Dual Blockade of the EP2 and EP4 PGE2 Receptors with TPST-1495 is an Optimal Approach for Drugging the Prostaglandin Pathway

Brian Francica[1], Justine Lopez[1], Franciele Kipper[2], Dingzhi Wang[3], Dave Freund[1], Anja Holtz[1], Chan Whiting[1], Dipak Panigrahy[2], Raymond Dubois[3], Sam Whiting[1], Thomas W. Dubensky[1].

[1]Tempest Therapeutics, 7000 Shoreline Ct, South San Francisco; [2]Beth Israel Deaconess Medical Center, 330 Brookline Ave, Boston, MA 02215; [3]Medical University of South Carolina, 171 Ashley Ave, Charleston, SC 29425

ABSTRACT

Prostaglandin E2 (PGE2) is a bioactive lipid produced by tumor cells that drives disease progression through stimulating tumor proliferation, enhancing angiogenesis and suppressing immune function in the tumor microenvironment (TME)[1, 2, 3.] PGE2 is also a mediator of adaptive resistance to immune checkpoint inhibitor therapy via the upregulation of cyclooxygenase-2 (COX-2). While the role of PGE2 signaling in cancer is clear, how best to inhibit PGE2 for cancer treatment remains under investigation. Inhibition of COX-1 and/or COX-2 has shown promising results in observational studies and meta-analyses, but inconsistent results in prospective studies. PGE2 signals through four receptors, EP1-4, that are variably expressed on tumor and immune cells and have distinct biological activities. The EP2 and EP4 receptors signal through cAMP and drive pro-tumor activities, while the EP1 and EP3 receptors signal through calcium flux and IP3 and drive immune activation and inflammation. While COX-2 and single EP antagonists continue to be developed, the nature of PGE2 signaling supports our rationale to inhibit PGE2 by dual antagonism of the pro-tumor EP2/EP4 receptors, while sparing the pro-immune EP1/EP3 receptors. To our knowledge, TPST-1495 is the first clinical-stage dual inhibitor of both the EP2 and EP4 receptors and is distinguished from other methods of PGE2 inhibition being tested in patients.

In mouse and human whole blood assays, dual blockade of EP2 and EP4 receptors with TPST-1495 reversed PGE2-mediated suppression of LPS-induced TNF-α, while single EP4 receptor antagonists were unable to block suppression at higher PGE2 concentrations. Similarly, in murine and human T cells in vitro, TPST-1495 inhibited PGE2-mediated suppression, resulting in a significant increase of IFN-γ production in response to stimulation with cognate peptide antigen. In vivo, TPST-1495 therapy alone also significantly reduced tumor outgrowth in CT26 tumor-bearing mice, correlated with increased tumor infiltration by NK cells, $CD8^+$ T cells, AH1-specific $CD8^+$ T cells, and other anti-tumor myeloid and adaptive immune cell populations. The relative role of increased immune infiltration may be simultaneously dependent on both the tumor model immunogenicity and direct anti-tumor effect of TPST-1495, because we also observed significant tumor regression in NSG and $RAG2^{-/-}$ animals, which are both deficient in immune cell development. TPST-1495 monotherapy demonstrated a decrease of both the intestinal tumor size and number in Adenomatous Polyposis ($APC^{min/+}$) mice, as compared to a single EP4 antagonist. TPST-1495 is currently being evaluated in an ongoing Phase 1 first-in-human study (NCT04344795) to characterize PK, PD, safety, and to identify a recommended phase 2 dose for expansion cohorts in key indications and biomarker-selected patients.

INTRODUCTION

TPST-1495 is a First-in-Class[1] Dual EP2/EP4 PGE2 Receptor Antagonist

Rationally designed, based on current understanding of PGE2 signaling in cancer progression

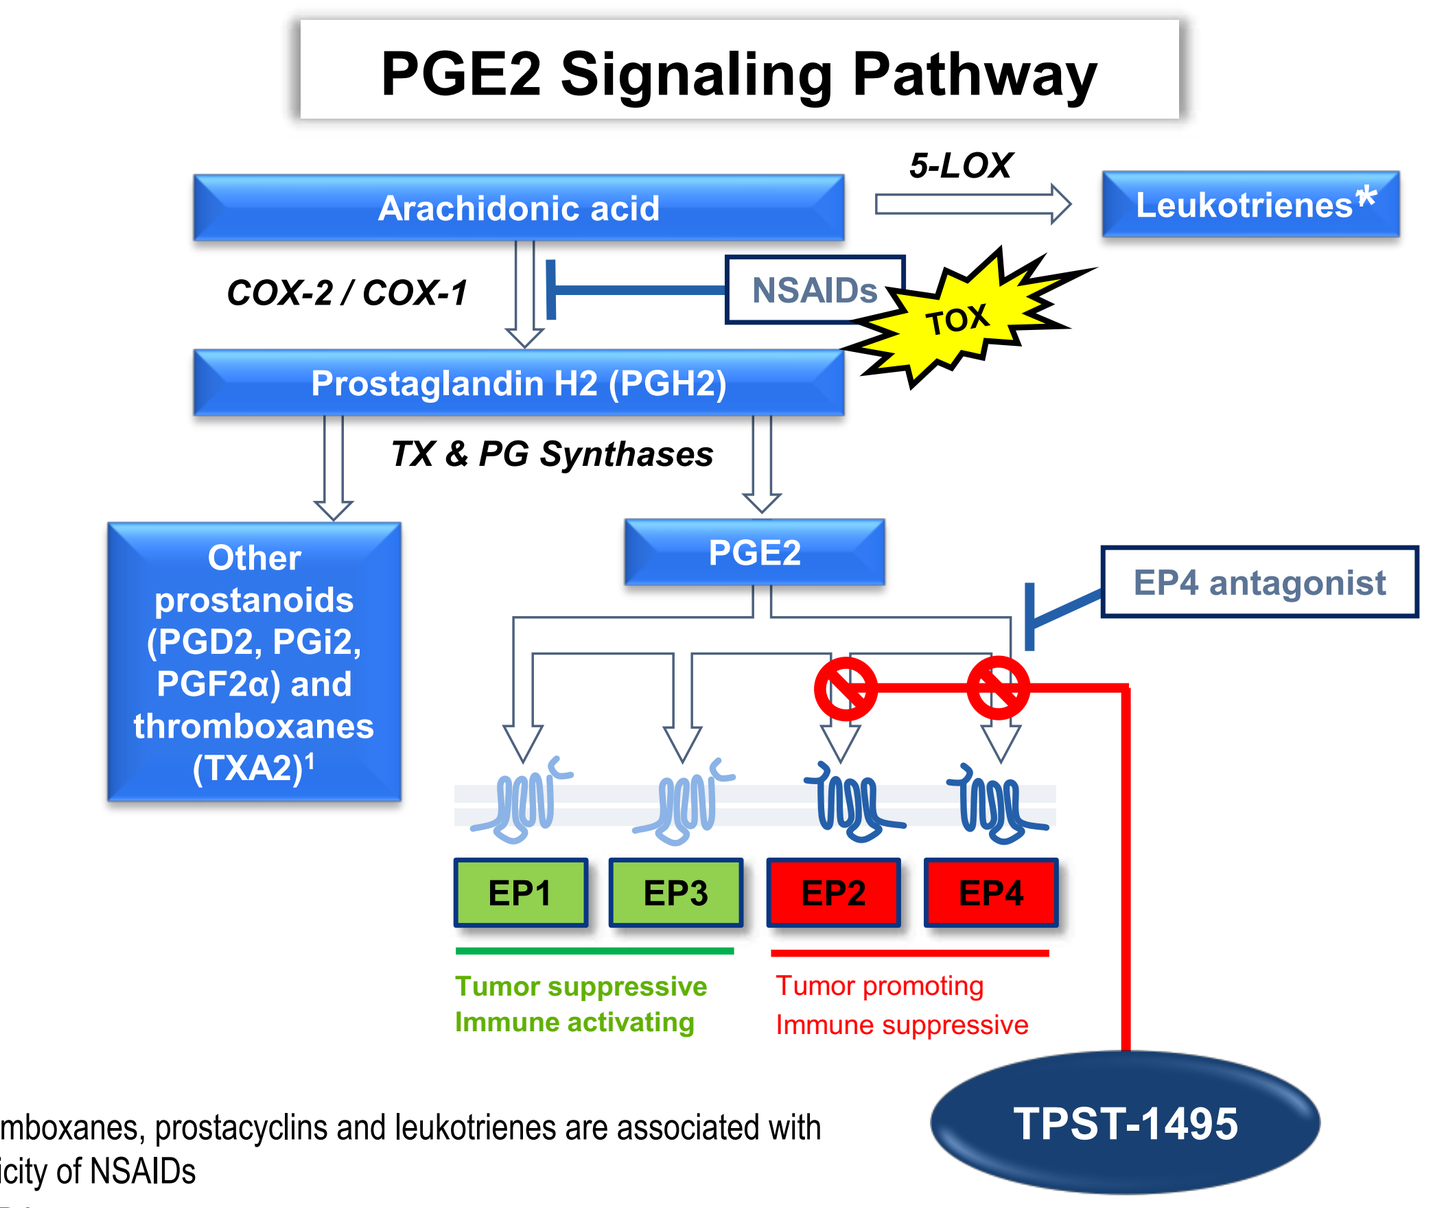
PGE2 Signaling Pathway
Arachidonic acid
5-LOX
Leukotrienes*
COX-2 / COX-1
NSAIDs
TOX
Prostaglandin H2 (PGH2)
TX & PG Synthases
Other prostanoids (PGD2, PGI2, PGF2α) and thromboxanes (TXA2)[1]
PGE2
EP4 antagonist
EP1
EP3
EP2
EP4
Tumor suppressive Immune activating
Tumor promoting Immune suppressive
TPST-1495

