P440: Phase 1/1b Multicenter Trial of TPST-1120, a Peroxisome Proliferator-Activated Receptor Alpha (PPARα) Antagonist as a Single Agent (SA) or in Combination in Patients with Advanced Solid Tumors

John D. Powderly II[1], Saurin Chokshi[2], Johanna Bendell[2], Leisha A. Emens[3], Jason J. Luke[3], Brian Francica[4], Dipak Panigrahy[5], Chan C Whiting[4], Thomas W Dubensky Jr[4], Ginna Laport[4]

[1]Carolina BioOncology Institute, Huntersville, NC; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TX; [3]University of Pittsburgh Medical Center, Pittsburg, PA ; [4]Tempest Therapeutics, Inc. San Francisco, CA, [5]Beth Israel Deaconess Medical Center, Harvard Medical School, Boston, MA

ABSTRACT

Background

Tumor cells initially rely on glucose consumption via aerobic glycolytic pathways. However, as tumor cells proliferate and metastasize in an increasingly hypoxic tumor microenvironment (TME), tumors increasingly utilize fatty acid oxidation (FAO) as glucose stores are depleted. FAO supports both tumor growth and suppressive immune cells in the TME, facilitating tumor progression. PPARα is a ligand-activated nuclear transcription factor which regulates lipid metabolism, FAO and inflammation. TPST-1120 is a first in class, oral, selective PPARα antagonist that blocks transcription of PPARα target genes leading to a metabolic shift from FAO to glycolysis. Antagonism of FAO in the TME leads to direct killing of tumor cells dependent on FAO and facilitates the cytotoxicity of effector cells. Preclinical studies with various tumor models demonstrate efficacy of TPST-1120 as monotherapy and in combination with anti-PD1 antibodies or chemotherapy. TPST-1120 has an IC50 of 0.04 nM with a >35 fold selectivity over other PPAR isoforms.

Methods

We have initiated a phase 1/1b multicenter, open label trial to evaluate TPST-1120 as a SA and in combination (combo) with other systemic therapies including nivolumab, an anti-PD1 monoclonal antibody; docetaxel, a cytotoxic chemotherapeutic agent and cetuximab, an anti-EGFR monoclonal antibody. The objectives are to 1) evaluate safety and tolerability of continuous dosing of TPST-1120 2) identify a recommended phase 2 dose (RP2D) 3) evaluate efficacy, and 4) evaluate PK/PD parameters. Eligibility criteria: 1) histologies: non-small cell lung cancer(ca), hepatocellular ca, renal cell ca, triple-negative breast ca, urothelial ca, pancreatic ca, cholangiocarcinoma, gastro-esophageal ca, castration resistant prostate ca, head and neck ca, sarcoma and MSS colorectal ca and 2) 1 but not more than 5 prior therapies. This phase 1/1b adaptive design is composed of Dose Escalation (DEs) and Dose Expansion (DEx) cohorts. DEs is comprised of 4 arms, 1 SA arm and 3 combination arms in which TPST-1120 is combined with nivolumab, docetaxel or cetuximab. The RP2D of TPST-1120 to proceed to DEx will be determined by safety and biomarkers including analysis of FAO/PPARα gene expression in the periphery and in tumor biopsies. The DEx arms will follow a 2-stage expansion design. This trial began accrual in May 2018 at U.S sites and is currently enrolling into the Monotherapy/Dose Escalation cohort. Expansion cohorts are projected to open in early 2019. The total sample size is up to 338 pts.

PPARα FUNCTION

FIGURE 1: PPARα Induces Fatty Acid Oxidation (FAO)

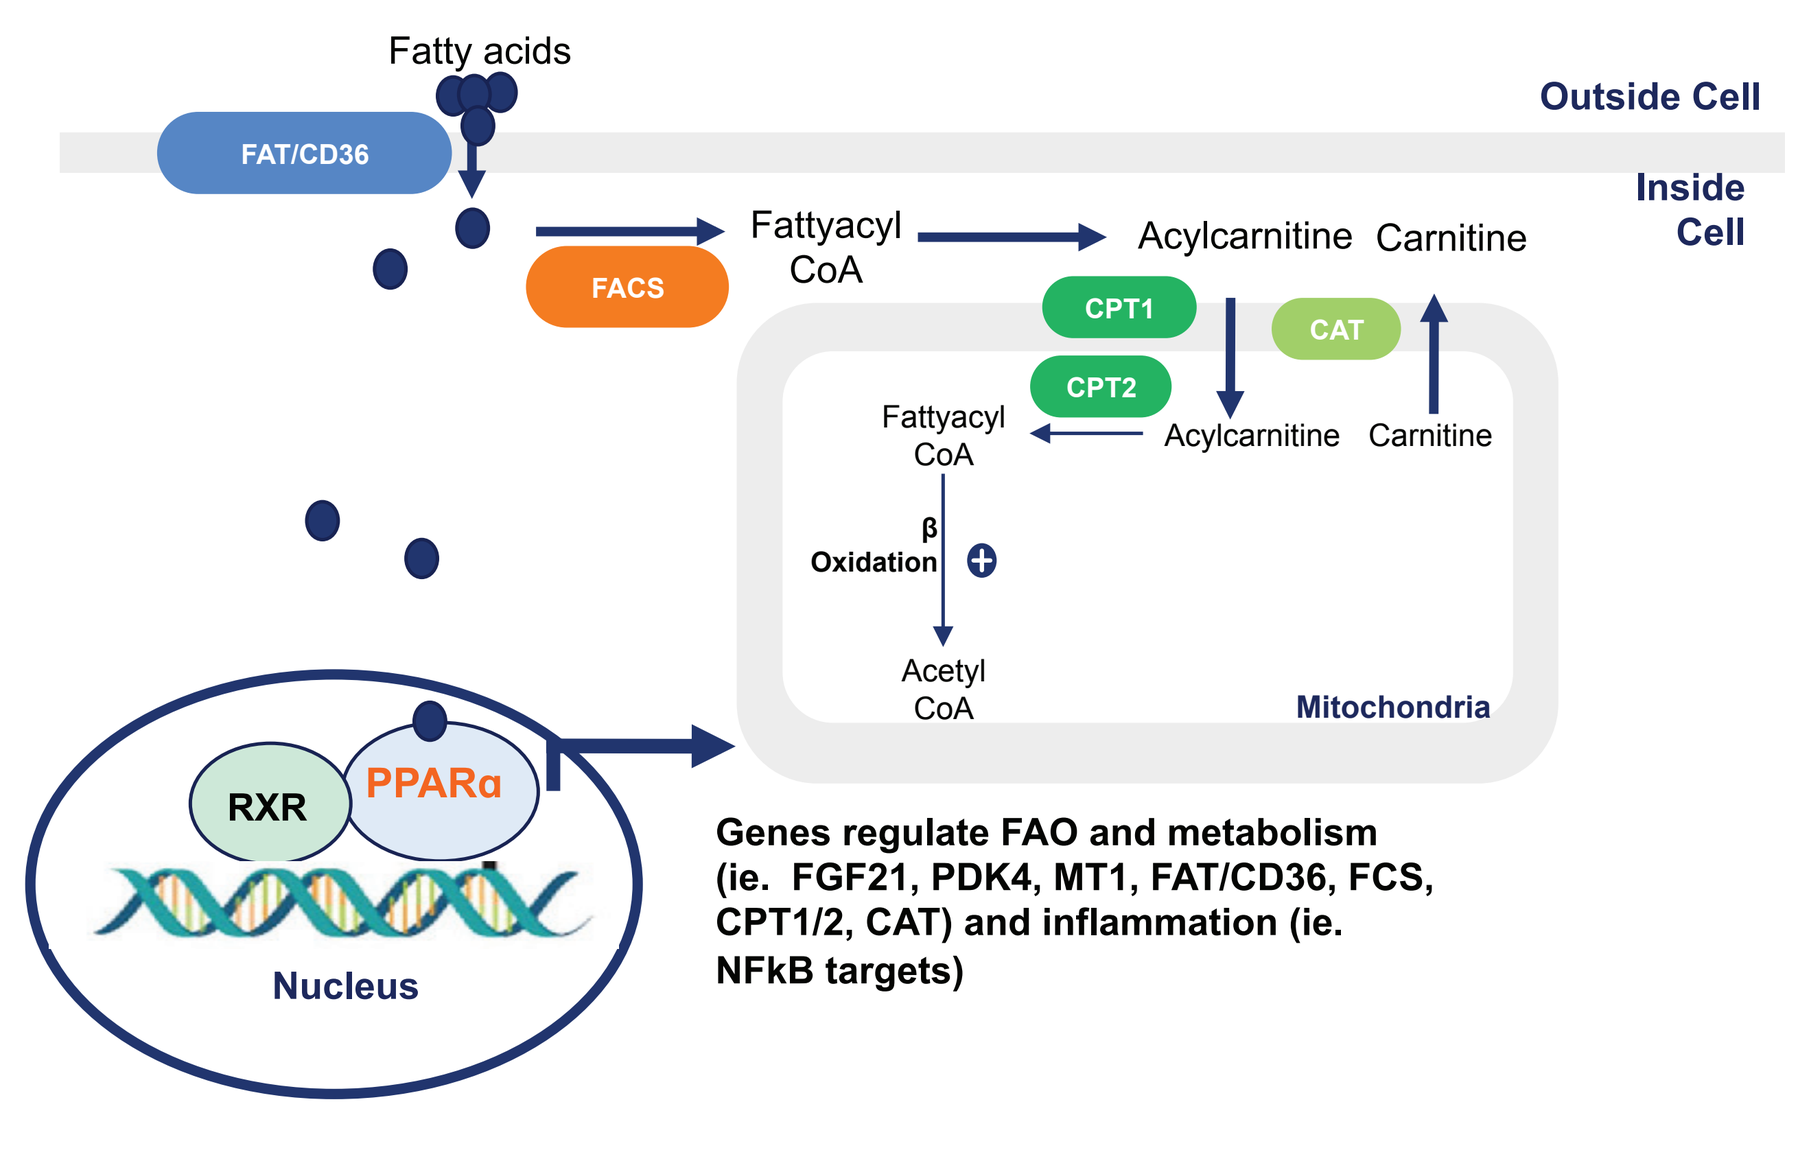
Fatty acids
Outside Cell
Inside Cell
FAT/CD36
FACS
Fattyacyl CoA
Acylcarnitine
Carnitine
CPT1
CAT
CPT2
Fattyacyl CoA
Acylcarnitine
Carnitine
β Oxidation
Acetyl CoA
Mitochondria
RXR
PPARα
Nucleus
Genes regulate FAO and metabolism (ie. FGF21, PDK4, MT1, FAT/CD36, FCS, CPT1/2, CAT) and inflammation (ie. NFkB targets)

