Antagonism of Peroxisome Proliferator Activated Receptor Alpha (PPARα) by TPST-1120 Suppresses Tumor Growth and Stimulates Anti-Tumor Immunity

Chan C. Whiting[1], Nick Stock[2], Davorka Messmer[2], Austin Chen[2], Traci Olafson[2], Allison Gartung[4], Karin Stebbins[2], Lisa Rahbaek[2], Catherine Lee[2], Chris Baccei[2], Alex Broadhead[2], Ryan Clark[2], Dan Lorrain[2], Alicia Levey[1], Derek Metzger[1], Amanda Enstrom[1], Jennifer McDevitt[1], David Spaner[3], Peppi Prasit[2], Dipak Panigrahy[4]

[1]Tempest Therapeutics, Inc., San Francisco, CA; [2]Inception Sciences, San Diego, CA; [3]Sunnybrook, Toronto, Canada; [4]Beth Israel Deaconess Medical Center, Harvard Medical School, Boston, MA

TEMPEST THERAPEUTICS

ABSTRACT

Background

TPST-1120 is a first-in-class selective antagonist of human PPARα, a transcription factor that induces expression of fatty acid oxidation (FAO) genes. Tumor metabolic adaptions promote its own survival and suppress tumor-specific immunity by upregulation of FAO. PPARα blockade has tumor intrinsic and extrinsic effects, and inhibits tumor cell growth and induces tumor-specific immunity, demonstrated among multiple syngeneic and xenograft mouse models.

Methods

TPST-1120 efficacy as a monotherapy or in combination with chemotherapy or anti-PD1 was evaluated in multiple syngeneic mouse models including B16 melanoma, MMTV mammary carcinoma, MC38 colon, Lewis lung carcinoma, ID8 ovarian, Panc07 pancreatic cancer, and xenograft models of CLL, melanoma, pancreatic and AML. To characterize its mechanism of anti-tumor immunity, TPST-1120 was evaluated in knock-out models of CCL2, MBL, TSP-1, STING and BatF3. Immune modulation was characterized by M2/M1 macrophage flow cytometry phenotyping and ELISA measurement of plasma and tumor matrix protein thrombospondin-1 (TSP-1), which is involved in granulocyte migration and angiogenesis.

Results

TPST-1120 mediated PPARα antagonism resulted in potent anti-tumor immune responses and significant tumor regression, either as a monotherapy or in combination with chemotherapy or anti-PD1. TPST-1120 showed anti-tumor efficacy against syngeneic models of breast, lung, colon, pancreatic and melanoma in addition to xenograft models of CLL, AML, pancreatic and melanoma cancers as a monotherapy or in combination with chemotherapy and anti-PD1. TPST-1120 demonstrated cytotoxic effect on tumor cells *in vitro.* In a pancreatic and breast cancer model, TPST-1120 combination with chemotherapies gemcitabine and eribulin, respectively, had additive effect on tumor growth. TPST-1120 combination with anti-PD1 in an ovarian orthotopic (ID8) and colon (MC38) models showed suppression of tumor growth and complete remission in some mice. Moreover, mice receiving the combination treatment conferred protection against autologous tumor re-challenge in the ID8 model, strongly suggesting immunological T cell memory against the primary tumor. Preliminary studies in genetic knock-out mice, suggest macrophages and antigen cross-presenting dendritic cells are required for TPST-1120 activity, potentially through STING activation and TSP-1. Consistent with prior reports of the involvement of PPARa activation in promoting M2 macrophages, TPST-1120 skews toward an M1 effector macrophage phenotype and in vivo pretreated peritoneal macrophages enhance the uptake of whole tumor cells by FACS.

Conclusions

Through its unique mechanism of restricting FAO, TPST-1120 targets a metabolic pathway that is critical for the survival of both tumor cells and of suppressive immune cell populations infiltrating the tumor microenvironment. TPST-1120 represents a promising new approach for evaluation in patients with advanced malignancies.

INTRODUCTION

- TPST-1120 is a first-in-class, orally administered, small molecule selective antagonist of the human peroxisome proliferator-activated receptor alpha (PPARα)
- PPARα is a transcription factor which induces the expression of genes that regulate fatty acid oxidation (FAO) and inflammation (Figure 2)
- PPARα and FAO gene signatures can be enriched in metastatic tumors[4-5] (Figure 1)
- FAO supports the metabolism of immune suppressive cell populations that inhibit anti-tumor immunity[1-3]
- The growth and progression of diverse tumor types are completely suppressed in PPARα-deficient mice[6-7]
- TPST-1120 has significant anti-tumor activity as a monotherapy and in combination with chemotherapy or anti-PD1 antibodies
- Anti-tumor activity of TPST-1120 is mediated through: 1) direct tumor cell killing; 2) release of immune suppression; and 3) restoration of TSP-1 to homeostatic levels