*Alterations in thromboxanes, prostacyclins and leukotrienes are associated with cardiovascular toxicity of NSAIDs
1 If approved by FDA
2 IC50s: 17 nM for EP2, 3 nM for EP4, and 51 nM in human whole blood assay

- Prostaglandin (PGE2) is produced by diverse advancing malignancies and drives tumor progression through autocrine signaling and immune suppression
- Enhanced COX-2 expression and PGE2 production is a mechanism of adaptive immune resistance in response to immune checkpoint blockade therapy
- PGE2 signals through four receptors, EP1-EP4, which affect distinct tumor-promoting and tumor-inhibiting activities
 - NSAIDs are toxic at high dose levels and block production of PGE2, thus signaling through all four EP receptors including the anti-tumor signaling through EP1 and EP3.
- TPST-1495 features
 - First-in-class, highly specific antagonist inhibits *only* the tumor-promoting EP2 and EP4 receptors[2]
 - Oral therapy
 - Nanomolar potency
 - Targets *both* tumor cells and immune suppressive cells

RESULTS

Whole Blood Assay to Measure TPST-1495 Immune Activation

TNF-α readout to measure TPST-1495 mediated prevention of PGE2 suppressive activity

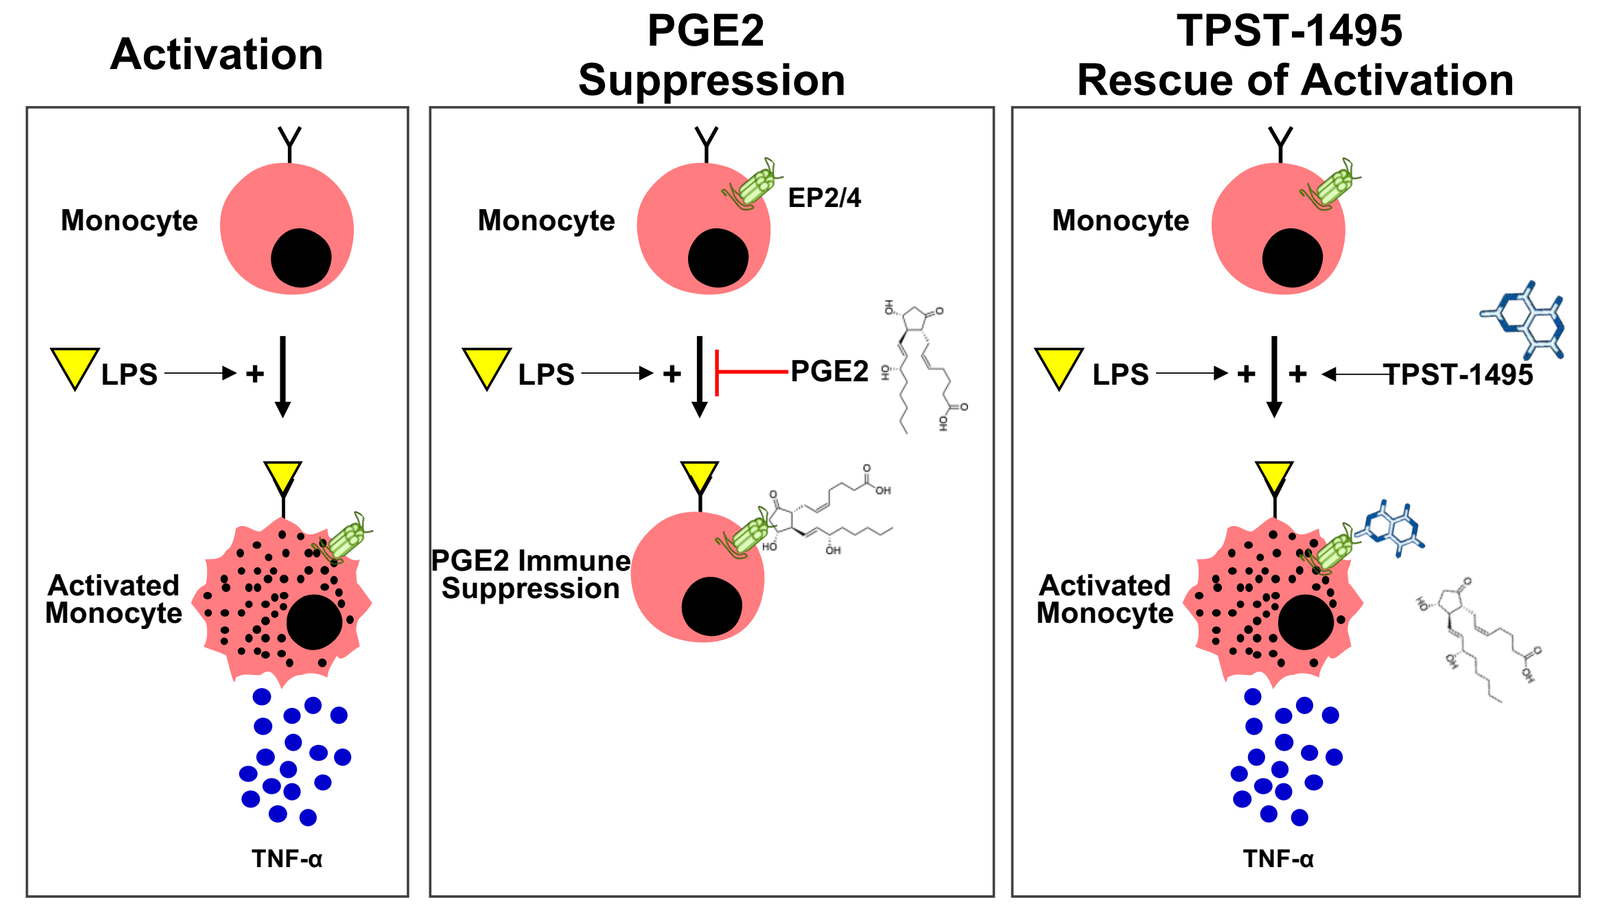
Activation
PGE2 Suppression
TPST-1495 Rescue of Activation
Monocyte
EP2/4
LPS
PGE2
TPST-1495
Activated Monocyte
PGE2 Immune Suppression
TNF-α

Whole blood assay is performed on fresh healthy whole blood specimens. Whole blood is treated with EP inhibitors for 30 minutes @37°C, followed by 30 minute incubation with 10nM or 500nM PGE2 @37°C and finally with 0.5ug/ml LPS overnight. TNFα is then read out by ELISA.

FIGURE 1: TPST-1495 prevents suppression of human and murine monocytes at high PGE2 concentrations