Figure 1: PPARα regulates key genes involved in FAO, metabolism and inflammation. Upon ligand binding, PPARα heterodimerizes with retinoic acid receptor (RXR) and stimulates transcription of target genes involved in FAO and lipid metabolism. PPARα also negatively modulates gene transcription by inhibiting DNA binding of several other transcription factors, such as NF-κB.

BACKGROUND

- Tumor cells initially favor glucose metabolism via aerobic glycolysis
- As tumor proliferate and metastasize, glucose stores are depleted in context of hypoxic tumor microenvironment (TME)
- Metabolic reprogramming shifts intracellular metabolism toward fatty acid oxidation (FAO)
- **PPARα** is a nuclear transcription factor which regulates lipid metabolism and FAO
- **TPST-1120 is a 'first in class' oral selective PPARα antagonist that blocks transcription of PPAR target genes**
 - TPST-1120 leads to intracellular metabolism shift from FAO to glycolysis
 - TPST-1120 has an IC50 of 0.04 nM with > 35 fold selectivity over other PPAR isoforms
 - Preclinical models shows efficacy of TPST-1120 as a single agent and in combination with an anti-PD1 antibody and chemotherapy

FIGURE 2: Rationale for PPARα Antagonist

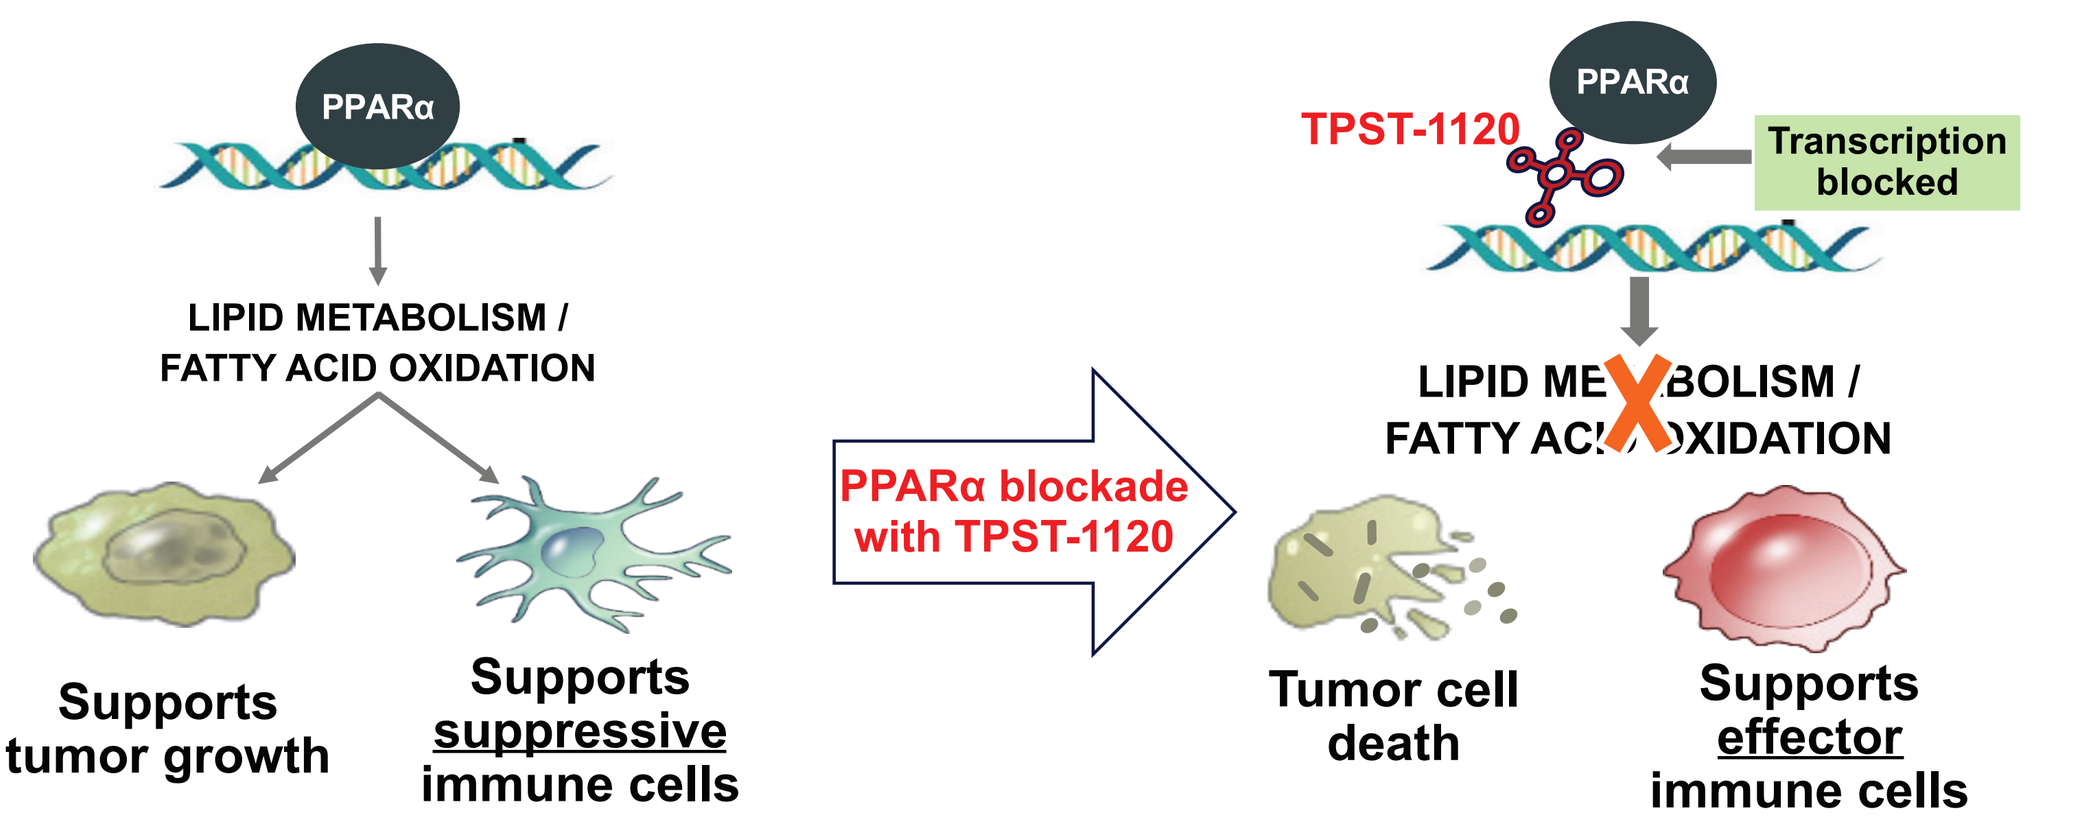
PPARα
LIPID METABOLISM / FATTY ACID OXIDATION
Supports tumor growth
Supports suppressive immune cells
PPARα blockade with TPST-1120
TPST-1120
PPARα
Transcription blocked
Tumor cell death
Supports effector immune cells

Figure 2:
• Fatty acids within the TME support tumor growth and suppressive immune cells (Tregs, MDSCs, etc)
• PPARα blockade inhibits FAO → direct tumor cell death and support of effector immune cells

FIGURE 3: Upregulation of PPARα and FAO genes in diverse tumors (TCGA-based analysis of tumor metabolic profiles)