FIGURE 1: Upregulation of PPARα and FAO Genes in Diverse Tumors

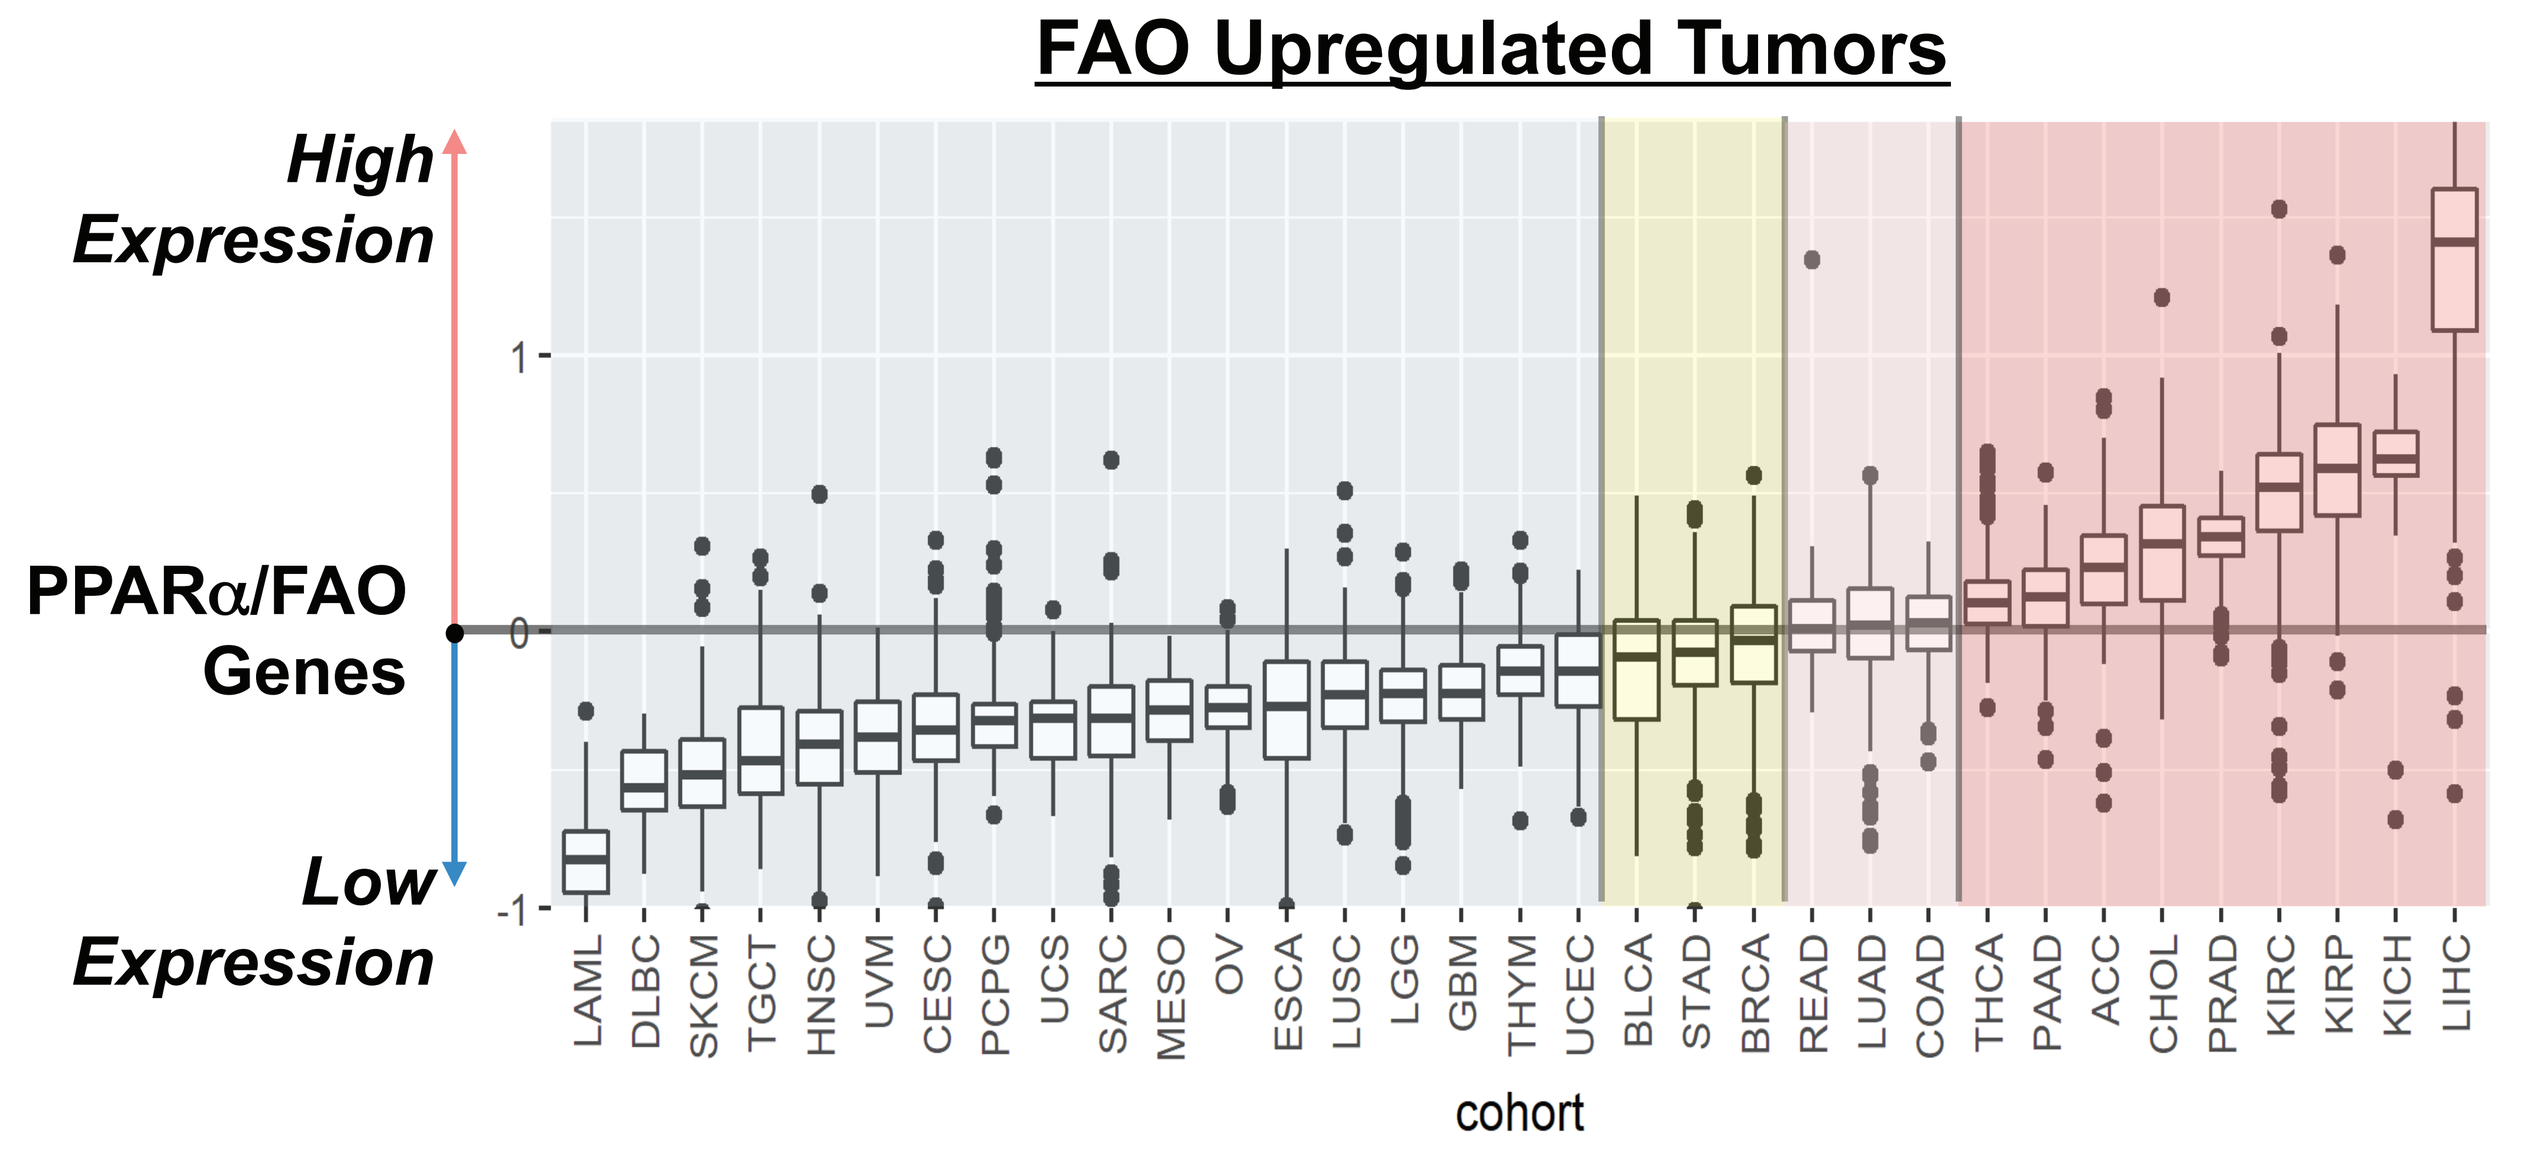
FAO Upregulated Tumors
High Expression
PPARα/FAO Genes
Low Expression
1
0
-1
LAML DLBC SKCM TGCT HNSC UVM CESC PCPG UCS SARC MESO OV ESCA LUSC LGG GBM THYM UCEC BLCA STAD BRCA READ LUAD COAD THCA PAAD ACC CHOL PRAD KIRC KIRP KICH LIHC
cohort