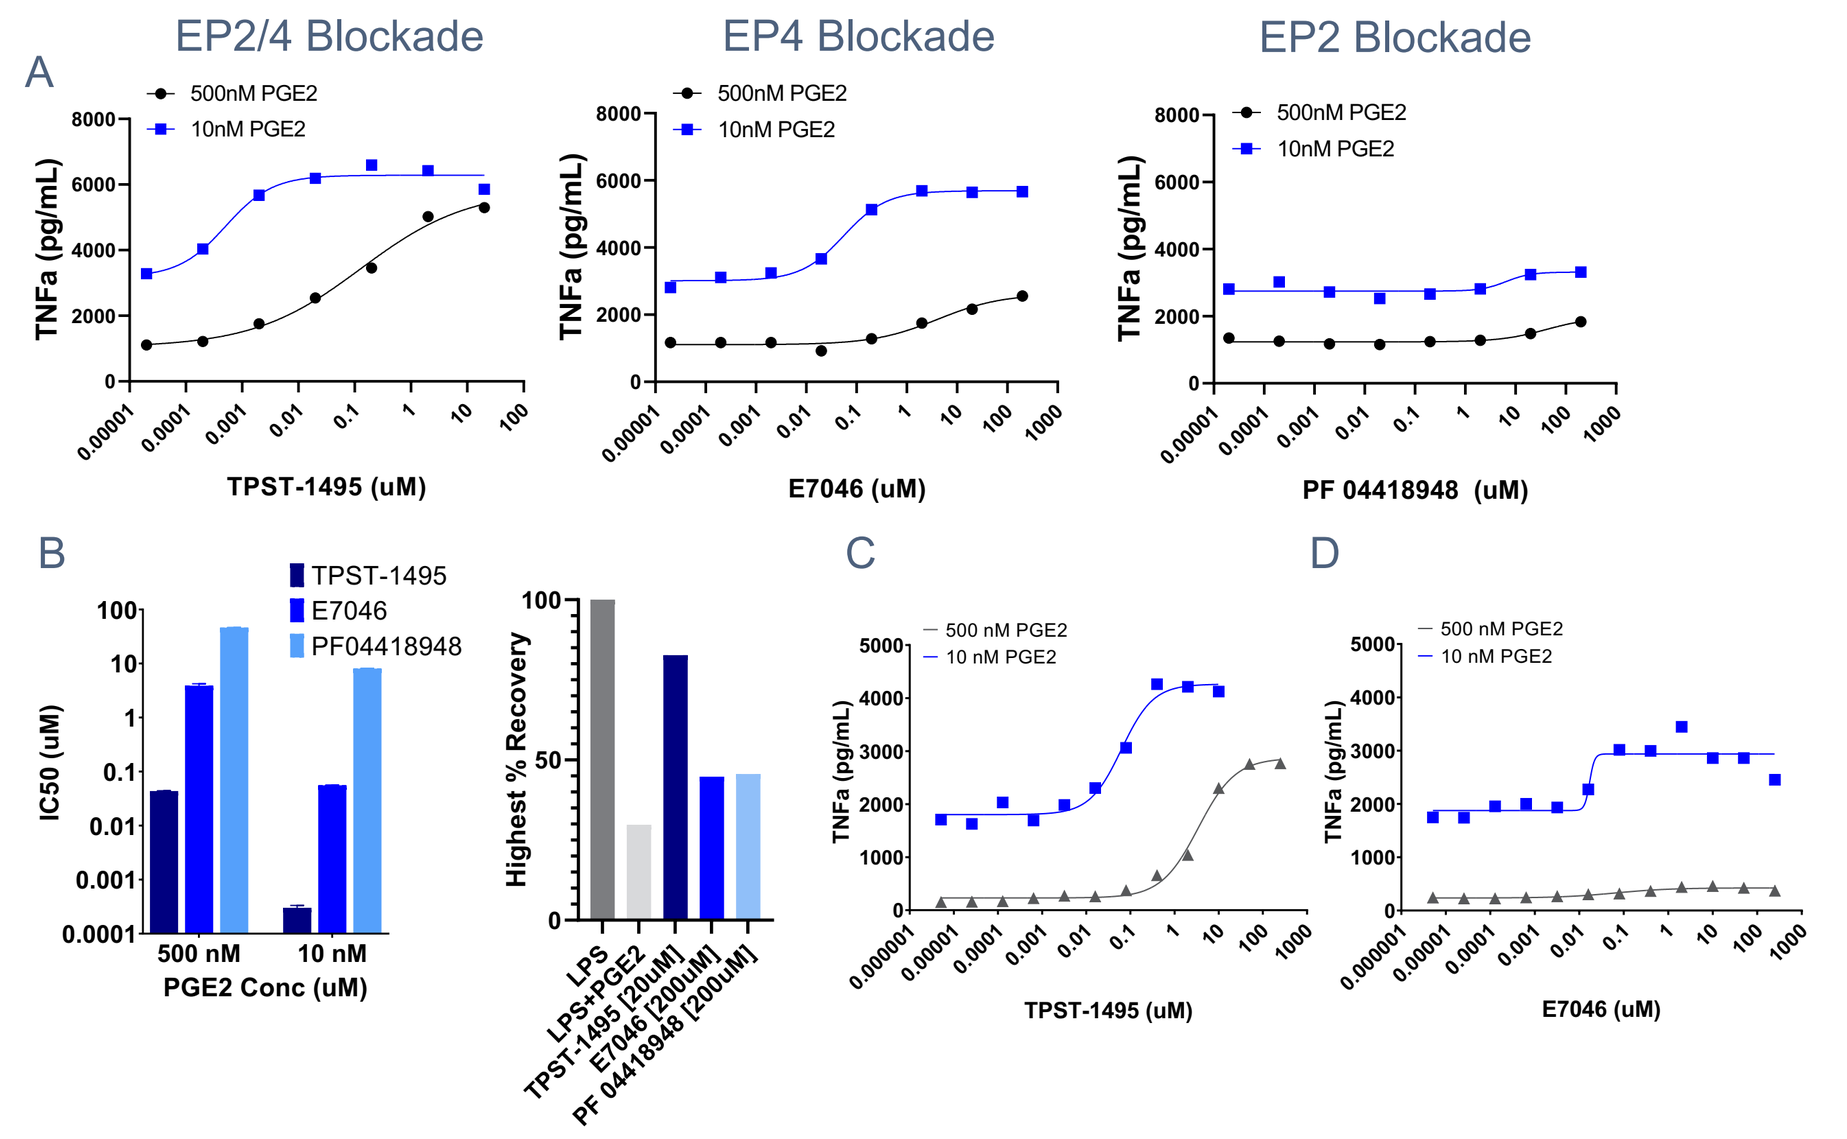
A
EP2/4 Blockade
EP4 Blockade
EP2 Blockade
500nM PGE2
10nM PGE2
TNFa (pg/mL)
TPST-1495 (uM)
E7046 (uM)
PF 04418948 (uM)
B
TPST-1495
E7046
PF04418948
IC50 (uM)
500 nM
10 nM
PGE2 Conc (uM)
Highest % Recovery
LPS
LPS+PGE2
TPST-1495 [20uM]
E7046 [200uM]
PF 04418948 [200uM]
C
D
500 nM PGE2
10 nM PGE2

Figure 1:
A,B) TNFa ELISA results from murine (A,B) and human (C,D) whole blood assays. Whole blood was procured from CROs and treated with increasing amounts of TPST-1495 (Dual EP2/4 inhibitor), E7046 (Single EP4 inhibitor), or PF 04418948 (Single EP2 inhibitor), followed by treatment with 10nM or 500nM PGE2 and 0.5ug/ml LPS ("whole blood assay"). B) IC50 and percent recovery results from whole blood assay described in (A).

FIGURE 2: TPST-1495 prevents suppression of human T Cells to a greater extent than Single EP2 or EP4 antagonism

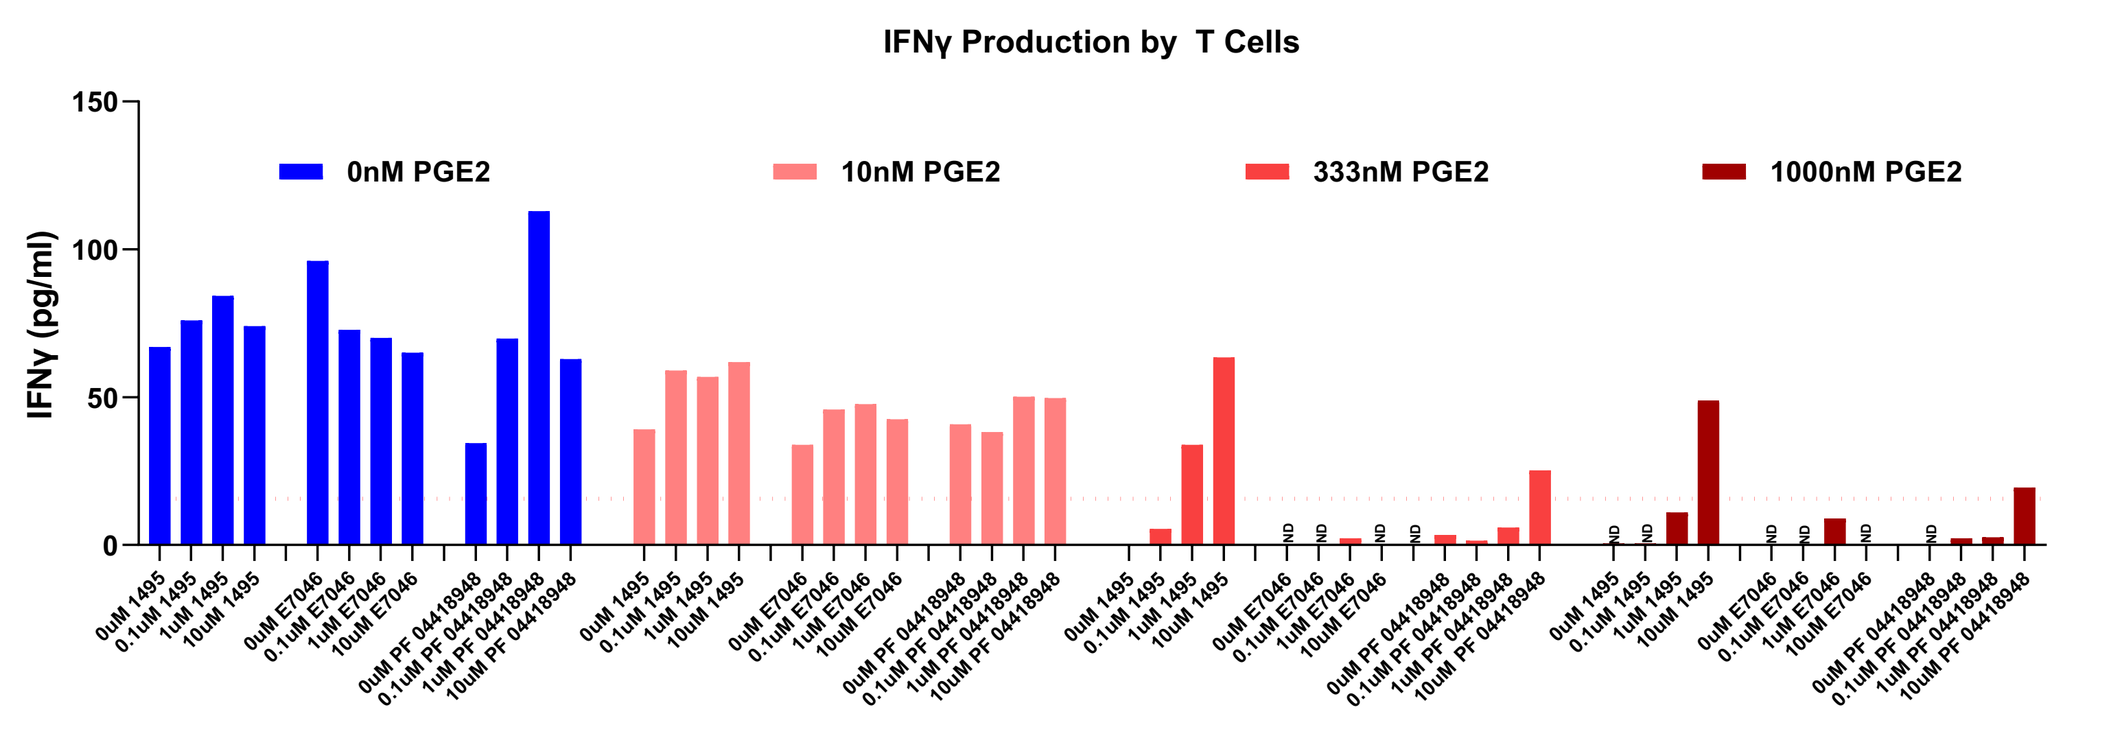
IFNγ Production by T Cells
IFNγ (pg/ml)
0nM PGE2
10nM PGE2
333nM PGE2
1000nM PGE2