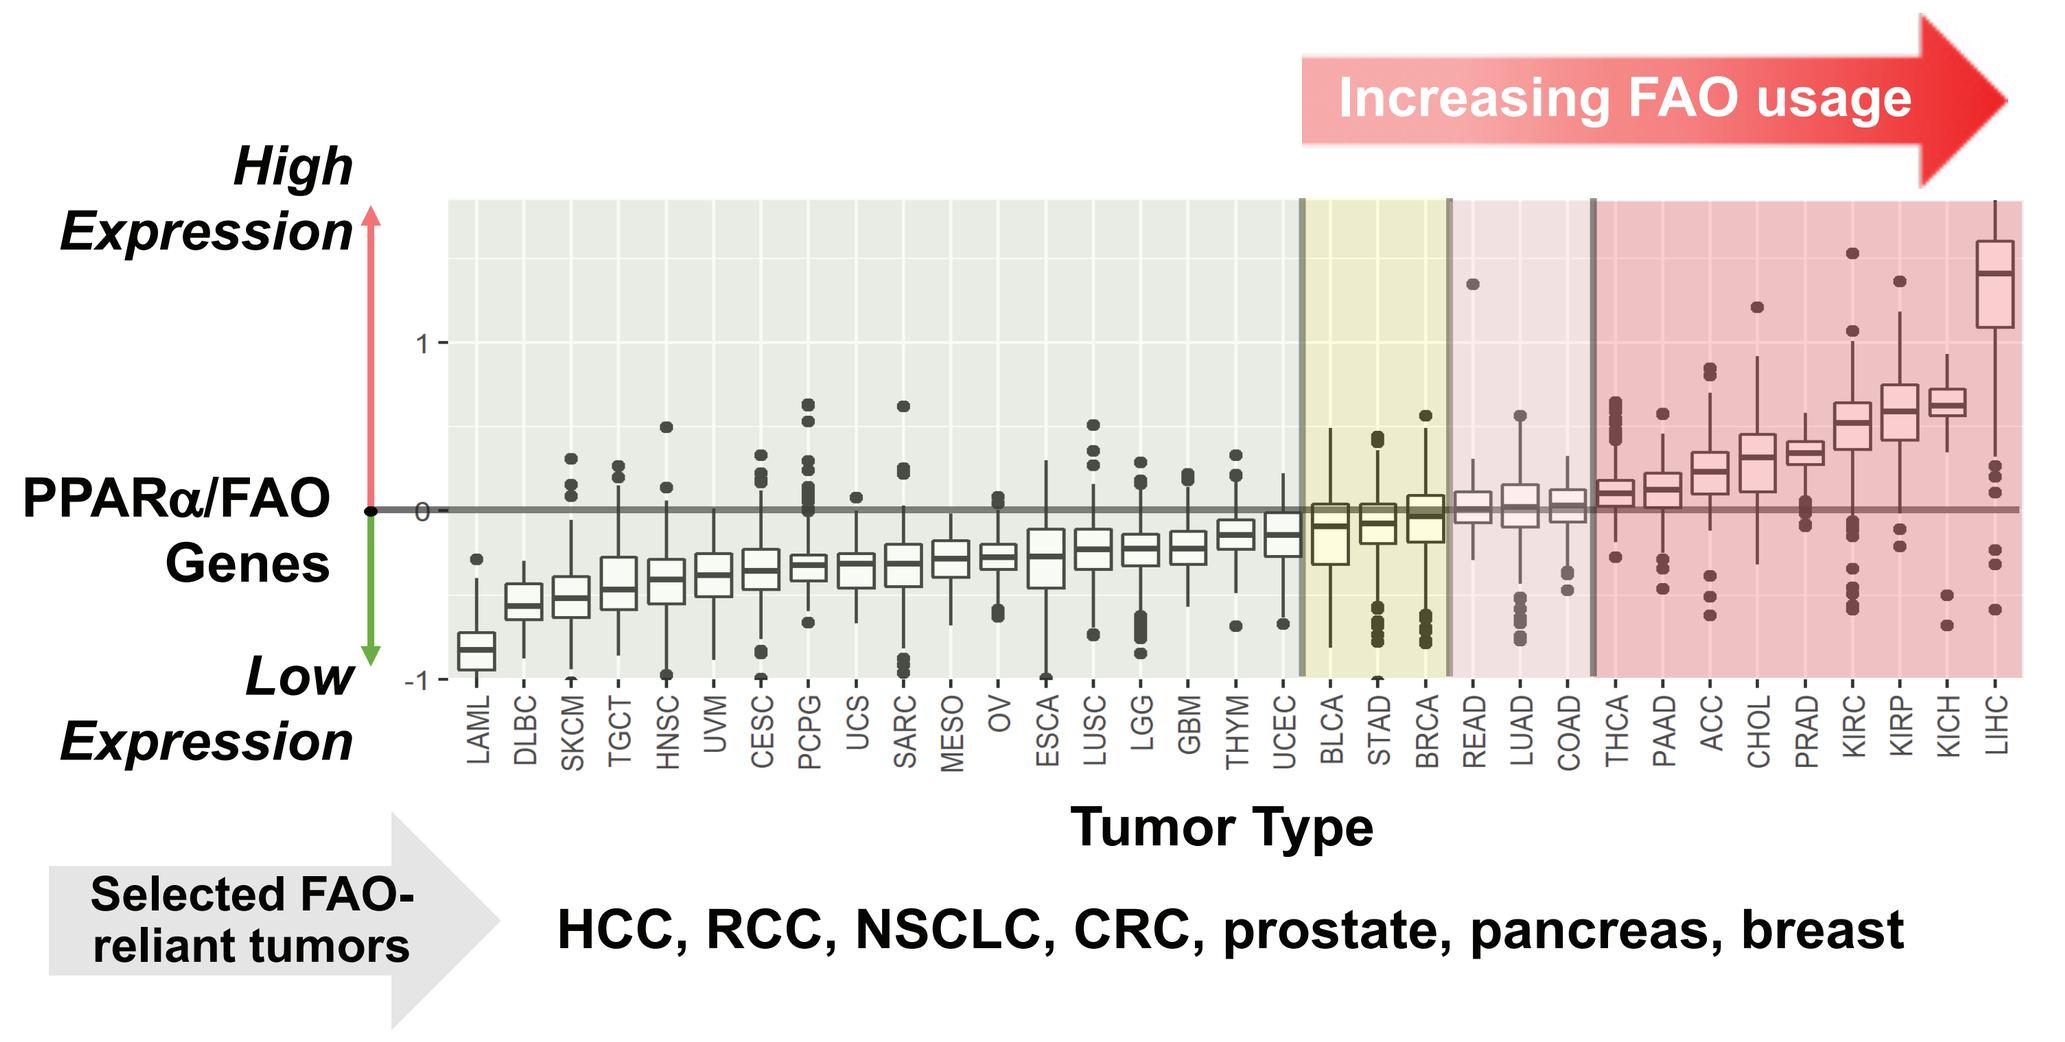
Increasing FAO usage
High Expression
PPARα/FAO Genes
Low Expression
LAML DLBC SKCM TGCT HNSC UVM CESC PCPG UCS SARC MESO OV ESCA LUSC LGG GBM THYM UCEC BLCA STAD BRCA READ LUAD COAD THCA PAAD ACC CHOL PRAD KIRC KIRP KICH LIHC
Tumor Type
Selected FAO-reliant tumors
HCC, RCC, NSCLC, CRC, prostate, pancreas, breast

