessed using the Nanostring nCounter® PanCancer Immune Profiling panel and liquid chromatography-tandem mass spectrometry, respectively

RESULTS

TPST-1120 is a Competitive Antagonist of PPARα

- X-ray co-crystal structures demonstrated that TPST-1120 binds to the PPARα LBD, inducing a repressive conformation (**Figure 2A**)
- In an *in vitro* cell-based PPARα assay, TPST-1120 potently competed against the PPARα-specific synthetic agonist GW7647 and endogenous agonist oleoylethanolamide (OEA) (**Figure 2B, 2C**)

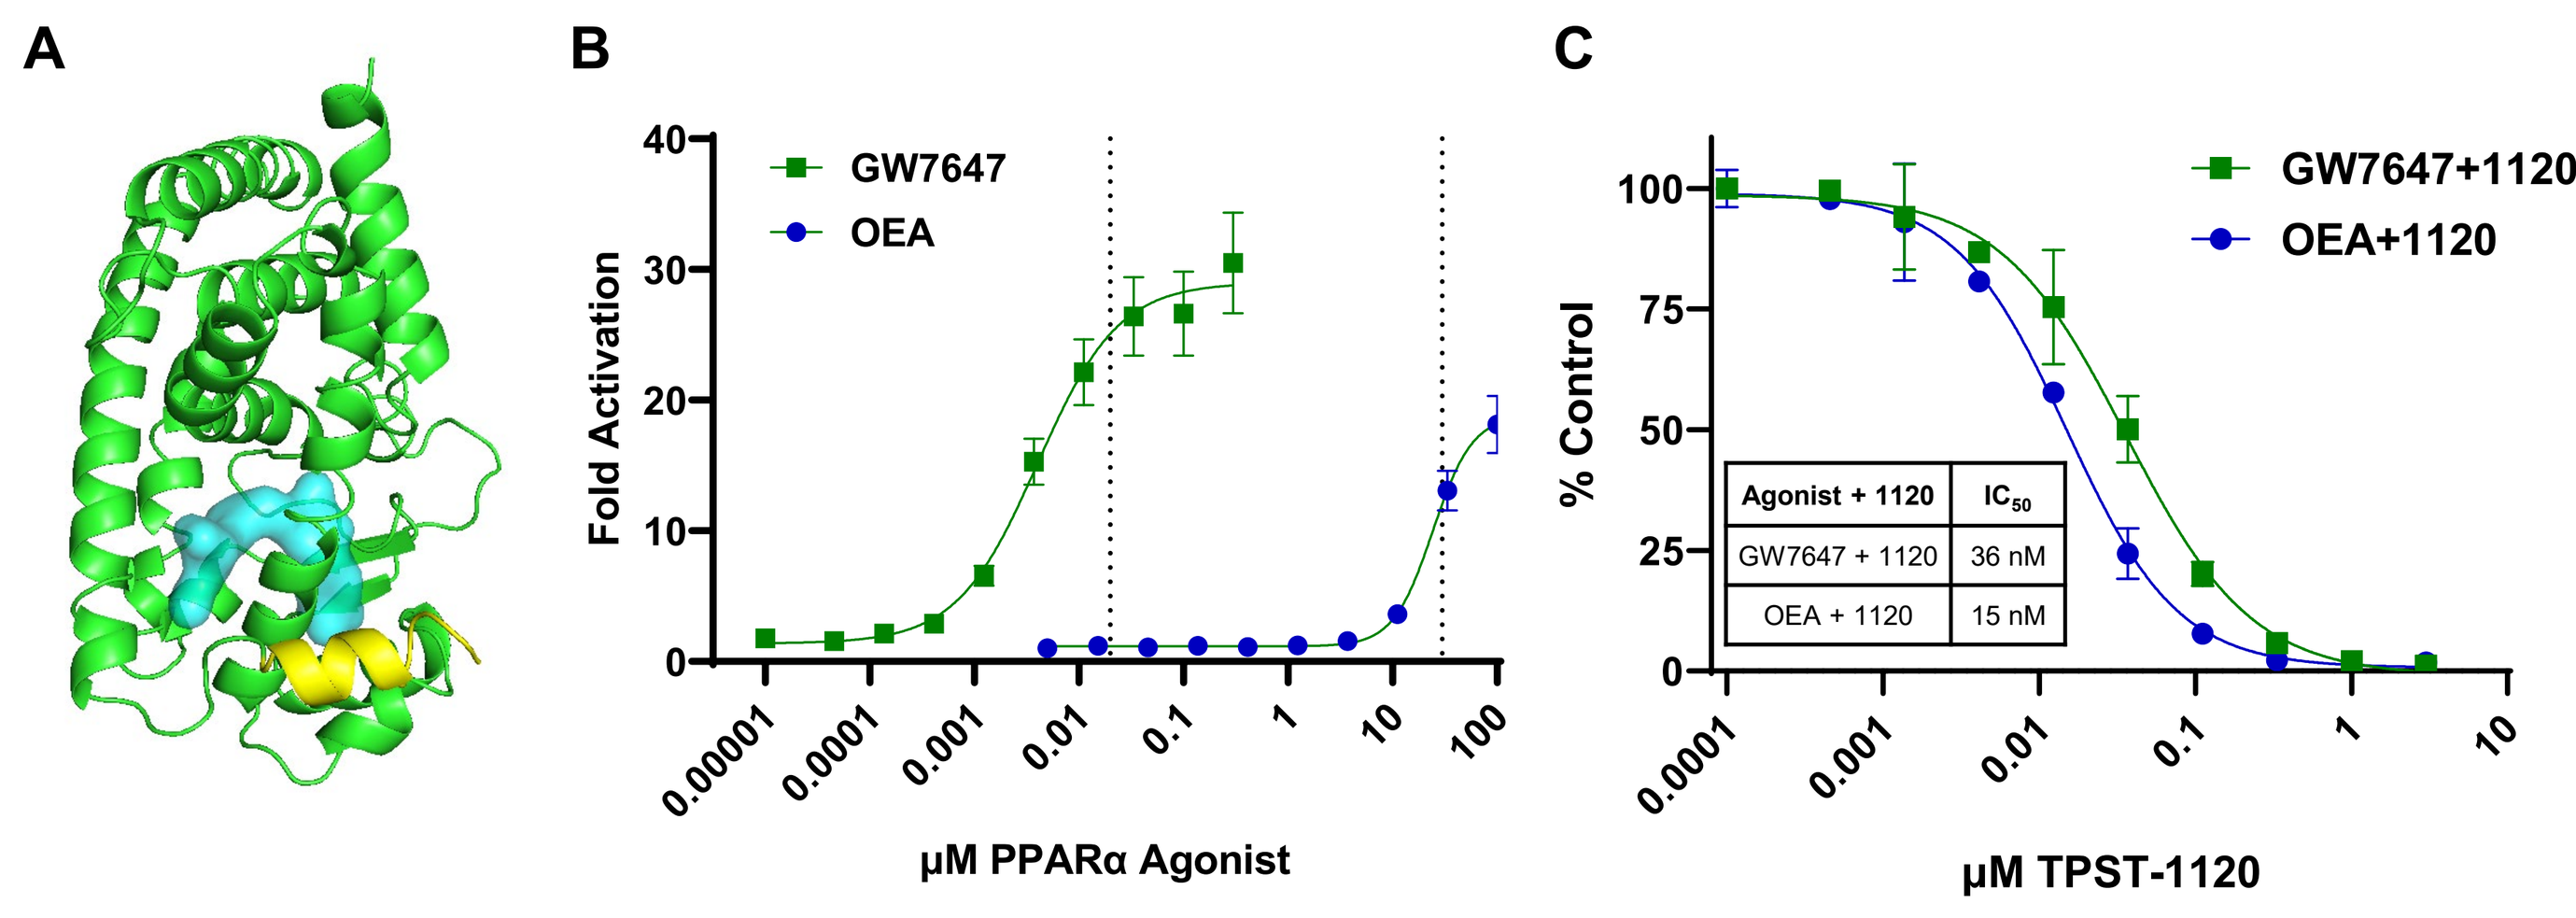
A
B
GW7647
OEA
Fold Activation
µM PPARα Agonist
C
GW7647+1120
OEA+1120
% Control
Agonist + 1120 | IC50
GW7647 + 1120 | 36 nM
OEA + 1120 | 15 nM
µM TPST-1120