Figure 2: Reversal of PGE2-mediated T cell suppression by EP2/4 antagonists. T Cells isolated from human healthy donor PBMC were treated with TPST-1495, E7046, or PF04418948 at the shown concentrations, then incubated with increasing concentrations of PGE2. After 30 minutes of drug+PGE2 exposure, PBMC were stimulated with a CEF peptide pool for 6 hours. IFNγ was then read out by ELISA. Results are representative of one donor.

FIGURE 3: TPST-1495 is more efficacious than either NSAIDs or single EP2 or EP4 pathway inhibitors

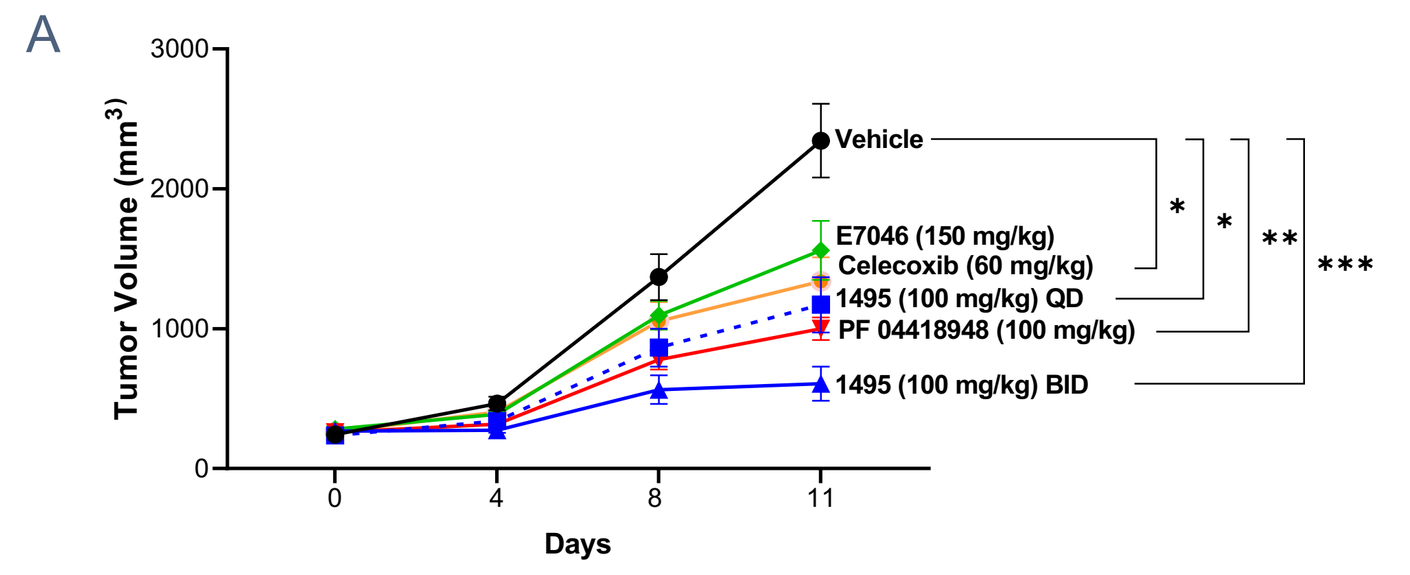
A
Tumor Volume (mm³)
Days
Vehicle
E7046 (150 mg/kg)
Celecoxib (60 mg/kg)
1495 (100 mg/kg) QD
PF 04418948 (100 mg/kg)
1495 (100 mg/kg) BID

Drug	Blockade
TPST-1495	EP2/EP4
E7046	EP4 only
PF04419848	EP2 only
Celecoxib	COX-2

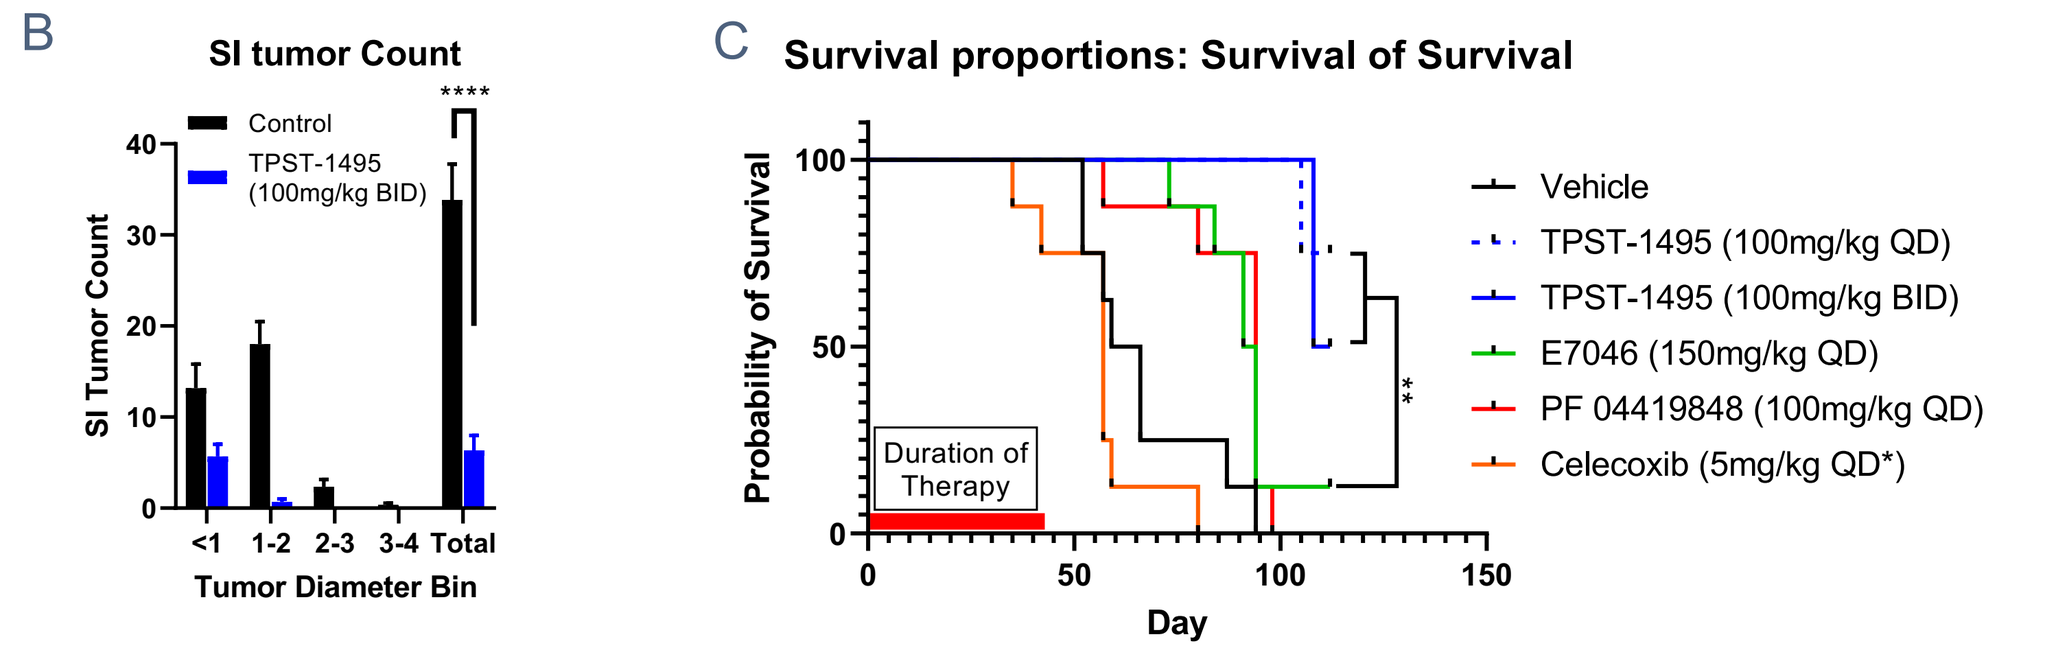
B
SI tumor Count
Control
TPST-1495 (100mg/kg BID)
SI Tumor Count
Tumor Diameter Bin
C
Survival proportions: Survival of Survival
Probability of Survival
Day
Duration of Therapy
Vehicle
TPST-1495 (100mg/kg QD)
TPST-1495 (100mg/kg BID)
E7046 (150mg/kg QD)
PF 04419848 (100mg/kg QD)
Celecoxib (5mg/kg QD*)