TCGA: Cancer Genome Atlas

Figure 3: Selected tumor types are FAO reliant

FIGURE 4a: Single Agent Therapeutic Anti-Tumor Efficacy

TPST-1120 single agent activity in various syngeneic mouse models

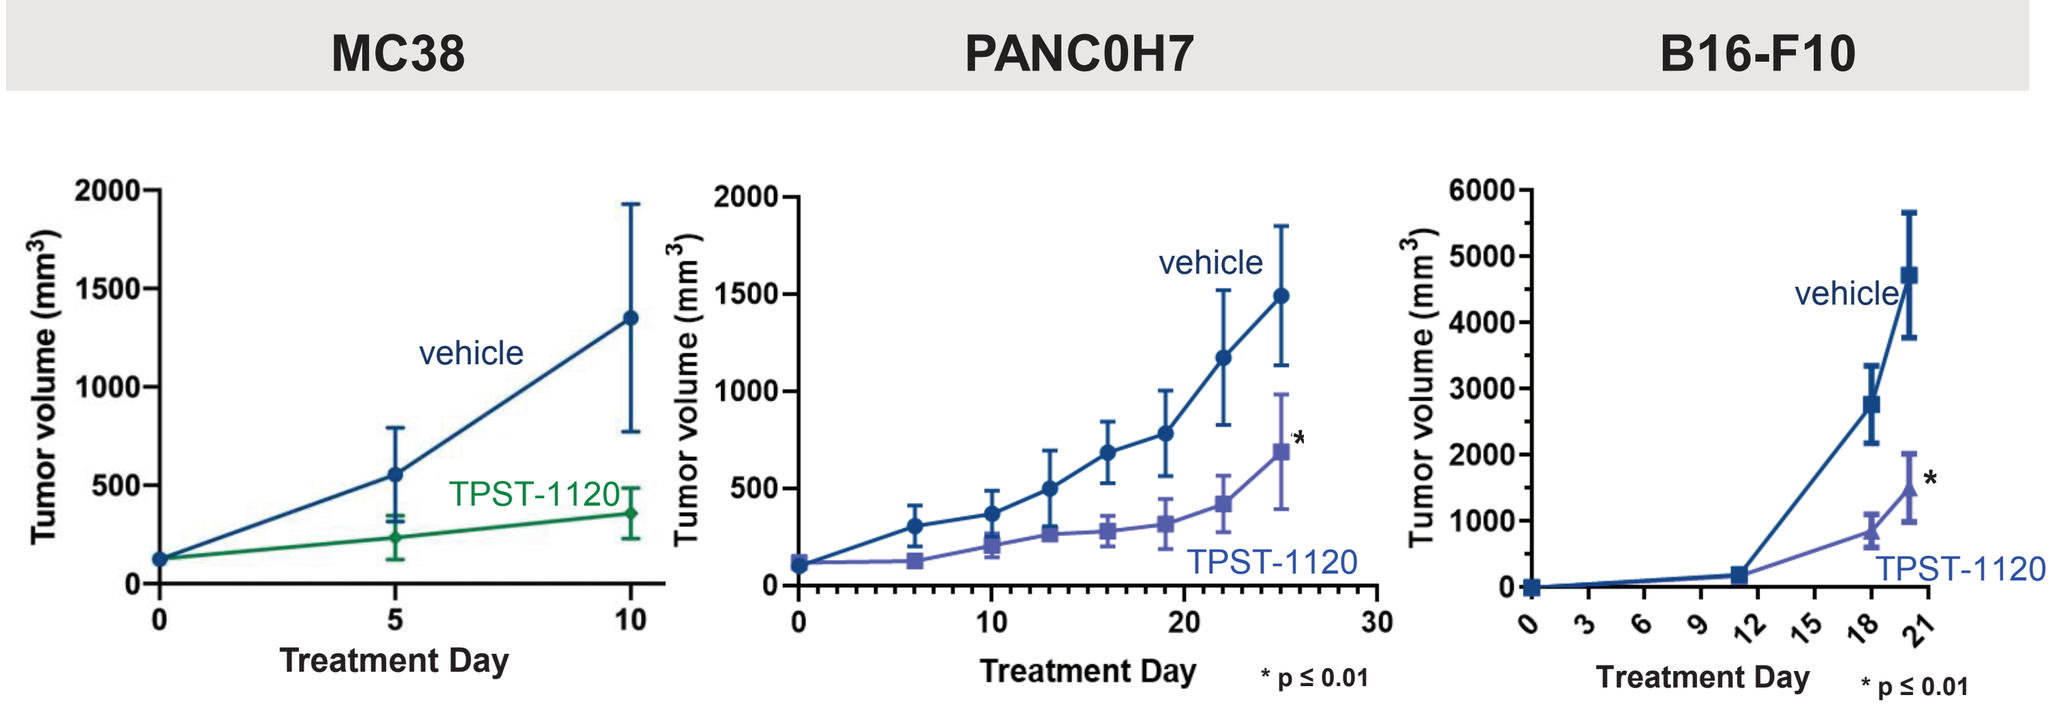
MC38
PANC0H7
B16-F10
Tumor volume (mm³)
vehicle
TPST-1120
Treatment Day
* p ≤ 0.01