Figure 1: Enrichment of FAO associated genes including PPARA was evaluated in 10,000 unique datasets representing 33 tumor types in the TCGA (cancergenome.nih.gov).

METHODS

- Efficacy as a monotherapy or in combination with chemotherapy or anti-PD1 was evaluated in multiple syngeneic mouse models
- TPST-1120 was evaluated in knock-out models of *Tsp-1, Sting* and *Batf3* to characterize its mechanism of anti-tumor immunity
- ELISA was used to measure plasma and tumor matrix protein Tsp-1 and Fgf21
- Gene expression analysis was performed by quantitative RT-PCR

RESULTS

TPST-1120: PPARα Target Validation and Target Engagement

FIGURE 2: PPARα Induces Fatty Acid Oxidation (FAO)

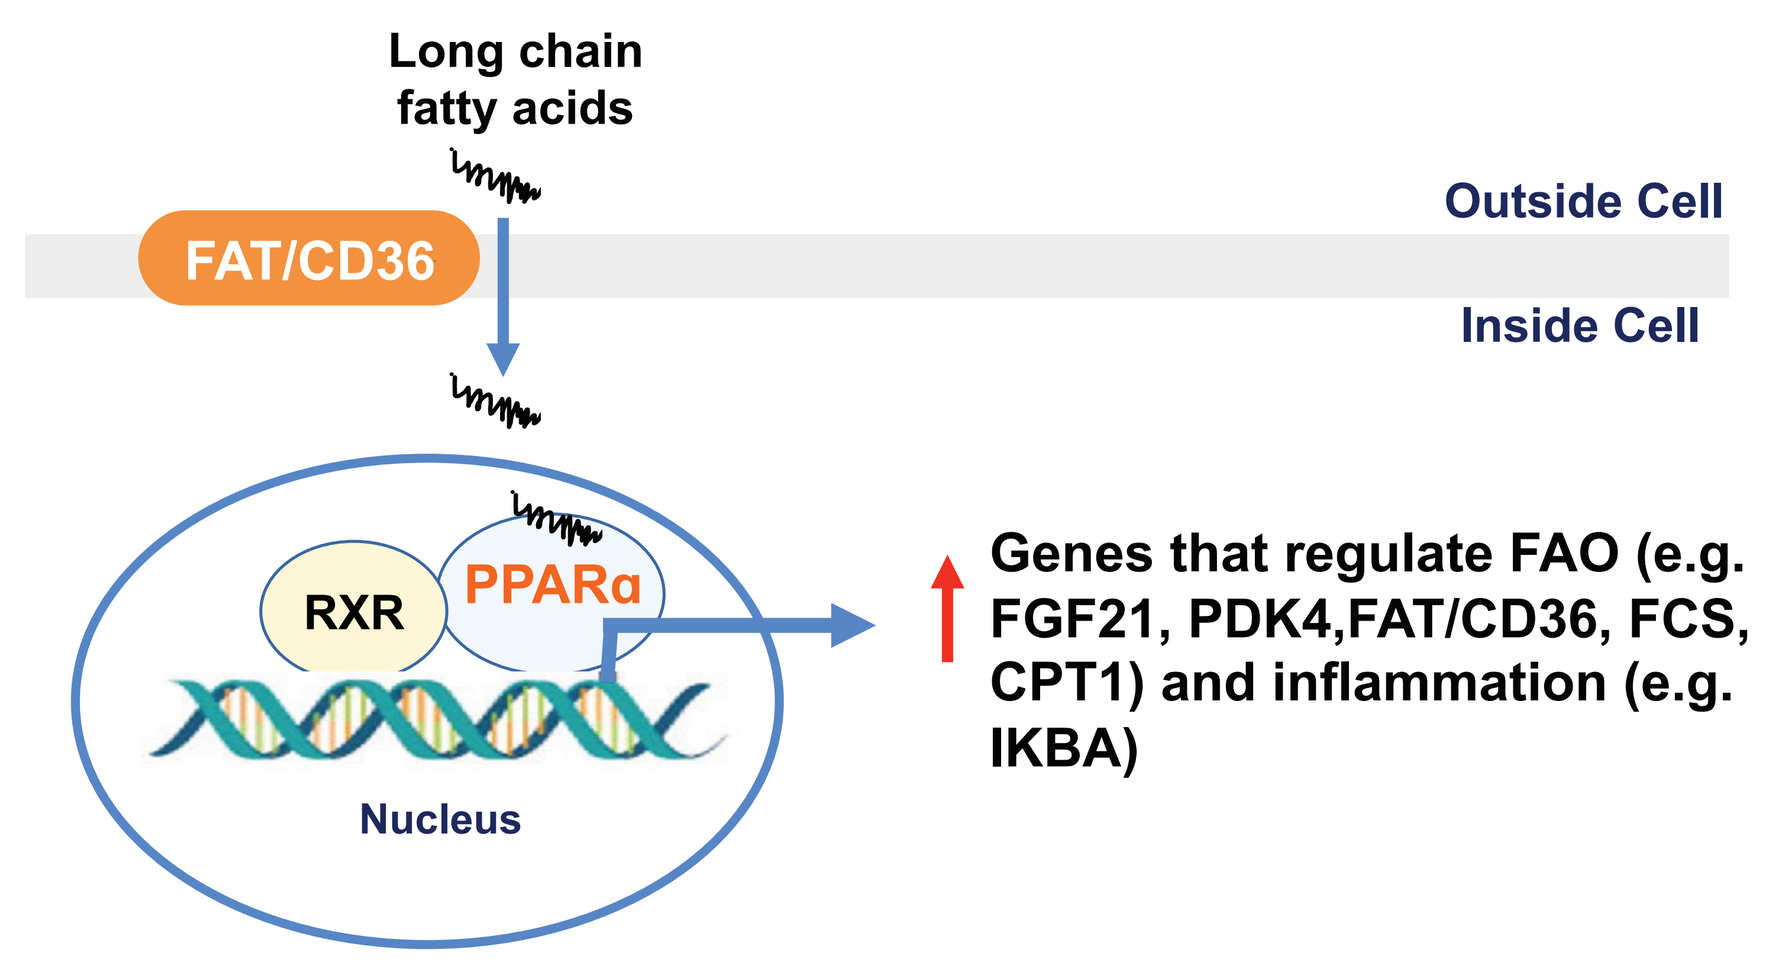
Long chain fatty acids
FAT/CD36
Outside Cell
Inside Cell
RXR
PPARα
Nucleus
Genes that regulate FAO (e.g. FGF21, PDK4,FAT/CD36, FCS, CPT1) and inflammation (e.g. IKBA)