Figure 2. TPST-1120 is a Competitive Antagonist of PPARα. (A) The TPST-1120 antagonist-bound ligand binding domain (green) shows that TPST-1120 (blue) situates the AF-2 activation helix (yellow) in an inactive conformation. The inactive conformation has a lower affinity to co-activator motifs and disfavors activation of PPARα regulated genes. (B) Fold-activation of CHO cell-based reporter cell line following treatment with PPARα-specific synthetic agonist GW7647 and endogenous agonist OEA. Vertical dotted lines correspond to agonist concentration tested in panel C. (C) Decrease in PPARα-dependent activation by increasing concentrations of TPST-1120.

RESULTS (cont.)

TPST-1120 Inhibits Tumor Growth in Pancreatic, Colon, and Liver Tumor Models

- TPST-1120 elicited > 50% tumor growth inhibition in PancOH7, MC38, and Hepa1-6 models with enhanced inhibition observed in the latter two models when co-administered with anti-PD-1 (**Figure 3**)

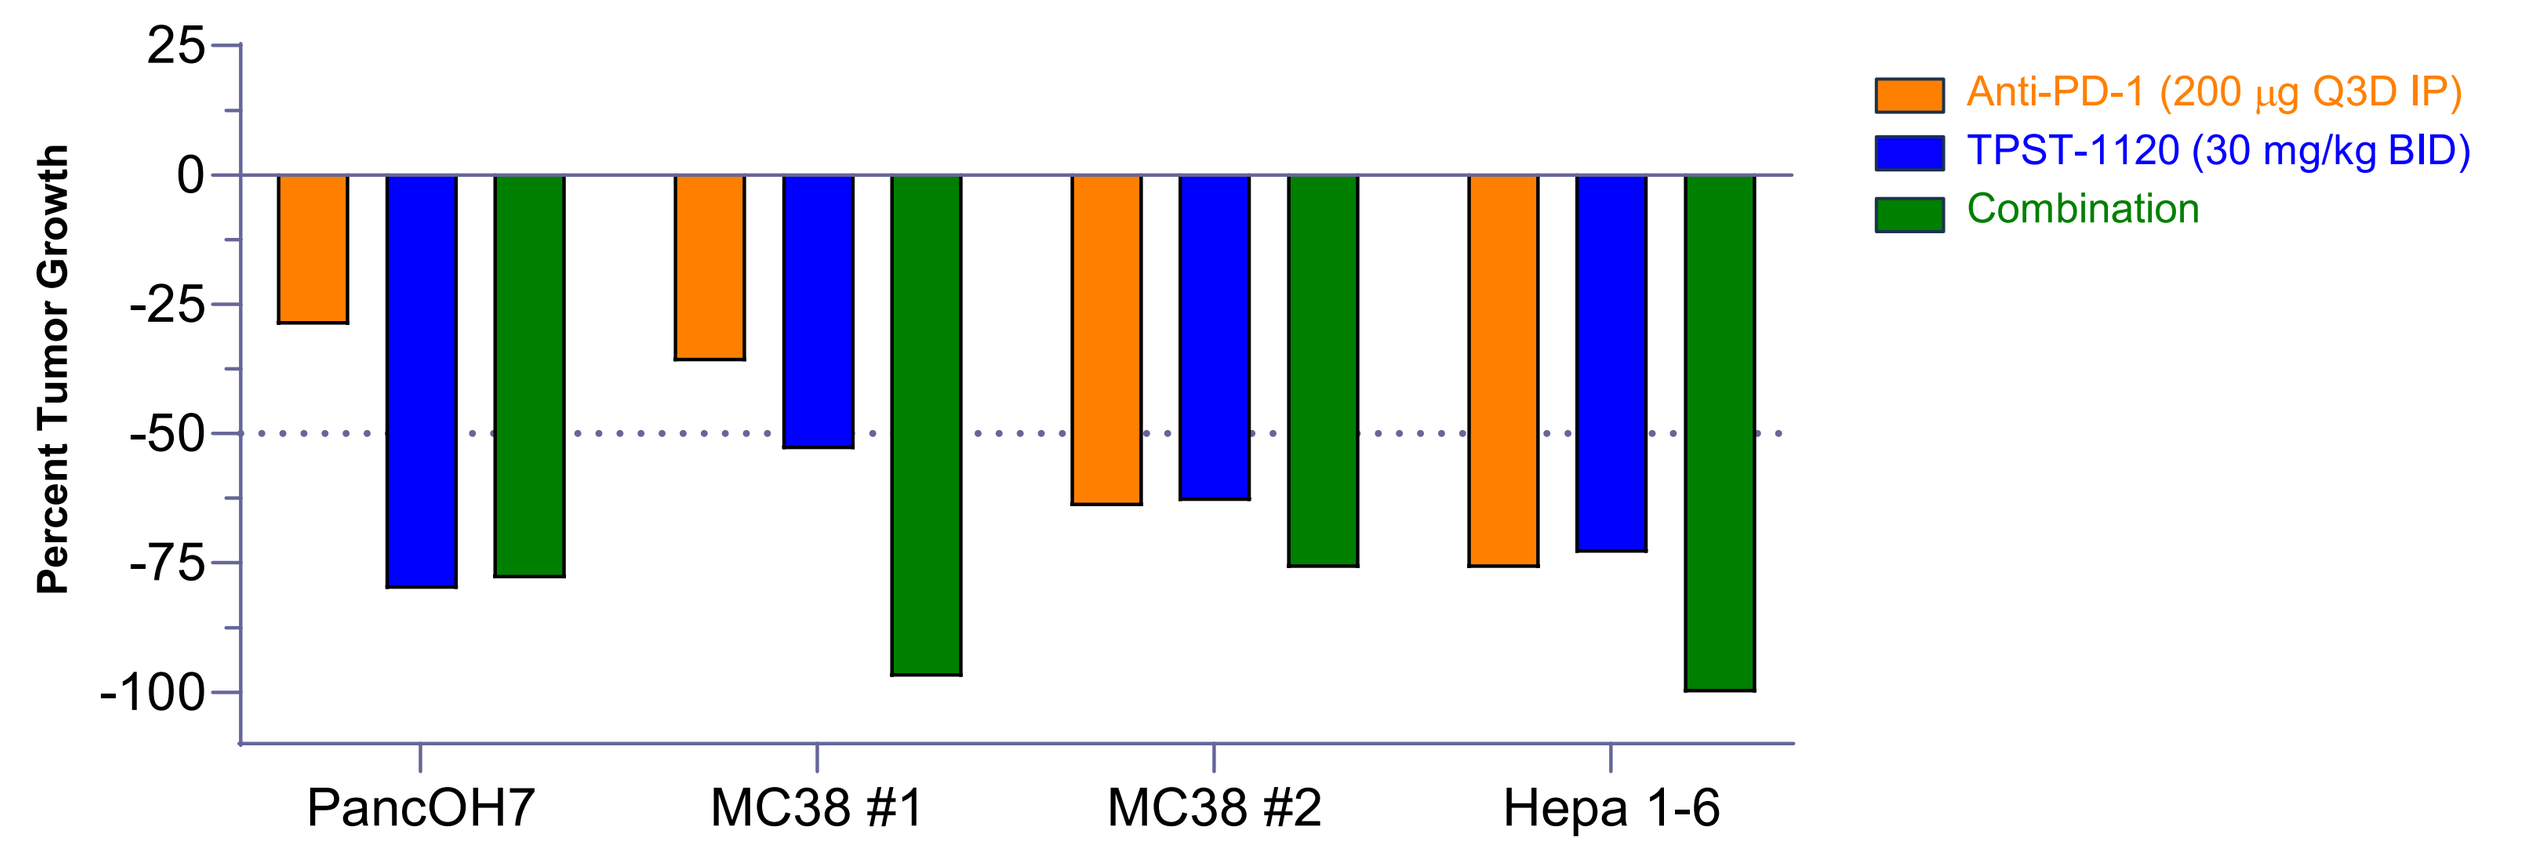
Percent Tumor Growth
PancOH7
MC38 #1
MC38 #2
Hepa 1-6
Anti-PD-1 (200 µg Q3D IP)
TPST-1120 (30 mg/kg BID)
Combination

Figure 3. Percent Tumor Growth Inhibition with TPST-1120. The mean change in tumor volume from treatment initiation was calculated at end of study (approximately day 21) for each tumor model treated with indicated therapy. Results from two independent experiments with MC38 shown.