FIGURE 4b: Significant Efficacy in HCC Mouse Model

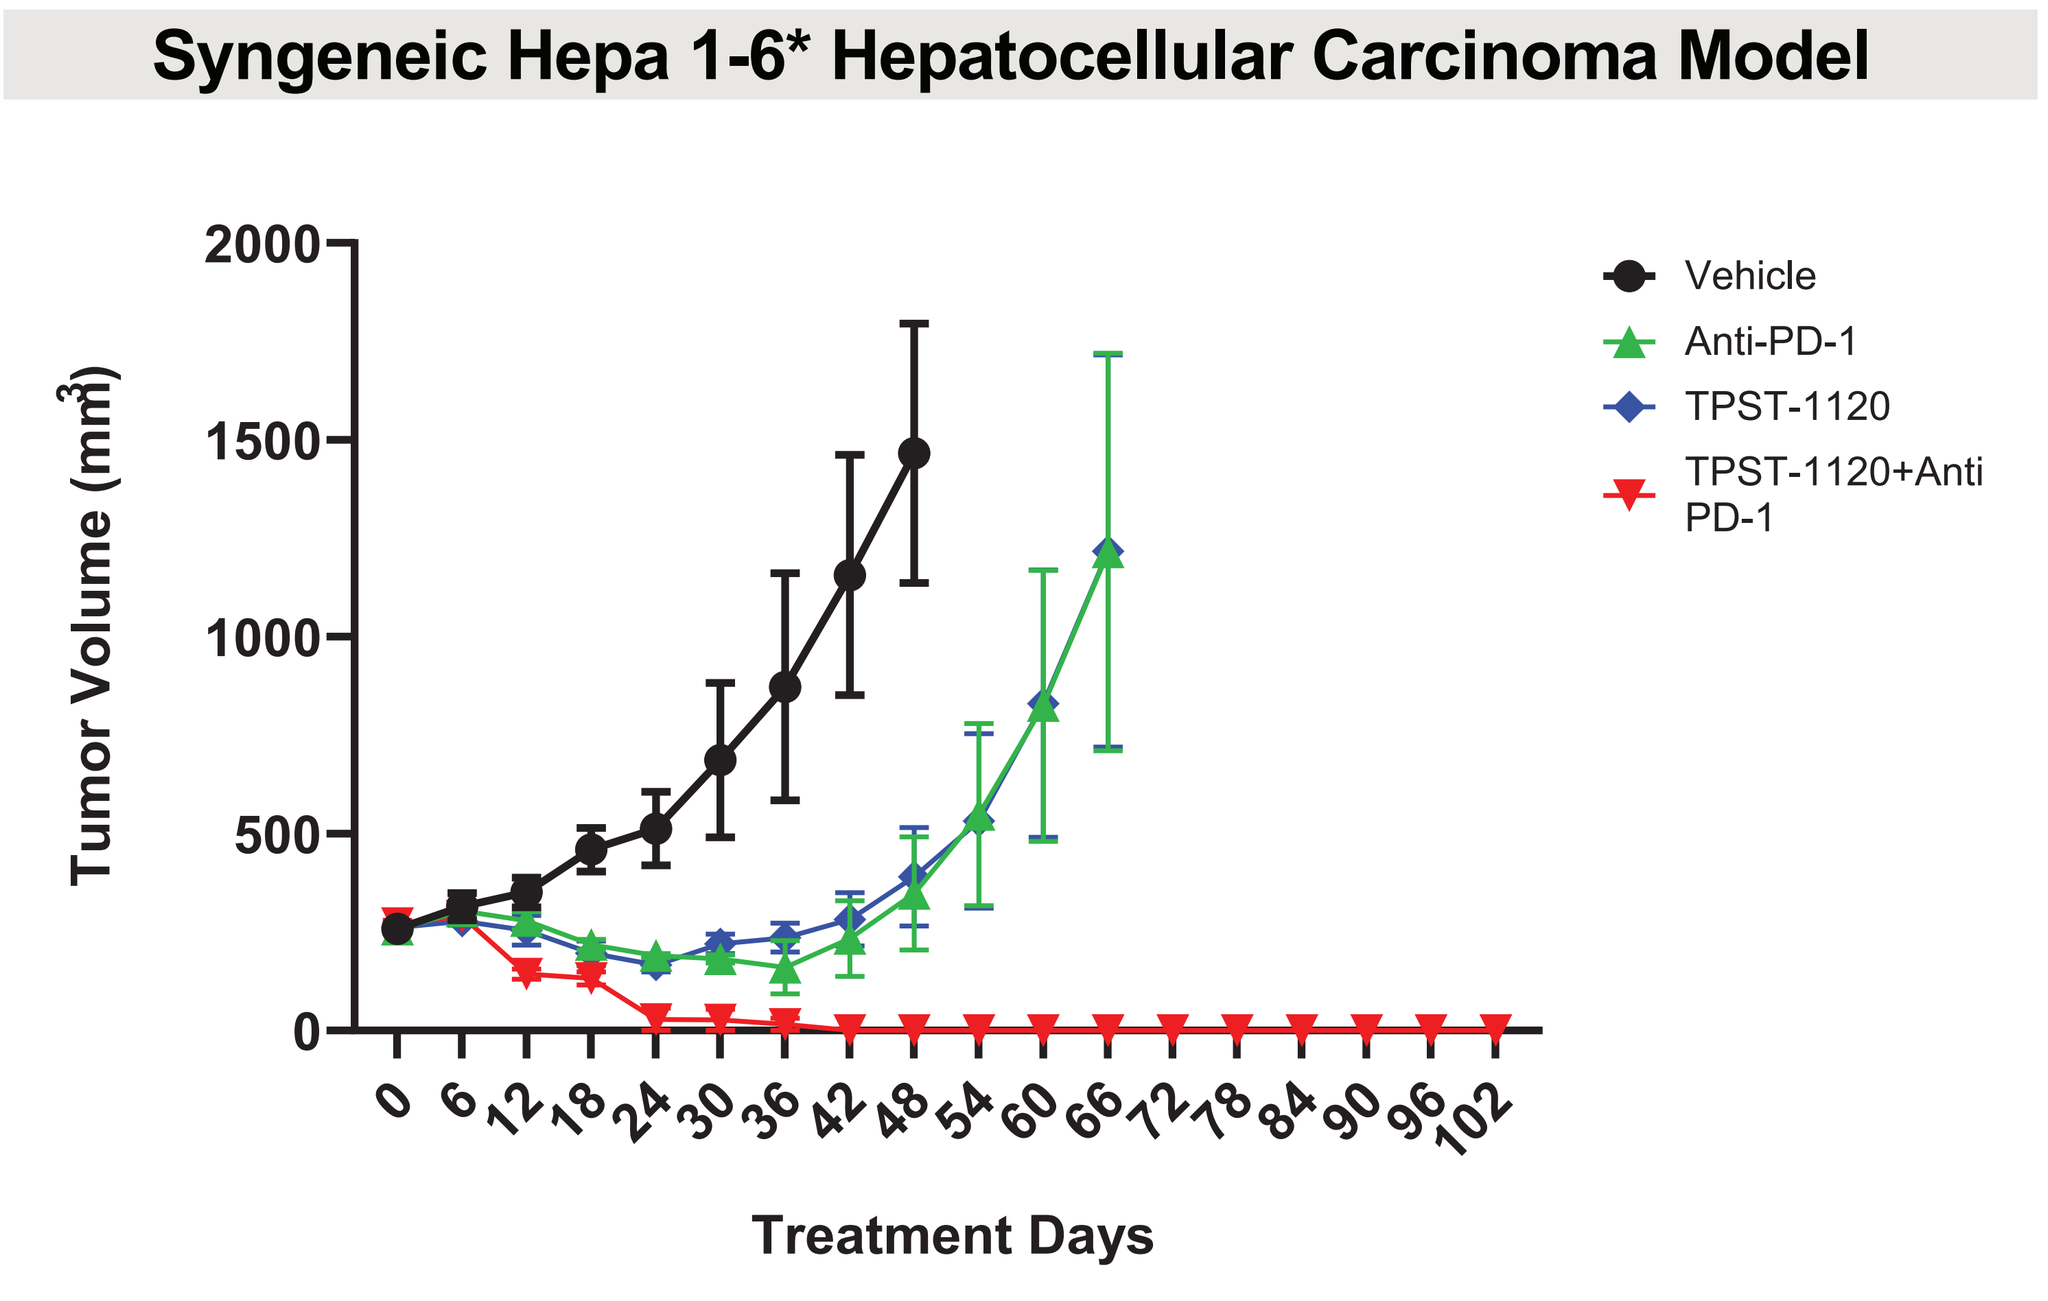
Syngeneic Hepa 1-6* Hepatocellular Carcinoma Model
Tumor Volume (mm³)
Vehicle
Anti-PD-1
TPST-1120
TPST-1120+Anti PD-1
Treatment Days