Figure 2: PPARα regulates genes that modulate FAO and inflammation. Upon ligand binding, PPARα heterodimerizes with RXR, stimulates gene transcription of target genes that regulate FAO and negative modulators of inflammation.

FIGURE 3: Tumors Spontaneously Regress in PPARα Knock-Out Mice

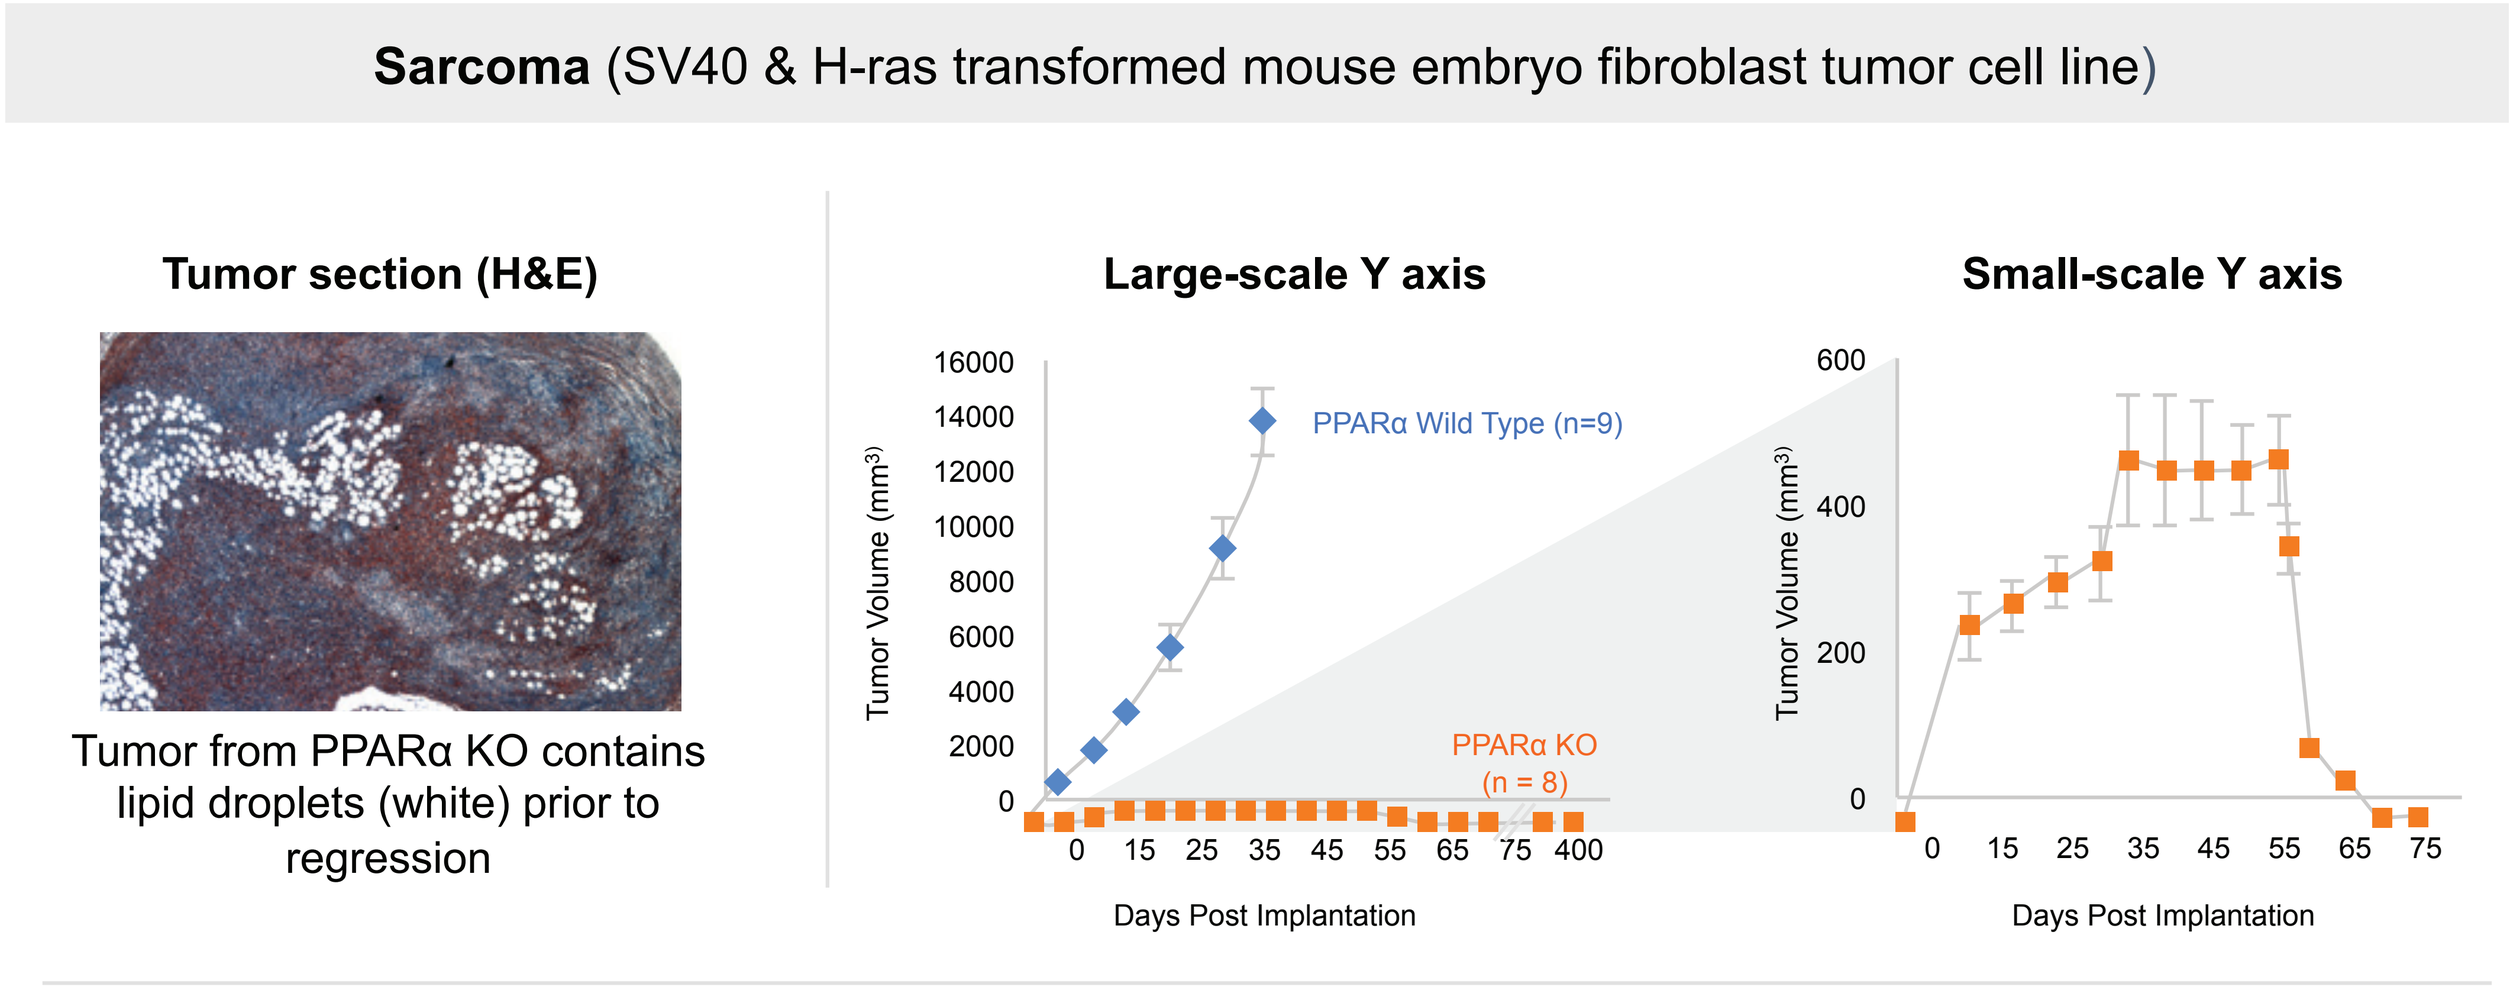
Sarcoma (SV40 & H-ras transformed mouse embryo fibroblast tumor cell line)
Tumor section (H&E)
Tumor from PPARα KO contains lipid droplets (white) prior to regression
Large-scale Y axis
Small-scale Y axis
Tumor Volume (mm3)
PPARα Wild Type (n=9)
PPARα KO (n = 8)
Days Post Implantation

Source: Kaipainen et al., PLoS ONE 2007, 2(2): e260 and unpublished results

Figure 3: Tumor growth is inhibited in PPARα knock-out (KO) mice. Tumors establish but spontaneously regress in KO mice.