Mice Exhibiting Complete MC38 Tumor Regression are Protected from Tumor Re-challenge

- At treatment Day 16, treated MC38 tumors were significantly smaller compared to control group (TPST-1120 $P = 0.002$; anti-PD-1 $P = 0.002$; Combo $P < 0.00001$) (**Figure 4A**)
- Three animals in the combination group exhibited complete tumor regression (considered cured) which was maintained at Day 60; when cured mice were rechallenged with fresh autologous tumors, they remained protected from tumor growth while tumors grew as expected in the naïve, age-matched control mice (**Figure 4B**)

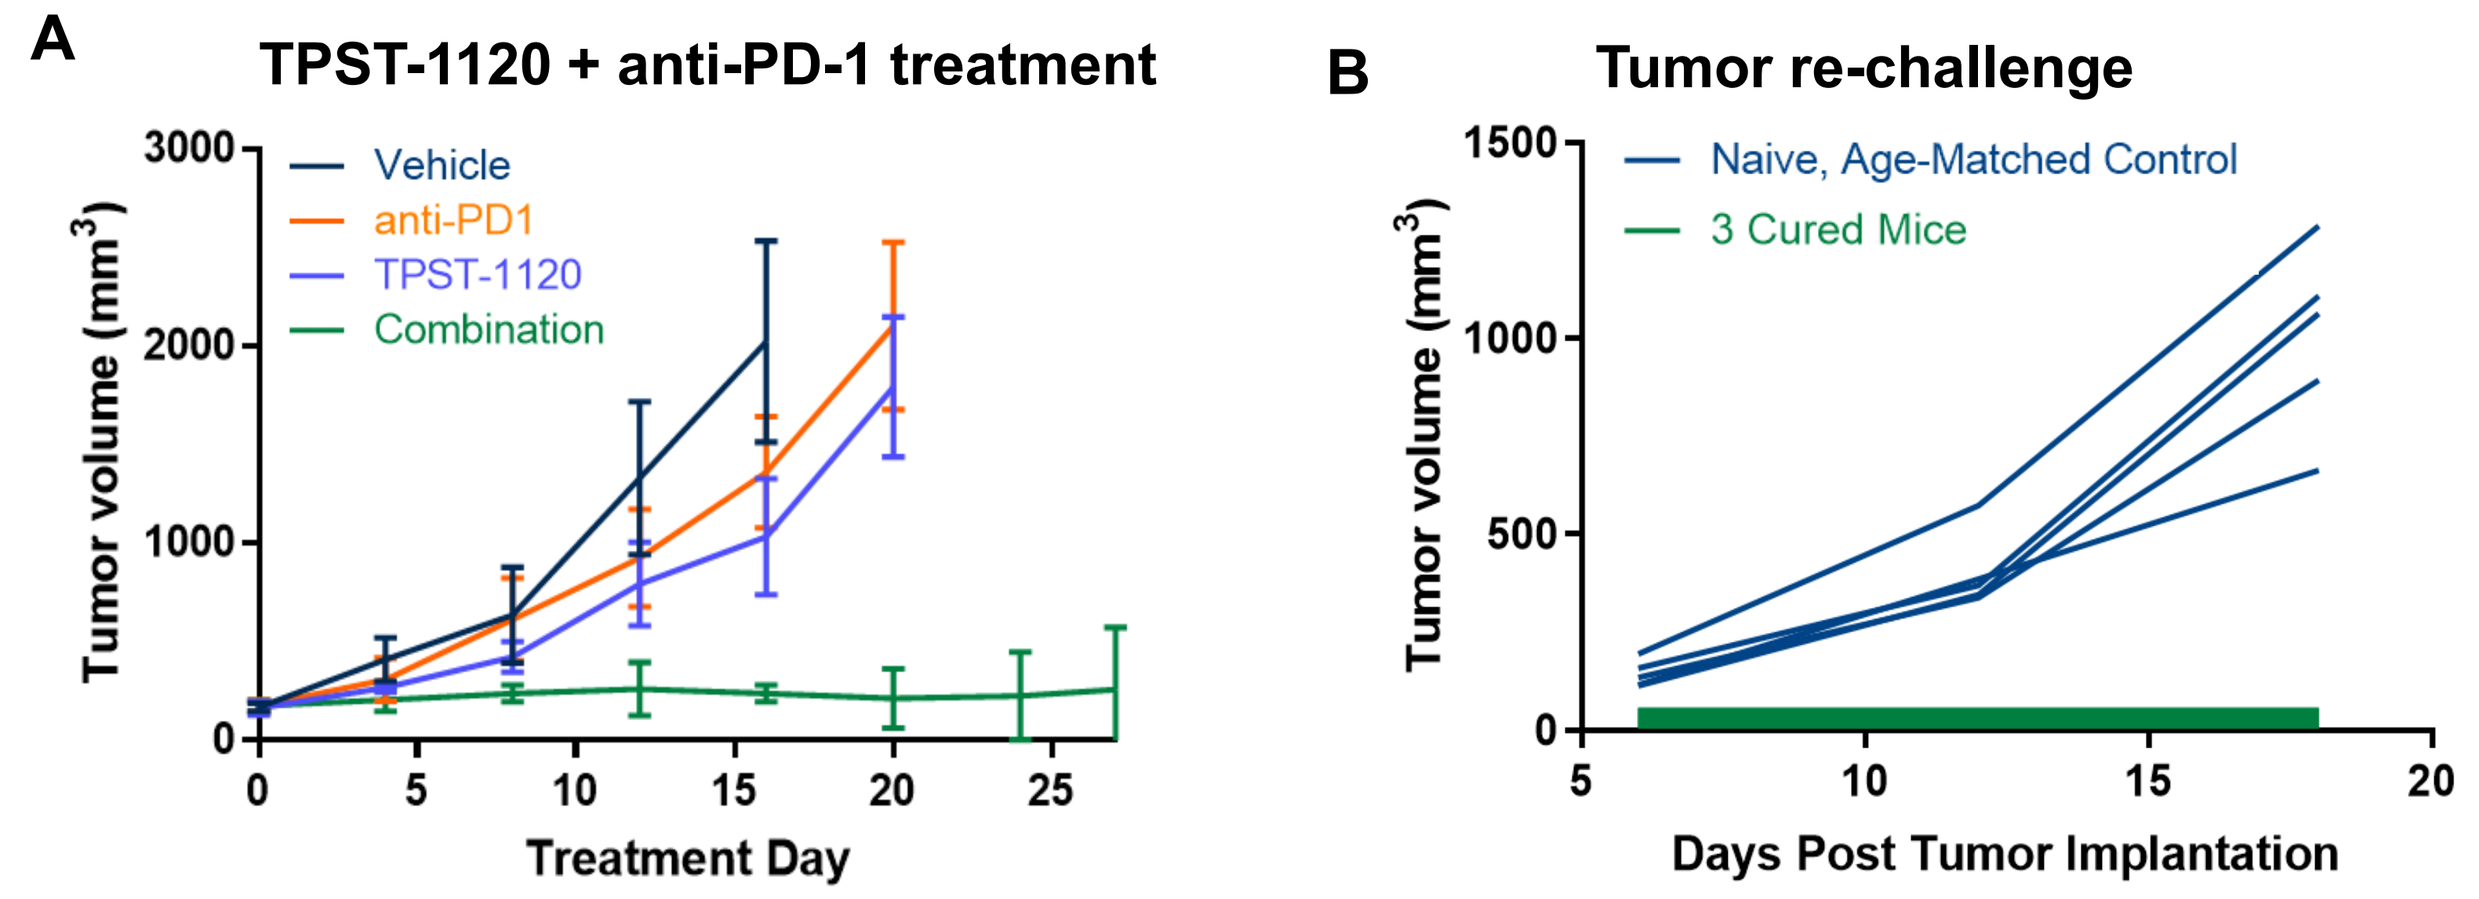
A
TPST-1120 + anti-PD-1 treatment
Vehicle
anti-PD1
TPST-1120
Combination
Tumor volume (mm³)
Treatment Day
B
Tumor re-challenge
Naive, Age-Matched Control
3 Cured Mice
Tumor volume (mm³)
Days Post Tumor Implantation