*Hepa 1-6 tumor cells are β-catenin dependent (*Pandit et al. BMC Cancer (2018) 18:783*)

Figure 4: (a) TPST-1120 has single agent activity in syngeneic mouse models. MC38 (colon), PANC0H7 (pancreatic), B16-F10 (melanoma) (data on file). (b) Efficacy of TPST-1120 alone and with anti-PD1 in HCC model. Hepa 1-6 tumor cells are β-catenin dependent (Pandit et al. BMC Cancer (2018) 18:783). Treatment initiated 12d post implantation; TPST-1120 30mg/kg BID; α-PD-1 200 µg Q3d. Source: Dipak Panigrahy collaboration (data on file).

Strong scientific rationale for TPST-1120 in Hepatocellular Carcinoma

- **High PPARα gene expression**
- **Tumor resistance to TKIs shows increasing reliance on FAO**
- **FAO is main energy source in β-catenin-induced HCC *(CTNNB1* subtype)**[1]
 - FAO inhibition eliminates tumor initiation and slows progression in preclinical *CTNNB1* HCC models
 - Prevalence of *CTNNB1* subtype: 19-27%[2,3]
 - ***CTNNB1:*** potential biomarker to select for HCC responsive to FAO inhibition

[1]Senni et al. Gut 2019;68(2):322-334
[2]Gao et al. Cell 2019;179(2):561-577
[2]TCGA. Cell 2017;169(7):1327-1341

PHASE 1/1B DOSE ESCALATION/ DOSE EXPANSION TRIAL DESIGN

- **Dose Escalation**
 - One monotherapy arm
 - Three combination arms in which TPST-1120 is combined with nivolumab, docetaxel or cetuximab
- **Dose Expansion**
 - Eight histology-specific cohorts following 2-stage expansion design
 - 4 monotherapy arms
 - 4 combination arms