FIGURE 4: PPARα Antagonist TPST-1120 Inhibits PPARα-Dependent Targets

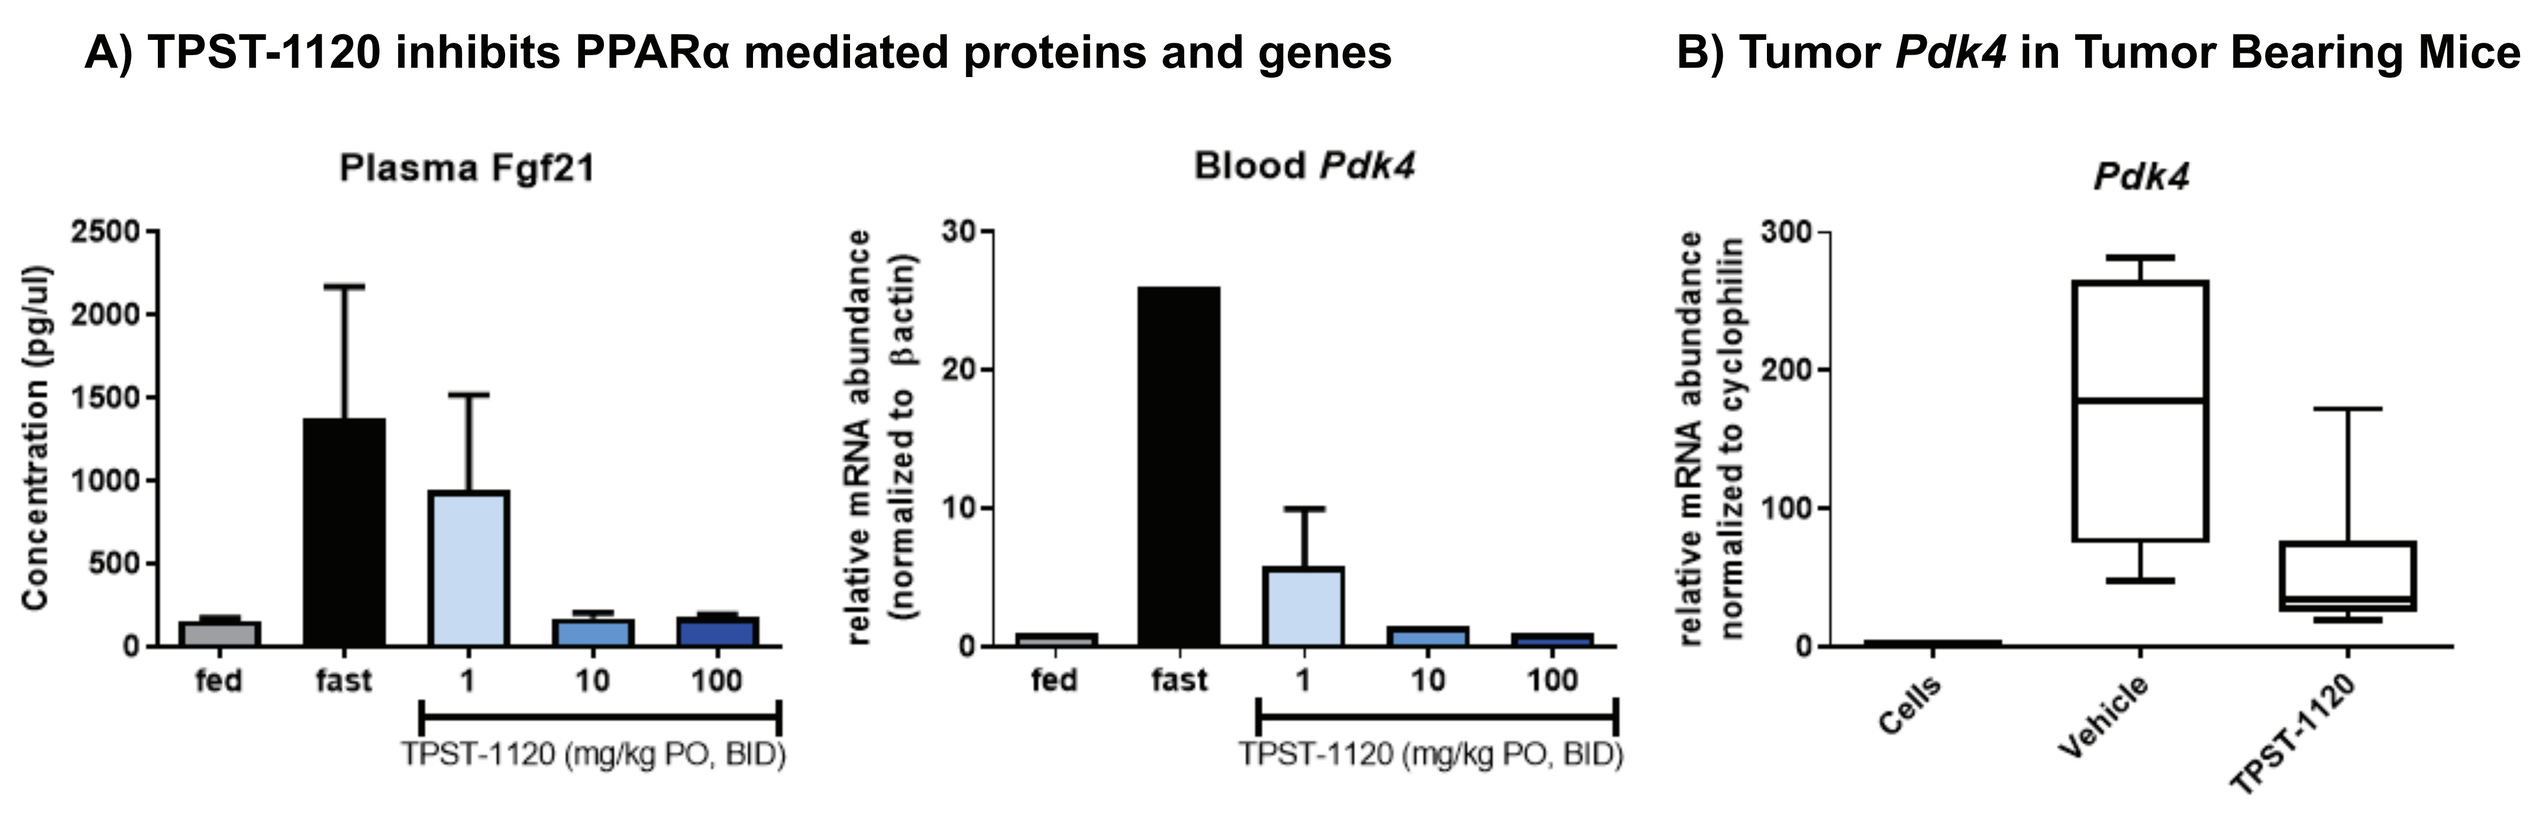
A) TPST-1120 inhibits PPARα mediated proteins and genes
Plasma Fgf21
Blood *Pdk4*
Concentration (pg/ul)
relative mRNA abundance (normalized to βactin)
fed
fast
1
10
100
TPST-1120 (mg/kg PO, BID)
B) Tumor *Pdk4* in Tumor Bearing Mice
Pdk4
relative mRNA abundance normalized to cyclophilin
Cells
Vehicle
TPST-1120

Figure 4: A.) PPARα induced Fgf21 protein and target gene *Pdk4* increases under fasting is inhibited with TPST-1120 at 1, 10, and 100 mg/kg PO BID in C57BL6/N mice (non-tumor bearing). B.) *Pdk4* gene expression in inhibited in B16-F10 melanoma tumors in mice treated with TPST-1120 30 mg/kg PO BID compared to vehicle treated.