Figure 4. TPST-1120 + Anti-PD-1 Elicits Tumor Regression and Long-Standing Immune Memory in MC38 Colorectal Cancer Tumor Model in C57BL/6 Immunocompetent Mice. (A) C57BL/6 mice bearing 150 mm³ MC38 flank tumors treated with TPST-1120 30 mg/kg BID, 200 µg α-PD-1 Q3D or combination. (B) Three mice from combination therapy group with no observable tumors at day 60 or 5 age-matched control received MC38 tumors in flank and were monitored for 18 days.

TPST-1120 Induces Increased Neutral Lipid Content and Elevated Immune-Related Markers

- The human myelomonocytic cell line THP-1 treated with TPST-1120 in the presence of oleic acid demonstrated changes in lipid content (**Figure 5A**), immune gene expression (**Figure 5B**) and corresponding protein levels (**Figure 5C**) as a function of dose.

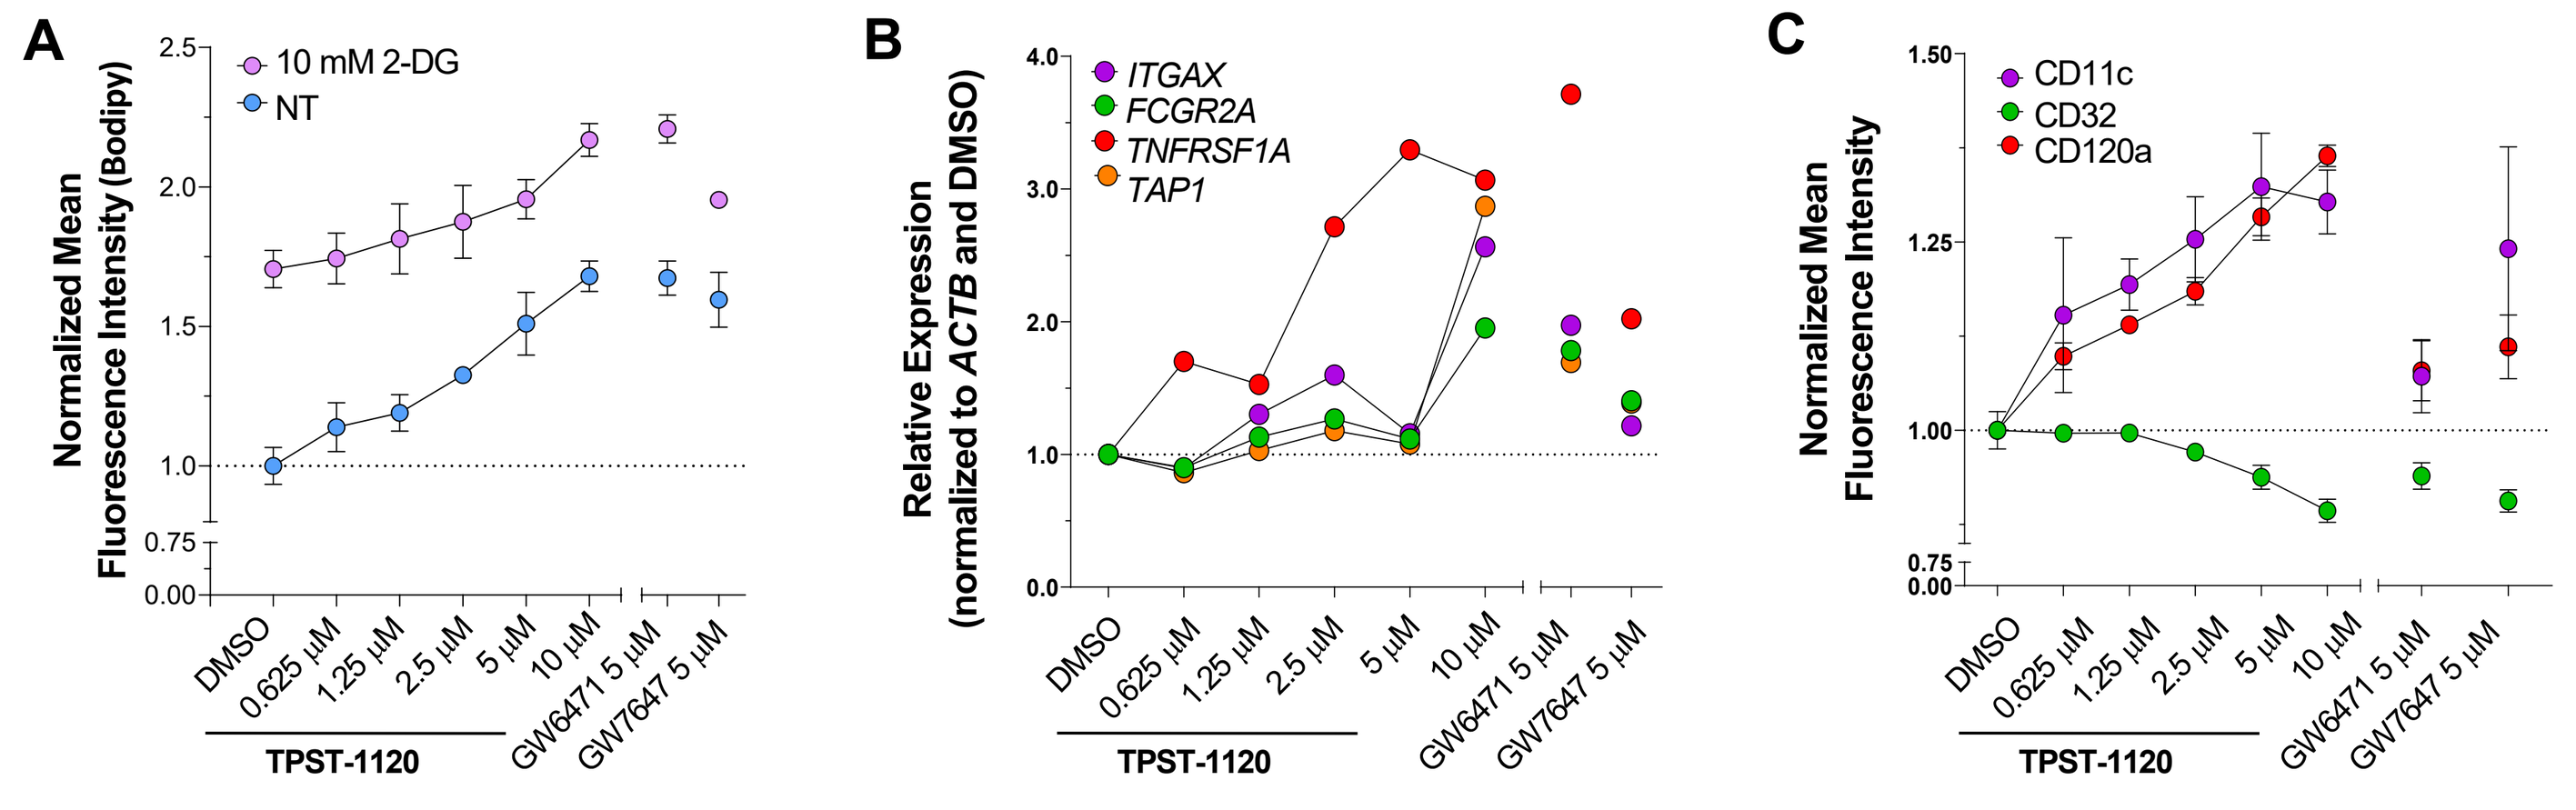
A
10 mM 2-DG
NT
Normalized Mean Fluorescence Intensity (Bodipy)
DMSO
0.625 µM
1.25 µM
2.5 µM
5 µM
10 µM
GW6471 5 µM
GW7647 5 µM
TPST-1120
B
ITGAX
FCGR2A
TNFRSF1A
TAP1
Relative Expression (normalized to ACTB and DMSO)
C
CD11c
CD32
CD120a
Normalized Mean Fluorescence Intensity