FIGURE 5: TPST-1120-001 Trial Design: MONOTHERAPY

Traditional 3+3 Design

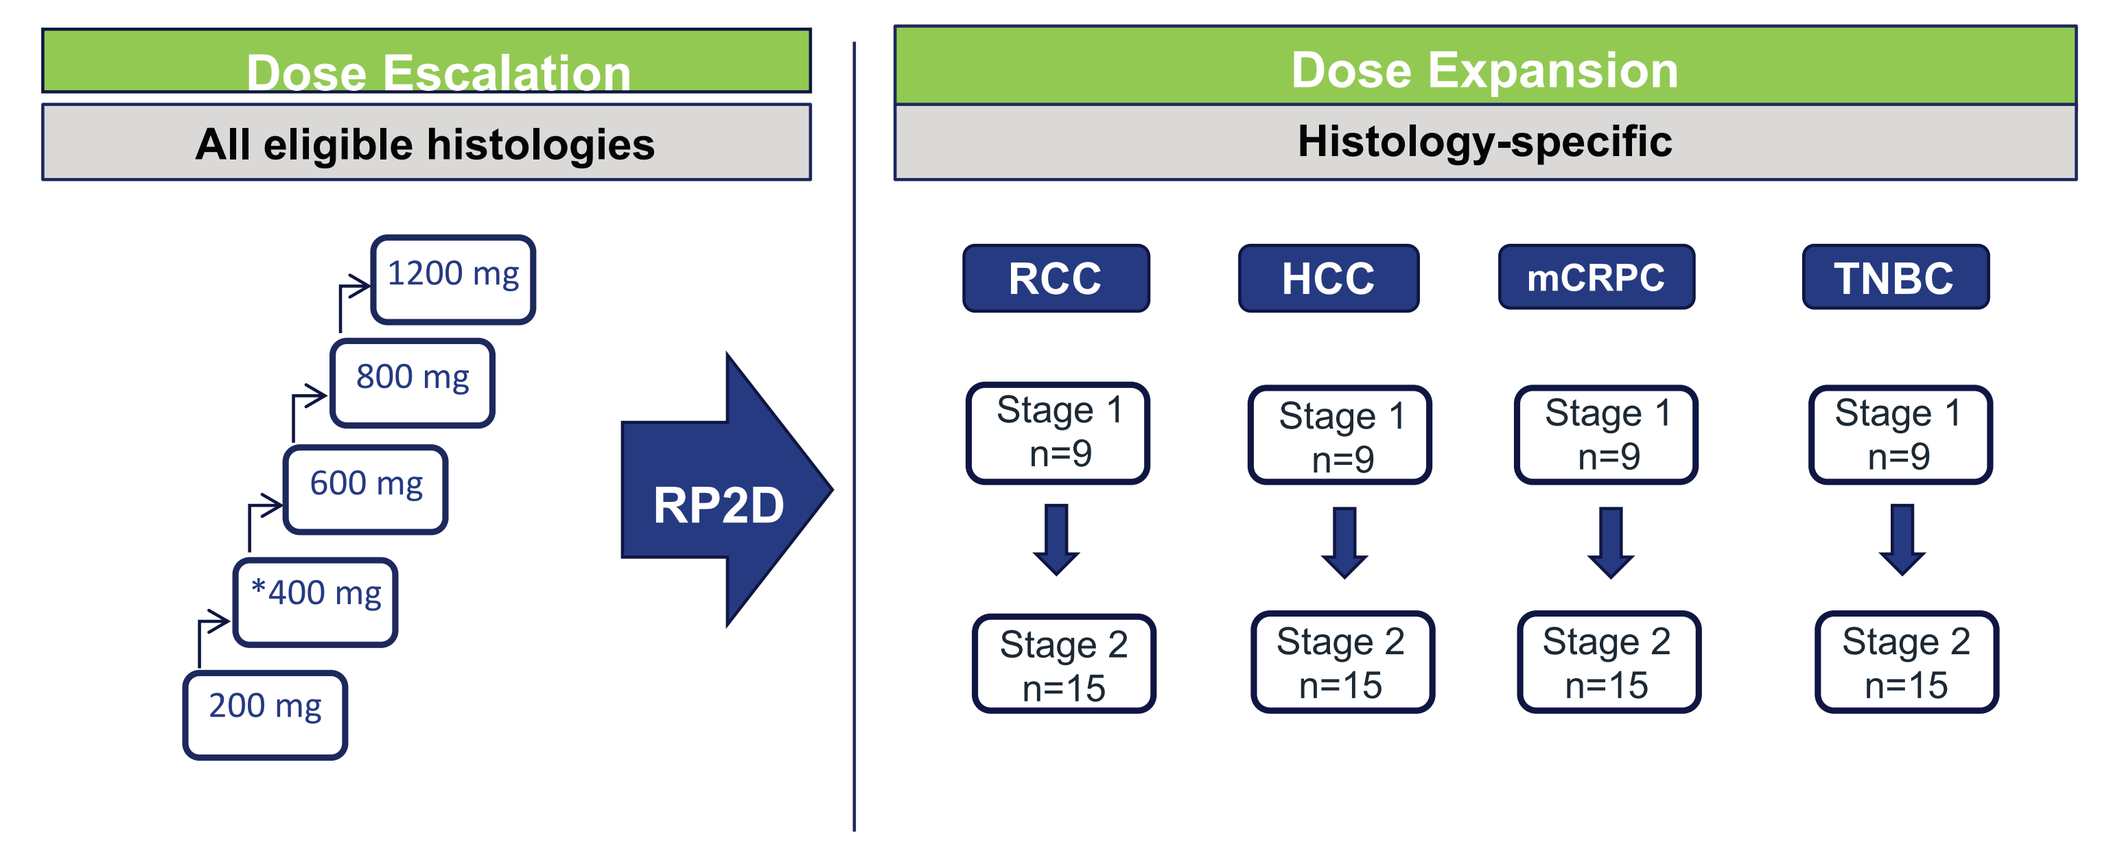
Dose Escalation
All eligible histologies
1200 mg
800 mg
600 mg
*400 mg
200 mg
RP2D
Dose Expansion
Histology-specific
RCC
HCC
mCRPC
TNBC
Stage 1 n=9
Stage 2 n=15