Figure 3:
A) Lewis Lung Carcinoma (LLC) Tumors treated with listed EP antagonists. Tumors were implanted in flanks of animals on day -11, then assorted into groups where tumor volumes averaged 200mm^3 and treated with listed EP antagonists.
B) Tumor count in small intestines of $APC^{min/+}$ mice treated for 8 weeks, starting at 6 weeks of age with orally administered TPST-1495 at 100mg/kg BID.
C) Survival of $APC^{min/+}$ animals treated beginning at 12 weeks of age with listed EP antagonists. All antagonists were administered PO for 6 weeks, except for Celecoxib which was administered IP. Statistics were calculated by the Gehan-Breslow-Wilcoxon test.

FIGURE 4: TPST-1495 exhibits immunomodulatory effects and adds to checkpoint therapy

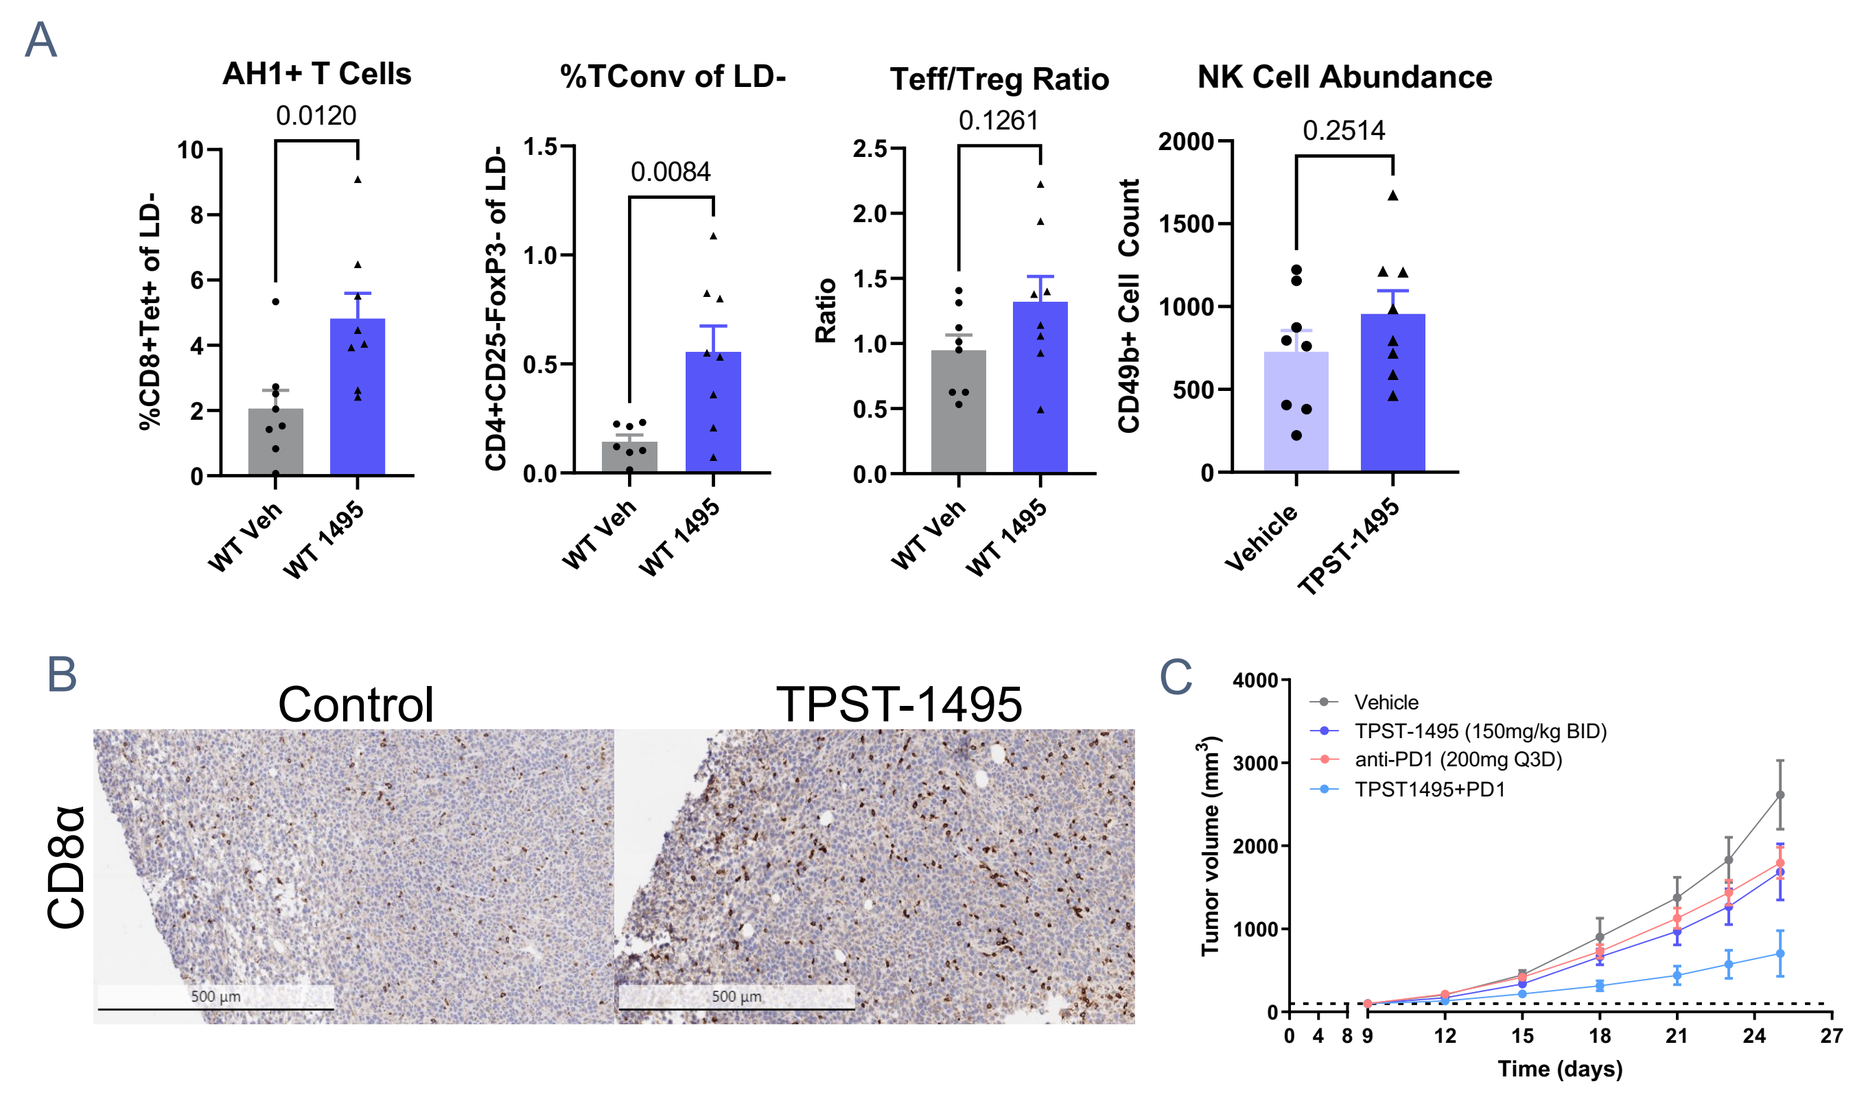
A
AH1+ T Cells
%TConv of LD-
Teff/Treg Ratio
NK Cell Abundance
0.0120
0.0084
0.1261
0.2514
WT Veh
WT 1495
Vehicle
TPST-1495
B
Control
TPST-1495
CD8α
C
Vehicle
TPST-1495 (150mg/kg BID)
anti-PD1 (200mg Q3D)
TPST1495+PD1
Tumor volume (mm³)
Time (days)

Figure 4:
A) Results of flow cytometry from flank-implanted CT26 tumors in BABL/c mice. Tumors were treated as group averages approached 100mm^3 in volume for 14 days, then sacrificed and TIL harvested. TPST-1495 was administered at 100mpk BID PO in methylcellulose.
B) CD8α Immunohistochemistry from flank-implanted CT26 tumors in BABL/c mice. Tumors were treated as group averages approached 100mm^3 in volume for 14 days, then fixed in formalin and imbedded in paraffin. TPST-1495 was administered at 100mpk BID PO in methylcellulose.
C) Tumor outgrowth of CT26 Tumors treated with 150mg/kg TPST-1495 BID PO. Anti-PD-1 was administered at 10mpk in PBS IP 2x weekly.