Figure 5. TPST-1120 Induces Increased Neutral Lipid Content and Elevated Immune-Related Genes in THP-1 Cells. (A) THP-1 cells were incubated for 24 hr in the presence of oleic acid (30 µM) with or without 2-deoxyglucose (2-DG) in increasing concentrations of TPST-1120 after which Bodipy staining was analyzed. Similar results observed with U937 cells (data not shown); (B) Expression levels of listed genes in THP-1 cells incubated with increasing TPST-1120; (C) Cell surface levels of listed proteins on THP-1 cells in increasing TPST-1120. 2-DG: glucose analog that inhibits glycolysis; GW6471: PPARα-selective antagonist; GW7647: PPARα-selective agonist.

Biomarker Analysis of Gene Expression and FFA Supports TPST-1120 Mechanism of Action

- In the phase 1 trial, statistically significant, exposure-dependent elevations in expression levels of multiple immune-related genes including *FCGR2A* (CD32), *ITGAX* (CD11c), *TAP1,* and *TNFRSF1A* (CD120a) were observed, and patients exhibiting objective responses displayed increased free fatty acids (FFA) in plasma (**Figure 6**)

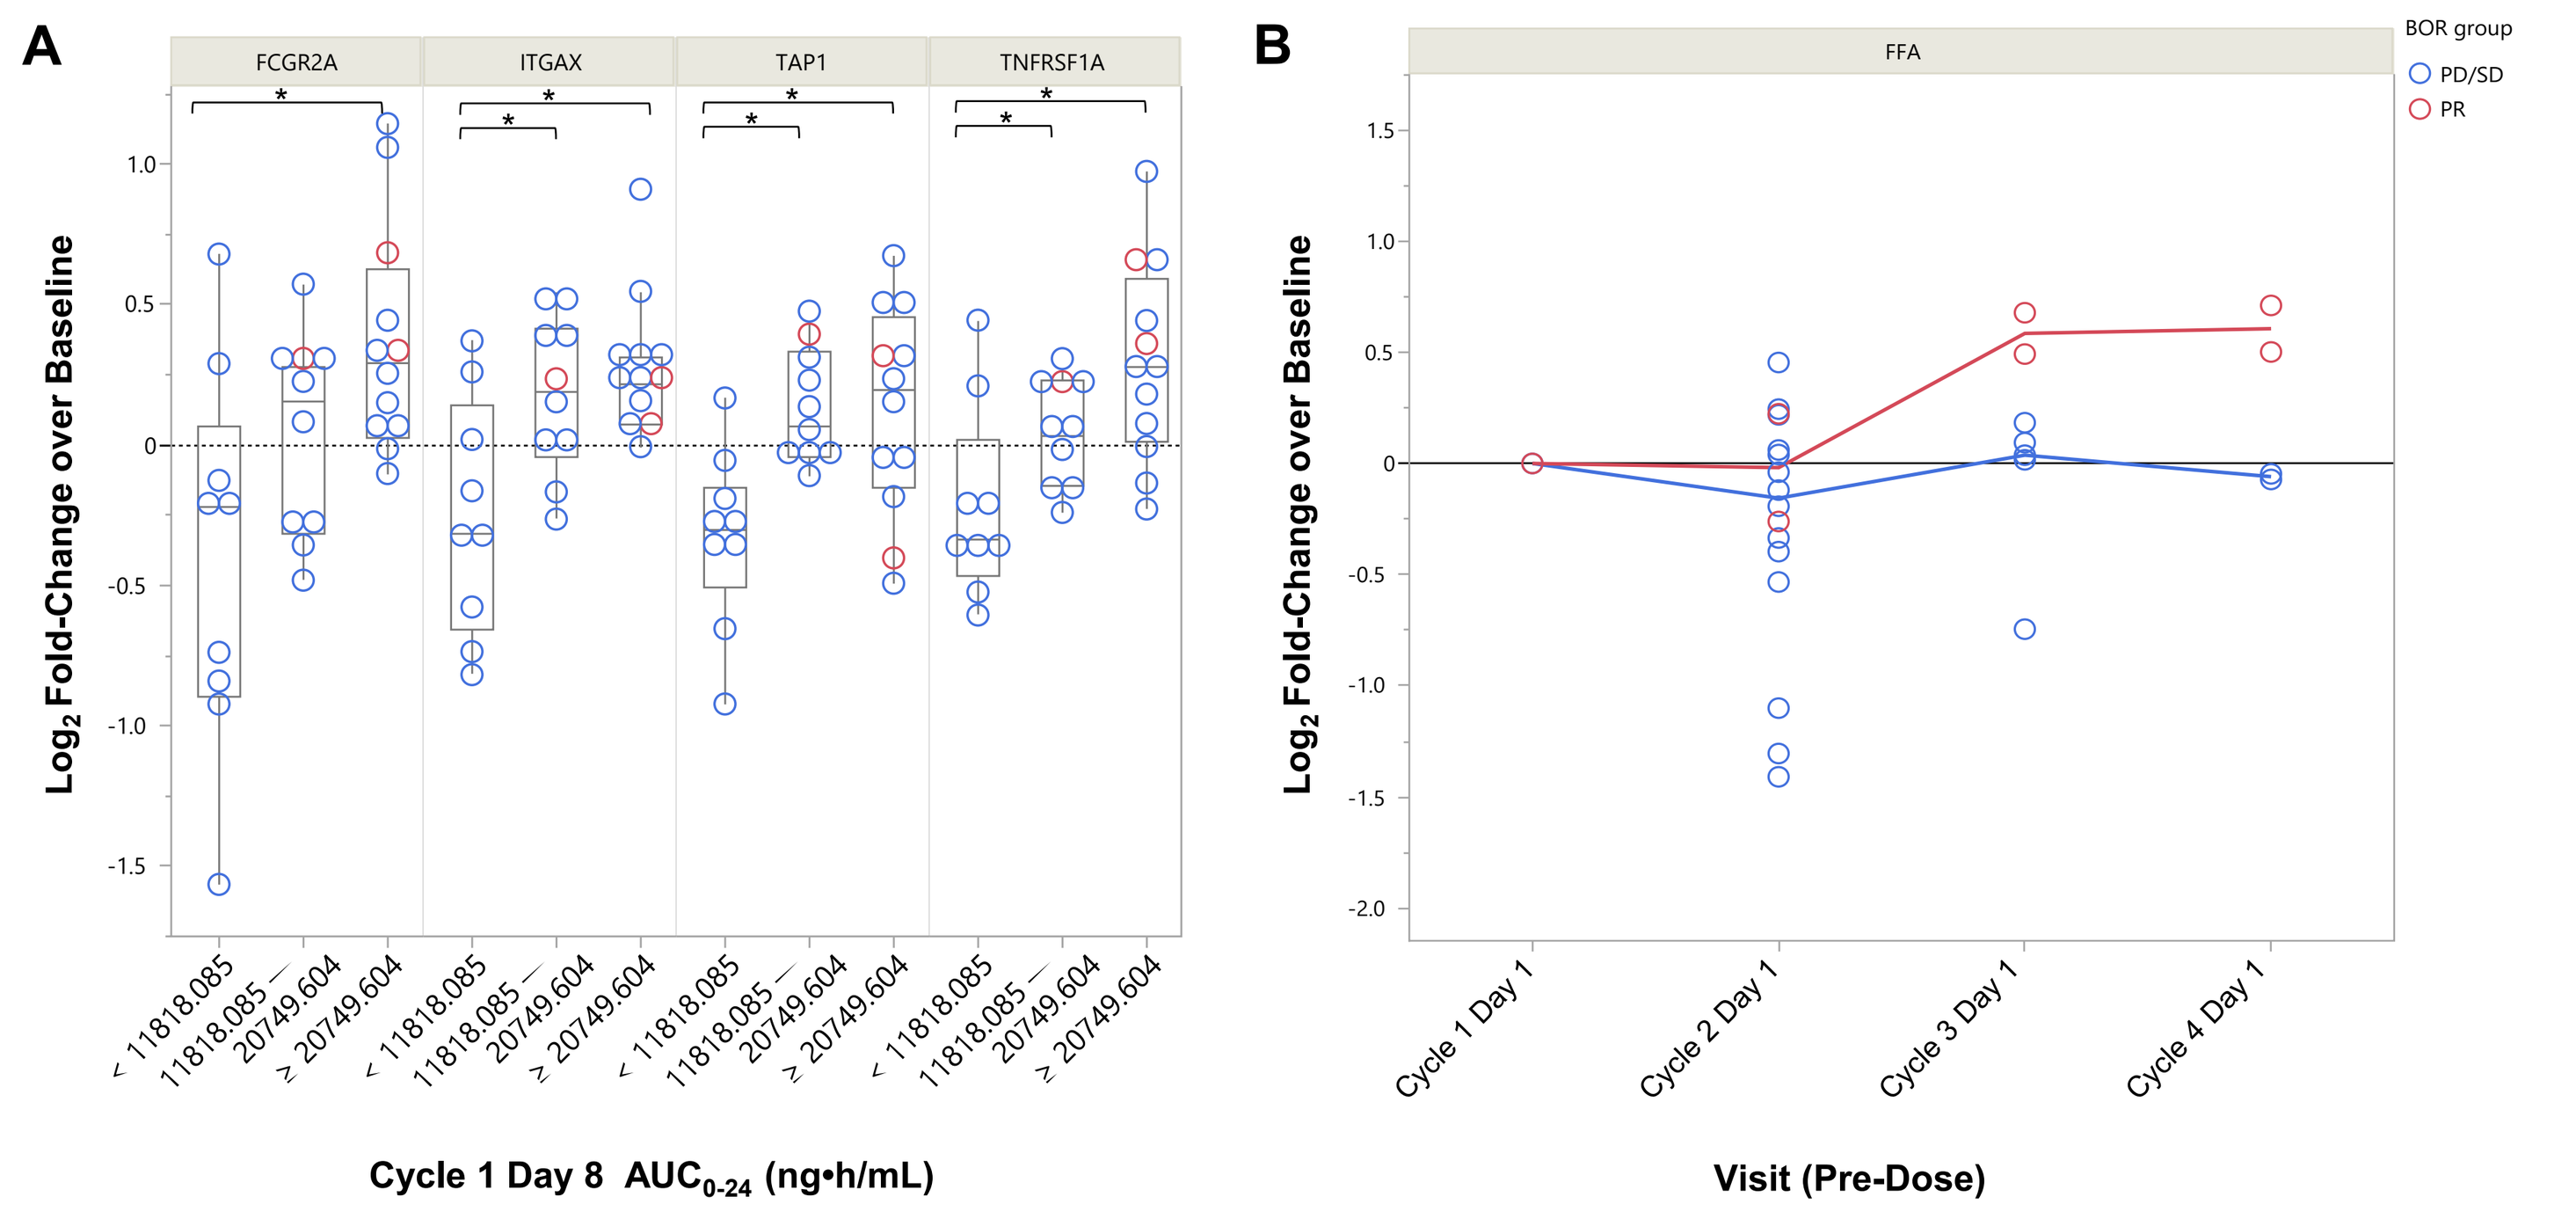
A
FCGR2A
ITGAX
TAP1
TNFRSF1A
Log2 Fold-Change over Baseline
< 11818.085
11818.085 – 20749.604
≥ 20749.604
Cycle 1 Day 8 AUC0-24 (ng•h/mL)
B
FFA
BOR group
PD/SD
PR
Cycle 1 Day 1
Cycle 2 Day 1
Cycle 3 Day 1
Cycle 4 Day 1
Visit (Pre-Dose)