NCT03829436

FIGURE 6: TPST-1120-001 Trial Design: COMBINATION ARMS

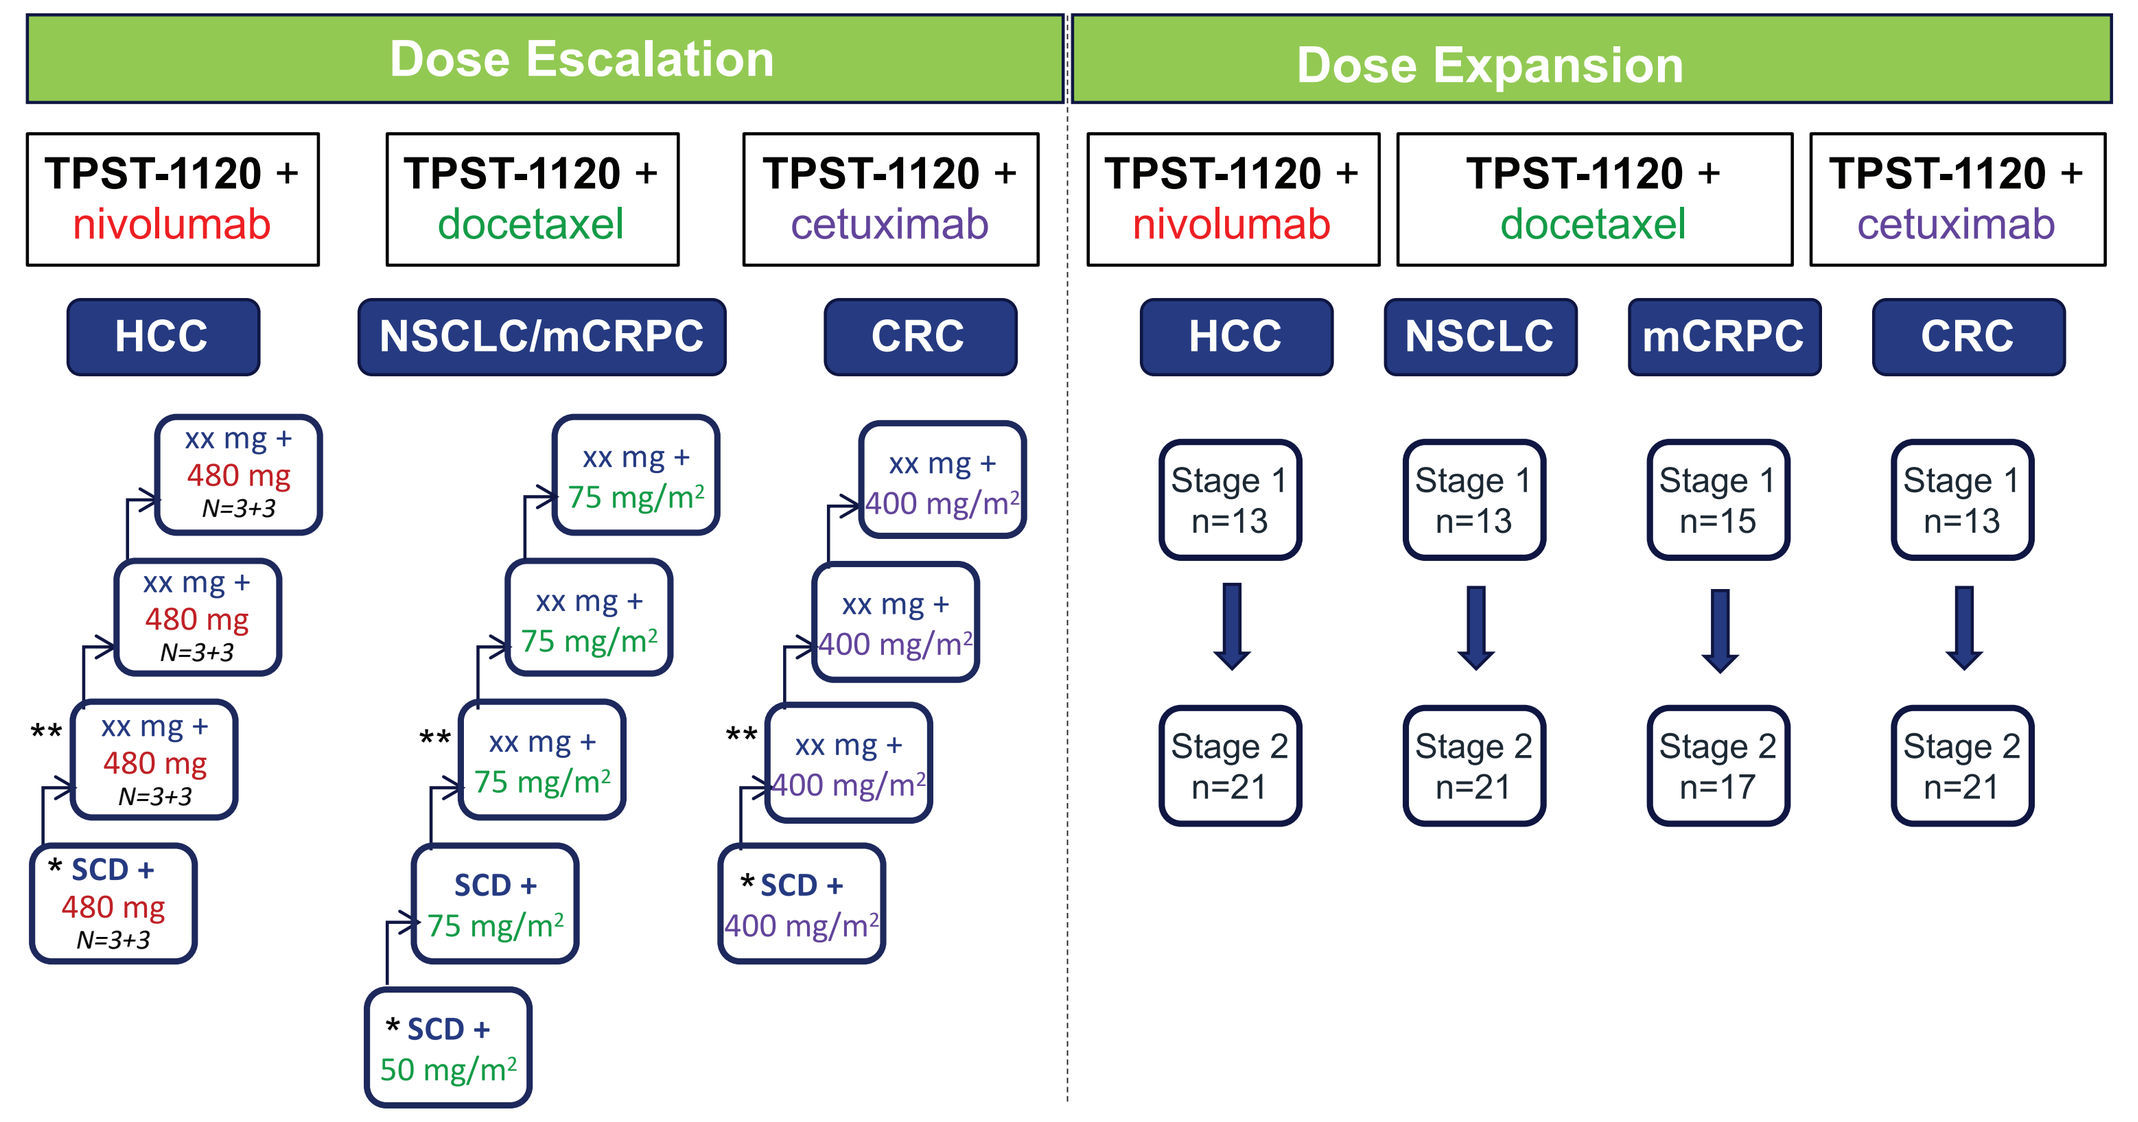
Dose Escalation
TPST-1120 + nivolumab
TPST-1120 + docetaxel
TPST-1120 + cetuximab
HCC
NSCLC/mCRPC
CRC
xx mg + 480 mg N=3+3
* SCD + 480 mg N=3+3
xx mg + 75 mg/m²
SCD + 75 mg/m²
* SCD + 50 mg/m²
xx mg + 400 mg/m²
* SCD + 400 mg/m²
Dose Expansion
TPST-1120 + nivolumab
TPST-1120 + docetaxel
TPST-1120 + cetuximab
HCC
NSCLC
mCRPC
CRC
Stage 1 n=13
Stage 1 n=15
Stage 2 n=21
Stage 2 n=17

Figure 6: **SCD** (starting combination dose) of TPST-1120 will be **400 mg** if the 600 mg dose level is cleared. However, this dose may be lower if MTD in monotherapy arm is lower than 600 mg. **Subsequent doses of TPST-1120 in combination arms will depend on SCD determined in TPST-1120 monotherapy arm

OBJECTIVES

- **Primary**
 - To evaluate safety/tolerability of TPST-1120 as monotherapy and in combination with systemic anti-cancer therapies
 - To determine MTD and/or optimal biologic dose of TPST-1120 as monotherapy and in combination with systemic anti-cancer therapies
- **Secondary**
 - Characterize the PK of TPST-1120
 - To evaluate anti-tumor activity of TPST-1120 as monotherapy and in combination with systemic anti-cancer therapies
 - Explore effects of TPST-1120 on immune function and PPARα gene expression

ELIGIBILITY

- **ECOG performance status of 0 or 1**
- **Life expectancy > 12 weeks**
- **Adequate organ function**
- **Must have received at least 1 but nor more than 4-5 prior regimens for advanced or recurrent disease**
- **Eligible histologies**
 - Mono Dose Escalation: RCC, HCC, mCRPC, TNBC, pancreatic, cholangiocarcinoma, sarcoma, UBC, NSCLC, MSS CRC, Gastroesophageal, SCCHN
 - Mono Dose Expansion: RCC, HCC, mCRPC, TNBC
 - Combo Esc/Exp: HCC, NSCLC, MSS CRC
- **Prior PD1/L1 or CTLA-4 agents allowed**
- **Paired tumor biopsies required**

BIOMARKER PLAN

- **Peripheral blood**
 - Gene expression profiling of PPARα-associated genes and immune activation genes
 - Serum lipid profiling
- **Tumor**
 - Gene expression profiling of PPARα-associated genes and immune activation genes
 - IHC for CD8+ tumor-infiltrating lymphocytes

PROTOCOL UPDATE

- Accrual commenced in May 2019 at U.S. sites
- Accrual continues in monotherapy dose escalation cohorts
- Expansion cohorts projected to open in early 2020