FIGURE 5: T Cell-Independent effects also play a significant role during TPST-1495 therapy

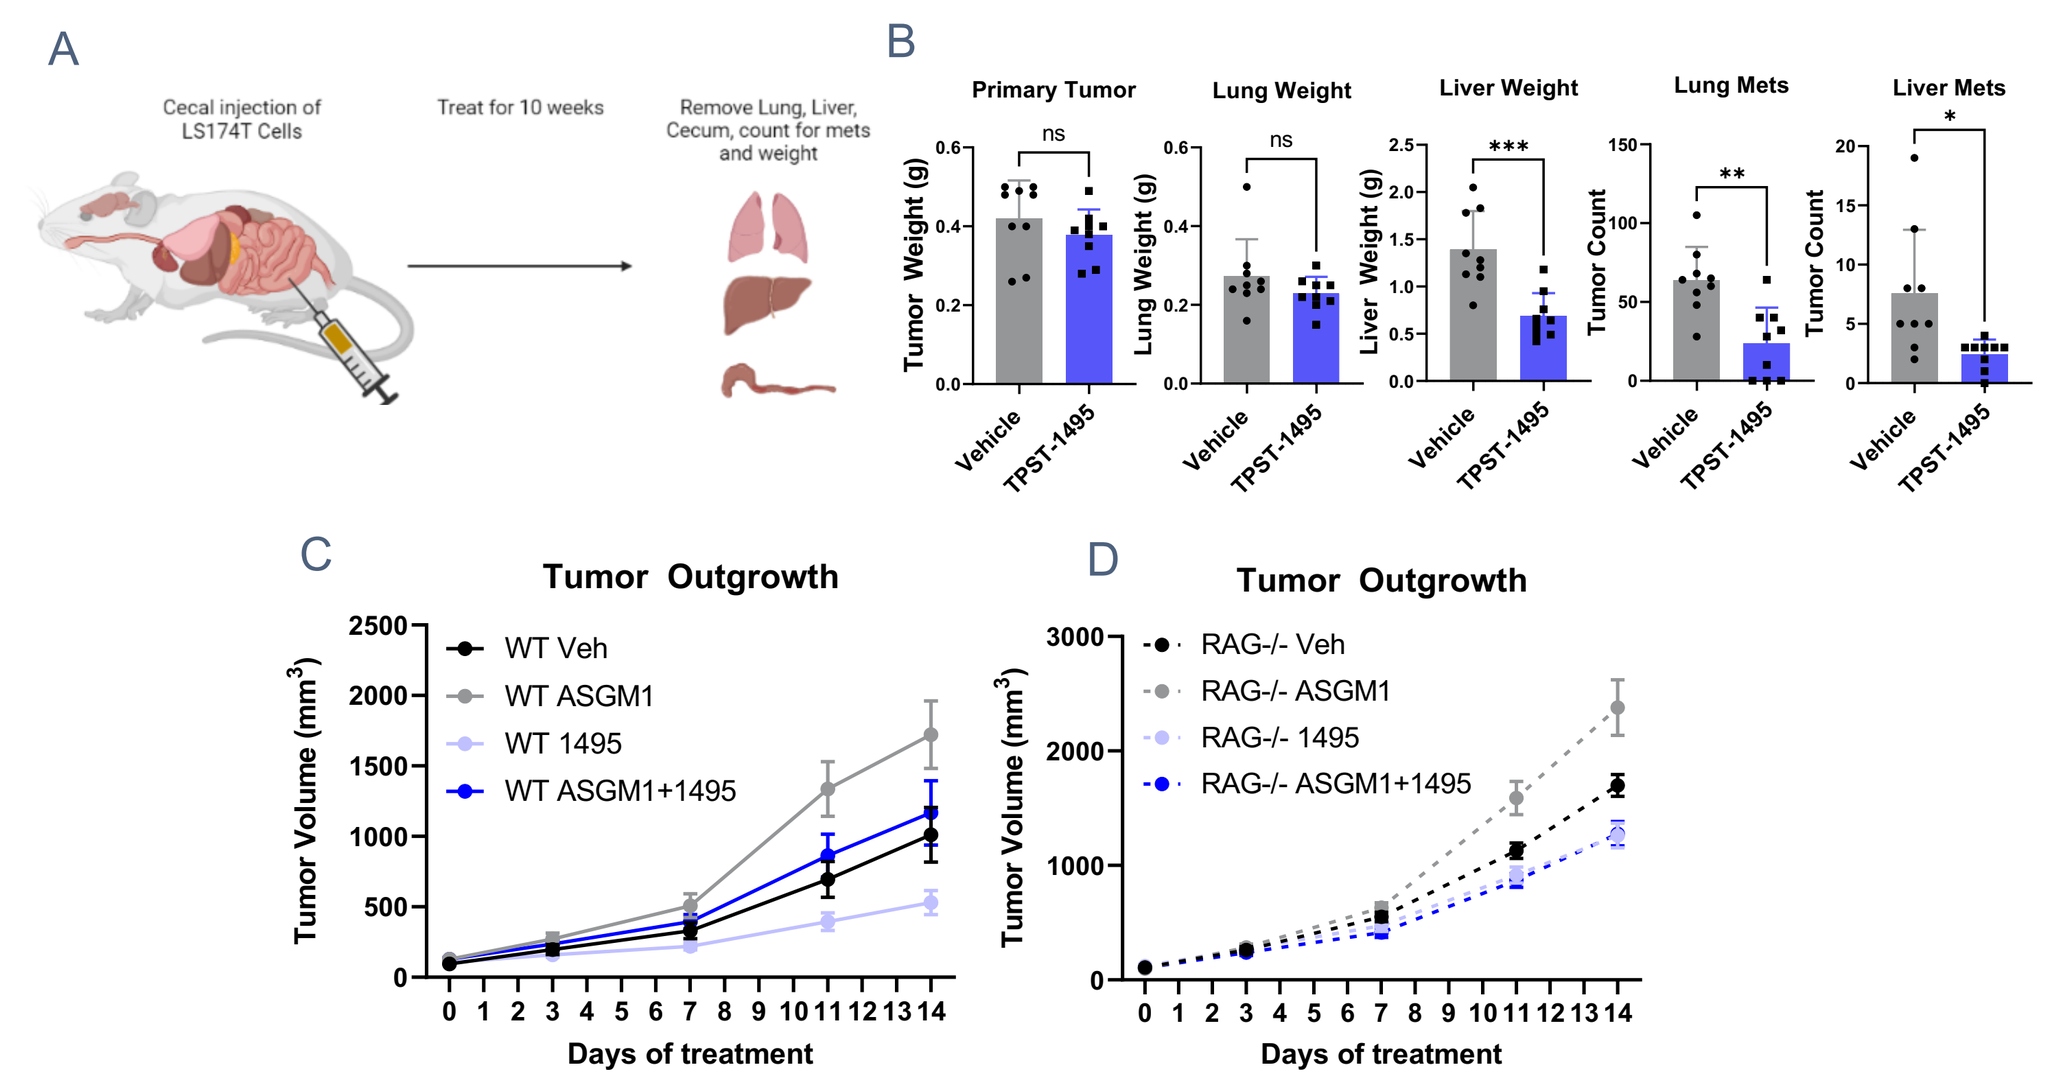
A
Cecal injection of LS174T Cells
Treat for 10 weeks
Remove Lung, Liver, Cecum, count for mets and weight
B
Primary Tumor
Lung Weight
Liver Weight
Lung Mets
Liver Mets
Vehicle
TPST-1495
C
Tumor Outgrowth
WT Veh
WT ASGM1
WT 1495
WT ASGM1+1495
Days of treatment
D
RAG-/- Veh
RAG-/- ASGM1
RAG-/- 1495
RAG-/- ASGM1+1495

Figure 5:
A,B) 2.5 x 10⁴ of LS-174T cells were injected into the cecal wall of male NSG mice at age of 8 weeks old. After 5 days, mice were randomly divided into 2 groups treated with methylcellulose or methylcellulose containing TPST-1495 (50mg/kg, BID) by gavage for 2 weeks. Then the mice were treated with methylcellulose or methylcellulose containing TPST-1495 (25mg/kg, BID) by gavage for 7 weeks. Tumor counts and organ weights at the time of sacrifice, 10 weeks after the initiation of treatment.
C) Tumor outgrowth of CT26 tumors implanted in WT BALB/c Mice. 1e6 CT26 tumor cells were injected in flanks of animals and treated as averaged group tumor volumes approached 100mm^3. ASGM1 antibody was used to deplete NK cells at -1 days and time of injection of other therapeutic interventions, followed by weekly injections thereafter.
D) RAG2-/- animals were treated in the same way as BALB/c mice in (C).