Figure 6. TPST-1120 Induces Increased Expression of Immune-Related Genes and Elevated Free Fatty Acids. (A) Genes differentially expressed on treatment day 8 vs. baseline, as a function of TPST-1120 exposure. Median elevation magnitudes in highest tertile were statistically increased above baseline values (*$P < 0.05$ by Wilcoxon's pair-wise analysis vs. first tertile [<11818.05]). (B) Log_2-fold-changes in baseline normalized free fatty acids (FFA) in patients enrolled in Part 2, presented by response.

CONCLUSIONS

- TPST-1120 is a potent competitive antagonist of PPARα that inhibits FAO and induces immune activation consistent with its proposed mechanism of action:
 - Co-crystal structure of TPST-1120 and the PPARα ligand-binding domain show a repressive conformation suggesting reduced transcription of target genes
 - TPST-1120 potently competed against PPARα-specific agonists in a cell-based assay, demonstrating specificity for the PPARα isoform
 - Tumor growth was inhibited > 50% in pancreatic, colon, and liver tumor models in mice, with immune memory induction in the colon cancer model
 - Dose-dependent increases in lipid content and immune gene expression support PPARα inhibition by TPST-1120
 - In a phase 1 study, patients with advanced solid tumors receiving TPST-1120 showed significant exposure-dependent elevation of several immune-related genes and increased plasma FFA associated with clinical response
- Clinical response and biomarker findings demonstrate that inhibition of PPARα is an effective therapeutic strategy for the treatment of cancer and has been a basis for the ongoing late-stage clinical development of TPST-1120

REFERENCES: 1. Bougarne N, et al. *Endocr Rev.* 2018;39:760–802. **2.** Nath A and Chan C. *Sci Rep.* 2016;6:18669 2; **3.** Yarchoan M, et al. *J Clin Oncol.* 2022; 40(16): Abstract 3005; **4.** Tempest Therapeutics Press Release. October 11, 2023. https://ir.tempesttx.com/news-releases/news-release-details/tempest-releases-new-data-demonstrating-superiority-tpst-1120.

ACKNOWLEDGMENTS: We thank the patients and their families, investigators, and coordinators for participation in the clinical trial, and Mark Yarchoan MD and Thomas W. Dubensky PhD for their expert insight in the conduct and interpretation of this study. Ingrid Koo, PhD, provided editorial support for the poster.

Society for Immunotherapy of Cancer (SITC) 2024 Spring Scientific Meeting, March 10-12, 2024, Miami, FL [This presentation is the intellectual property of the author/presenter. Contact nstandifer@tempesttx.com for permission to reprint and/or distribute]