FIGURE 5: TPST-1120 Inhibits Proliferation of Primary CLL Tumor Cells

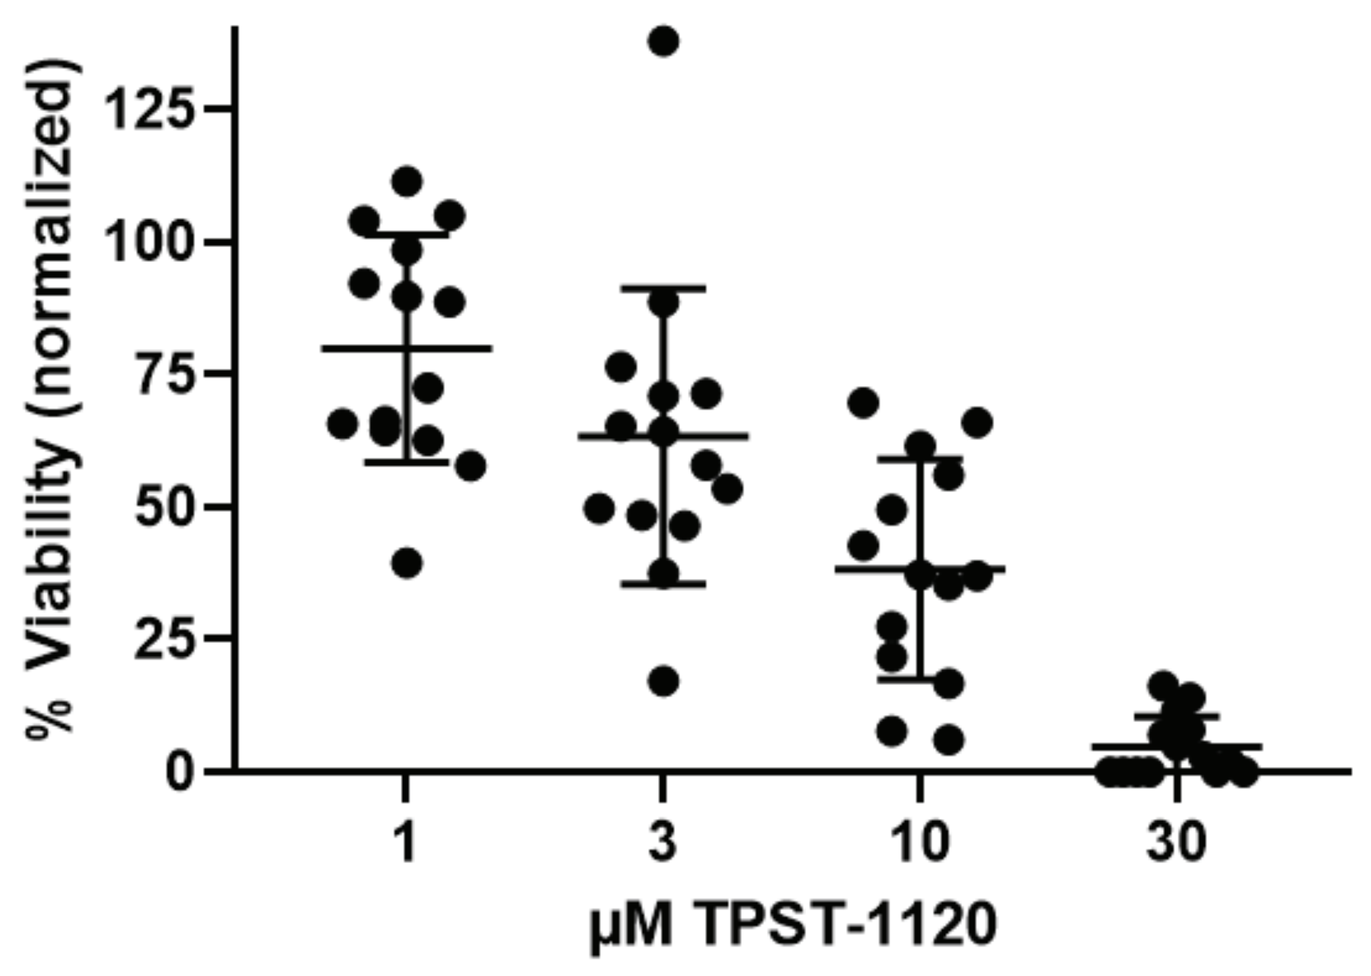
% Viability (normalized)
µM TPST-1120
1
3
10
30

Figure 5: TPST-1120 directly inhibits proliferation of primary CLL tumor cells in a dose dependent manner after 48 hours of incubation. Cells were obtained from consented patients (n=14).