CONCLUSIONS

- Prostaglandin (PGE2) is produced by diverse advancing malignancies and drives tumor progression through both autocrine signaling and immune suppression TPST-1495 is a novel, first-in-class, dual antagonist of both EP2 and EP4 prostaglandin (PGE2) receptors
- TPST-1495 confers near complete restoration of immune function in cellular assays even in the presence of super-physiological PGE2 concentrations, conditions in which single EP4 or EP2 inhibitors were not effective **(Fig 1)**
- TPST-1495 therapy promotes anti-tumor activity through both T cell-independent and T-cell dependent mechanisms, as evidenced by TME infiltration of effector immune cell populations and tumor Ag-specific CD8+ T cells **(Figs 2, 4, 5)**
- TPST-1495 therapy confers a significant survival advantage compared to therapy with single EP2, EP4 antagonists or the NSAID Celecoxib in the $APC^{min/+}$ spontaneous tumor mouse model of CRC **(Fig 3)**
- TPST-1495 is currently being evaluated in a schedule and dose-finding Phase 1a clinical study in patients with advanced solid tumors (NCT04344795)
- Additional Phase 1b hypothesis-driven clinical studies including in patients with PIK3CA mutant-driven cancers and in combination with pembrolizumab are planned **(Fig 6)**

CITATIONS

1. Pelly VS, Moeini A, Roelofsen LM, et al. Anti-Inflammatory Drugs Remodel the Tumor Immune Environment to Enhance Immune Checkpoint Blockade Efficacy. Cancer Discov. 2021;11(10):2602-2619. doi: 10.1158/2159-8290.CD-20-1815.

2. Tury S, Becette V, Assayag F, et al. Combination of COX-2 expression and PIK3CA mutation as prognostic and predictive markers for celecoxib treatment in breast cancer. Oncotarget. 2016;7(51):85124-85141. doi: 10.18632/oncotarget.13200.

3. Zelenay S, van der Veen AG, Böttcher JP, et al. Cyclooxygenase-Dependent Tumor Growth through Evasion of Immunity. Cell. 2015;162(6):1257-70. doi: 10.1016/j.cell.2015.08.015.

Ph1b EXPANSION DESIGN

FIGURE 6: PIK3CA Driver Mutation Promotes Tumor Growth and PGE2 Production

PIK3CA mutation predictive of NSAID benefit in CRC and SCCHN

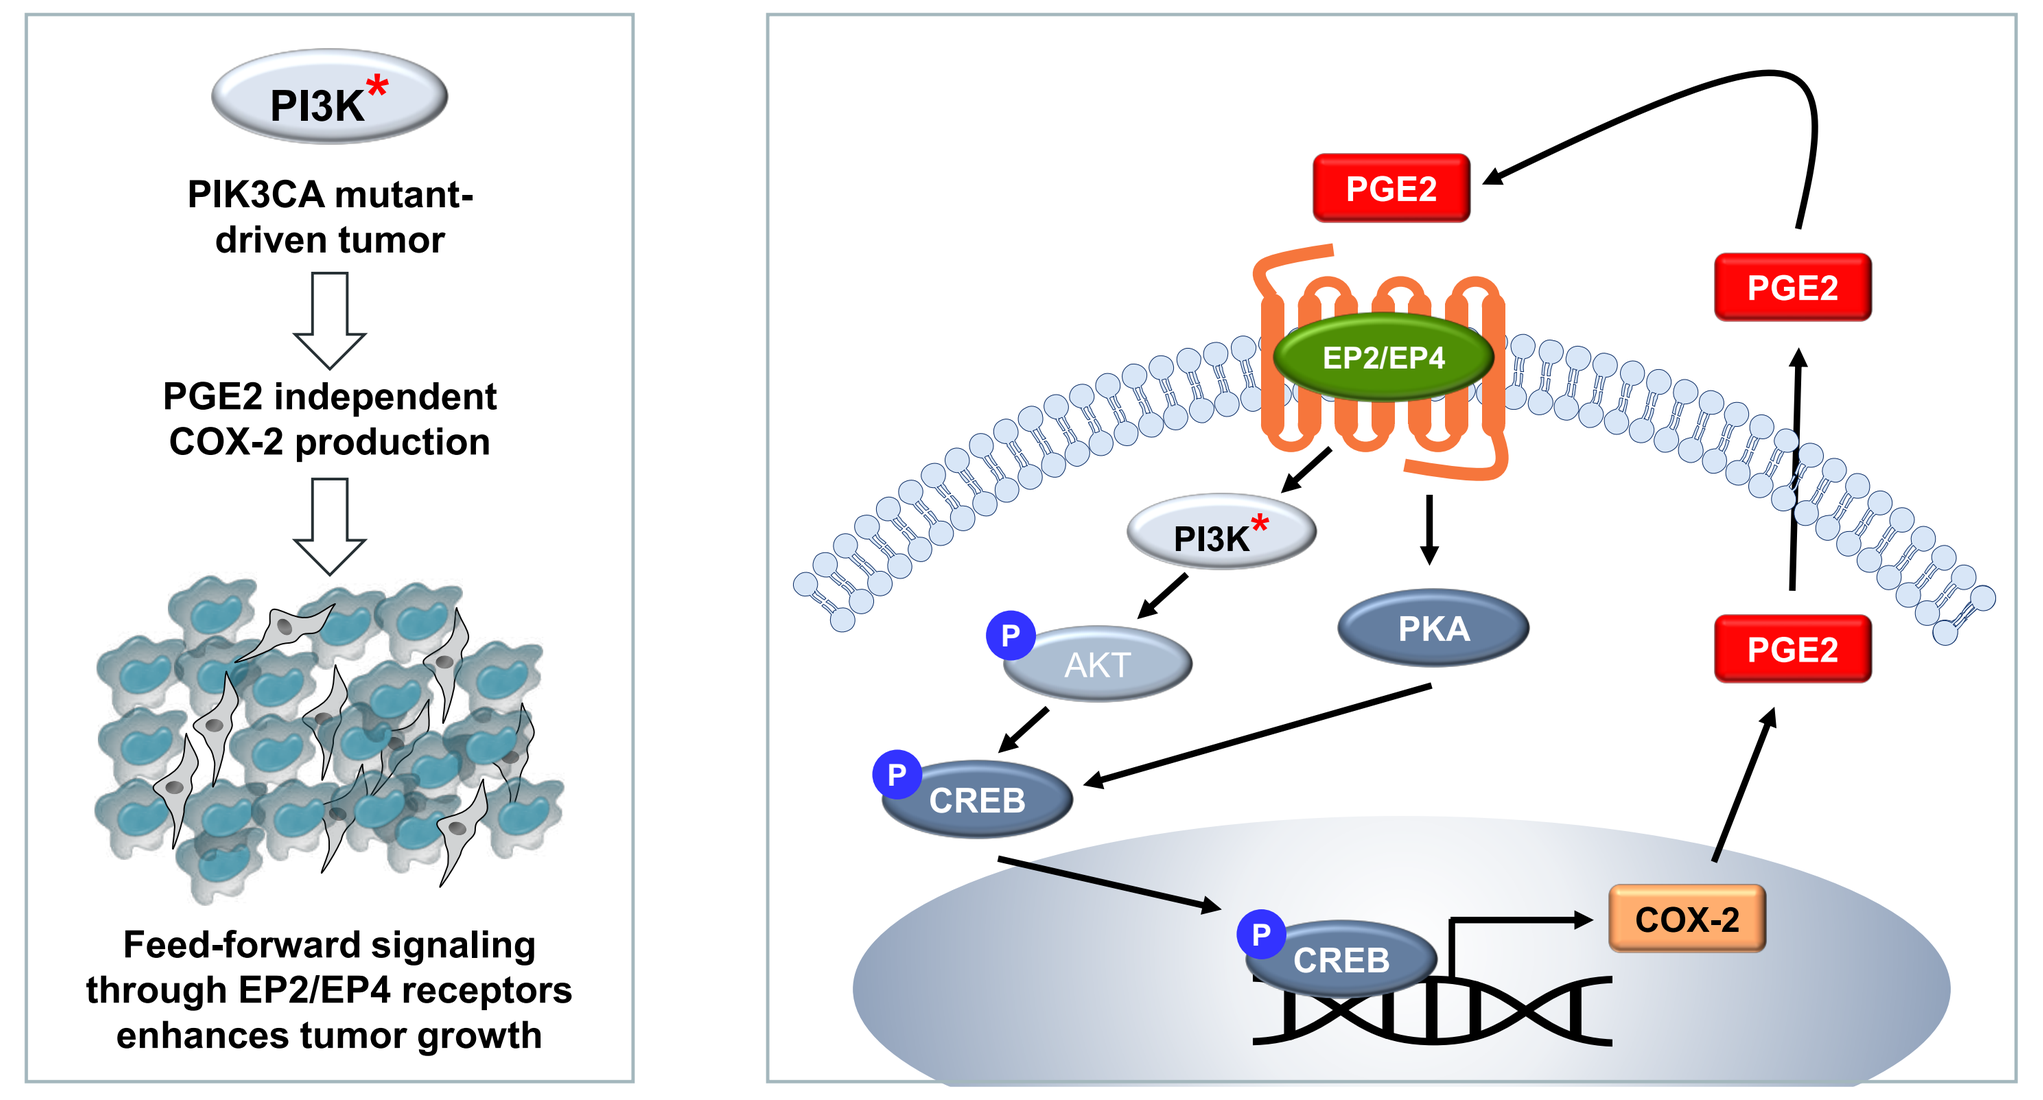
PI3K*
PIK3CA mutant-driven tumor
PGE2 independent COX-2 production
Feed-forward signaling through EP2/EP4 receptors enhances tumor growth
PGE2
EP2/EP4
PI3K*
AKT
PKA
CREB
COX-2

- PIK3CA tumor driver mutation constitutively activates cell proliferation and production of PGE2 and may be a biomarker for TPST-1495-responsive tumors

Adopted from Yang et. Al., OncoTargets and Therapy, 2020.

TPST-1495 Clinical Development Strategy

Maximize PTS for signal detection and potentially broader development opportunity

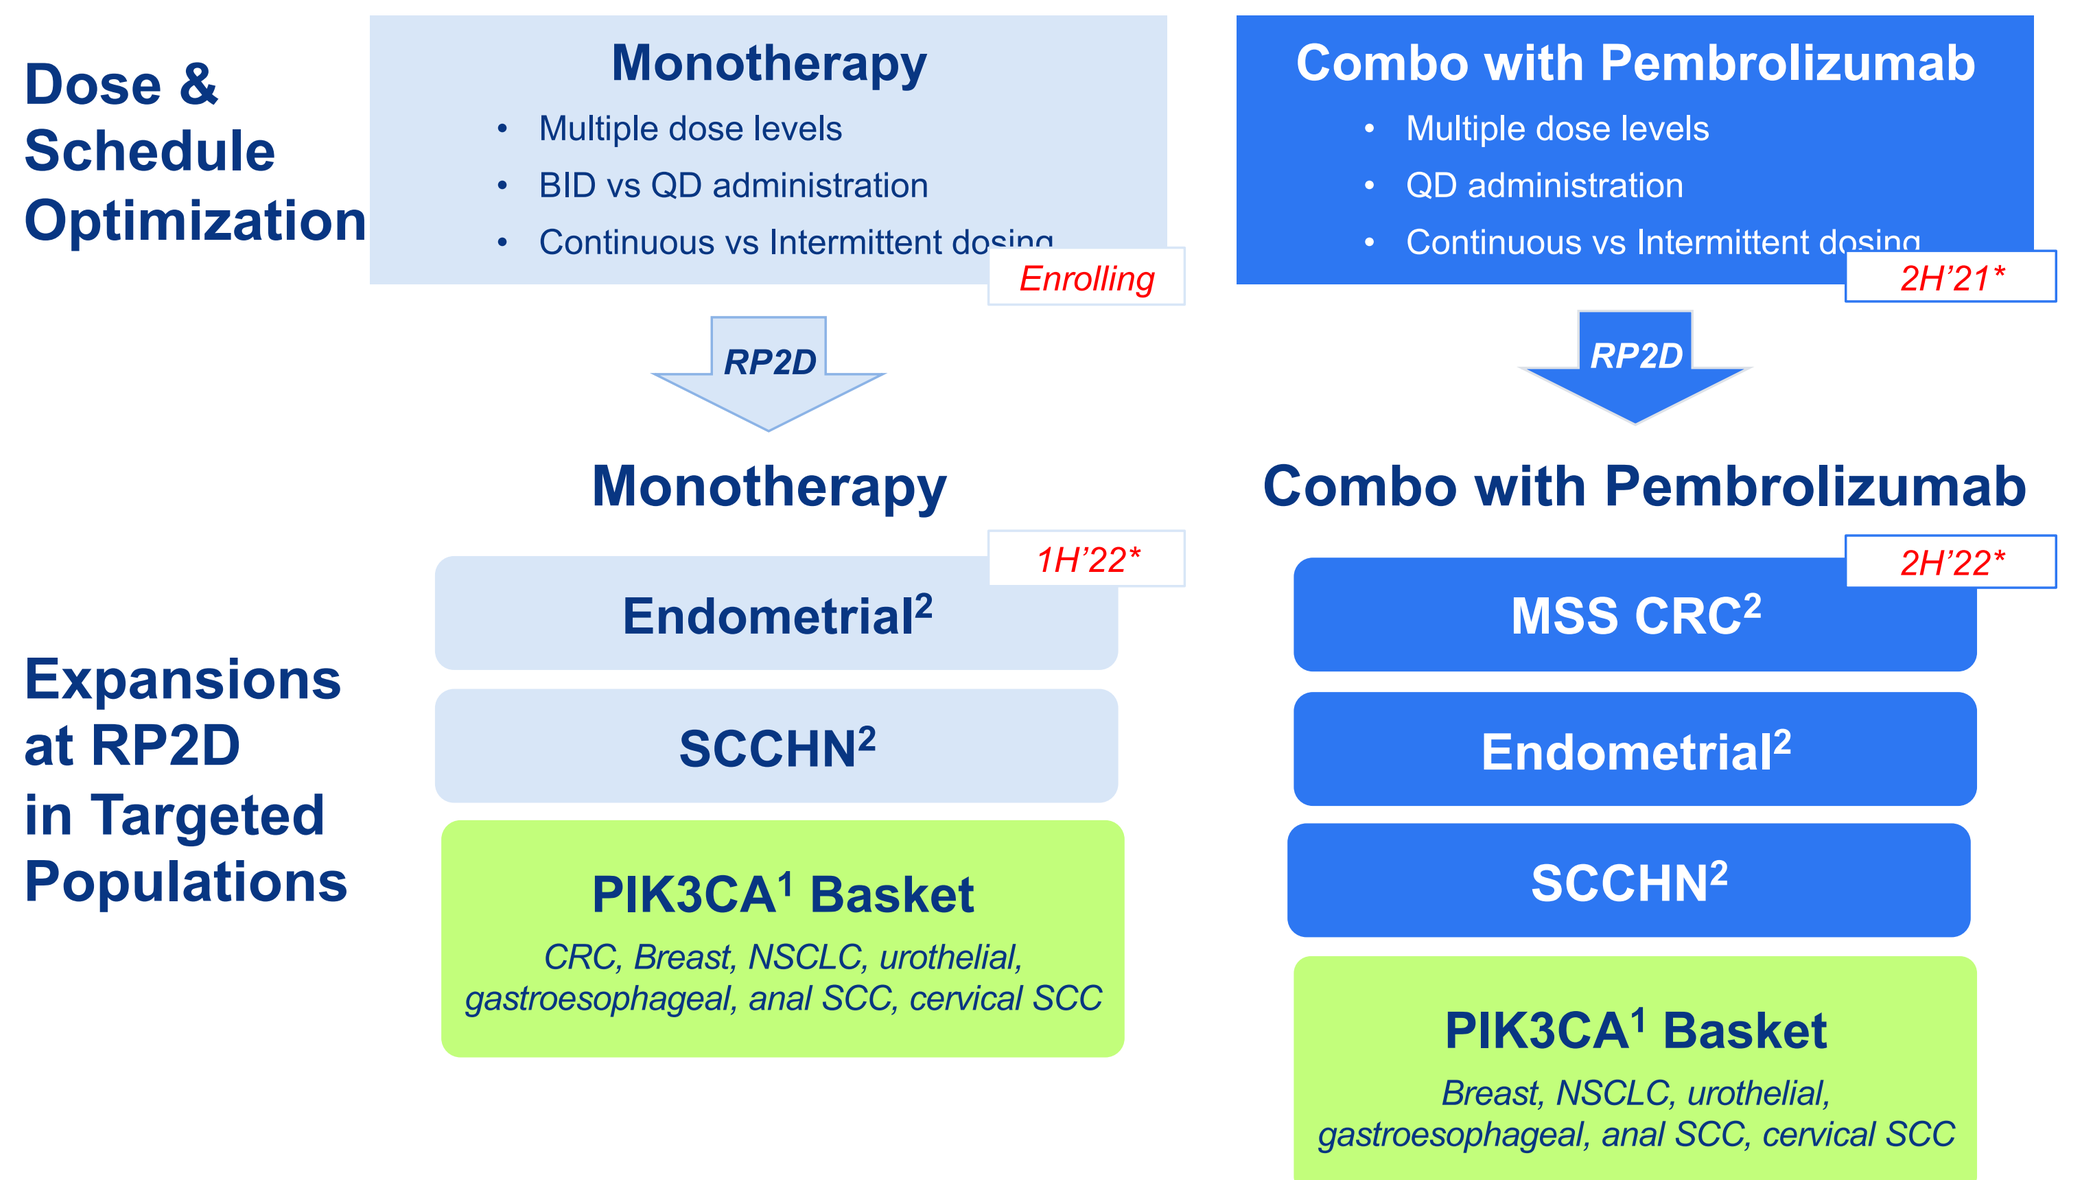
Dose & Schedule Optimization
Monotherapy
Multiple dose levels
BID vs QD administration
Continuous vs Intermittent dosing
Enrolling
Combo with Pembrolizumab
Multiple dose levels
QD administration
Continuous vs Intermittent dosing
2H'21*
RP2D
Expansions at RP2D in Targeted Populations
Monotherapy
1H'22*
Endometrial²
SCCHN²
PIK3CA¹ Basket
CRC, Breast, NSCLC, urothelial, gastroesophageal, anal SCC, cervical SCC
Combo with Pembrolizumab
2H'22*
MSS CRC²
Endometrial²
SCCHN²
PIK3CA¹ Basket
Breast, NSCLC, urothelial, gastroesophageal, anal SCC, cervical SCC

* Anticipated initiation
[1] *Must have documented pathogenic mutation in PIK3CA*
[2] *Both wild-type and mutant PIK3CA*