FIGURE 6: TPST-110 Inhibits the Outgrowth of Syngeneic Tumors

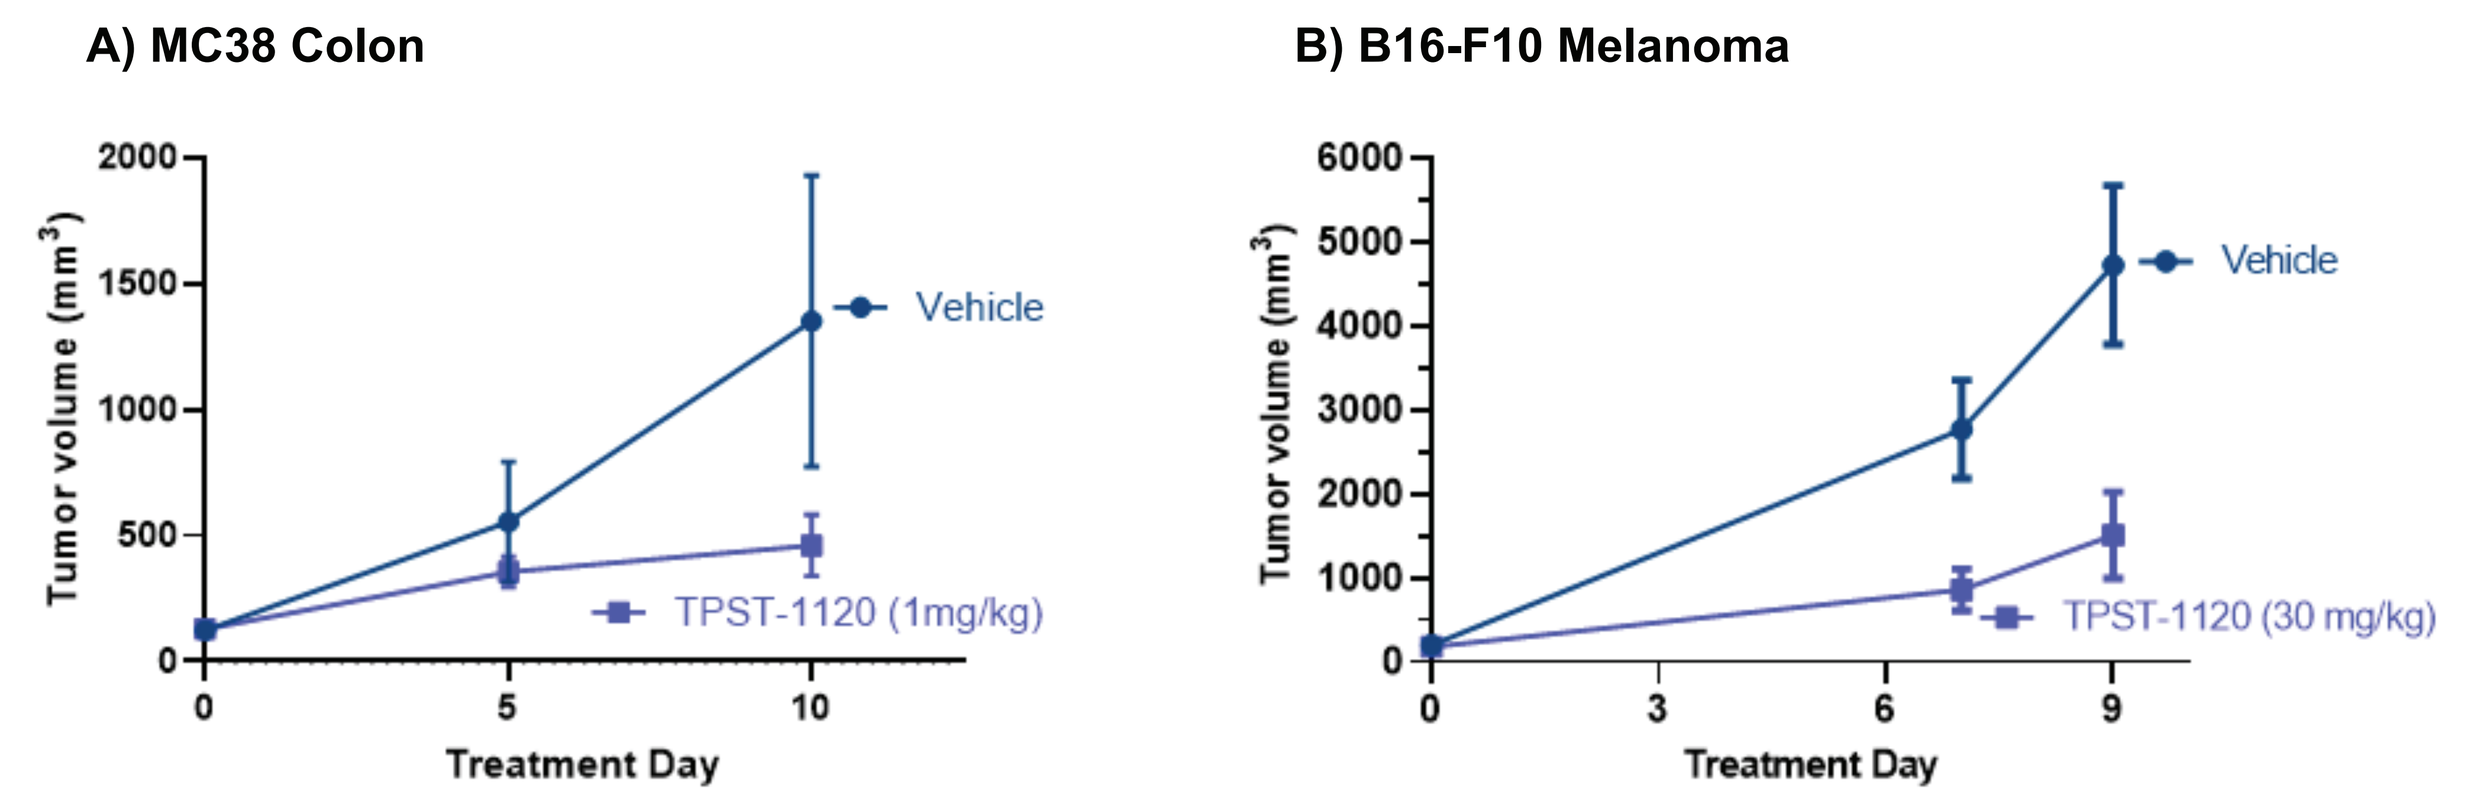
A) MC38 Colon
B) B16-F10 Melanoma
Tumor volume (mm3)
Vehicle
TPST-1120 (1mg/kg)
TPST-1120 (30 mg/kg)
Treatment Day

Figure 6: Tumor growth is suppressed compared to vehicle control by TPST-1120 in A) MC38 colon model at 1 mg/kg, and B) in B16F10 melanoma at 30 mg/kg BID PO.

FIGURE 7: TPST-1120 Significantly Enhances Benefit of Chemotherapy

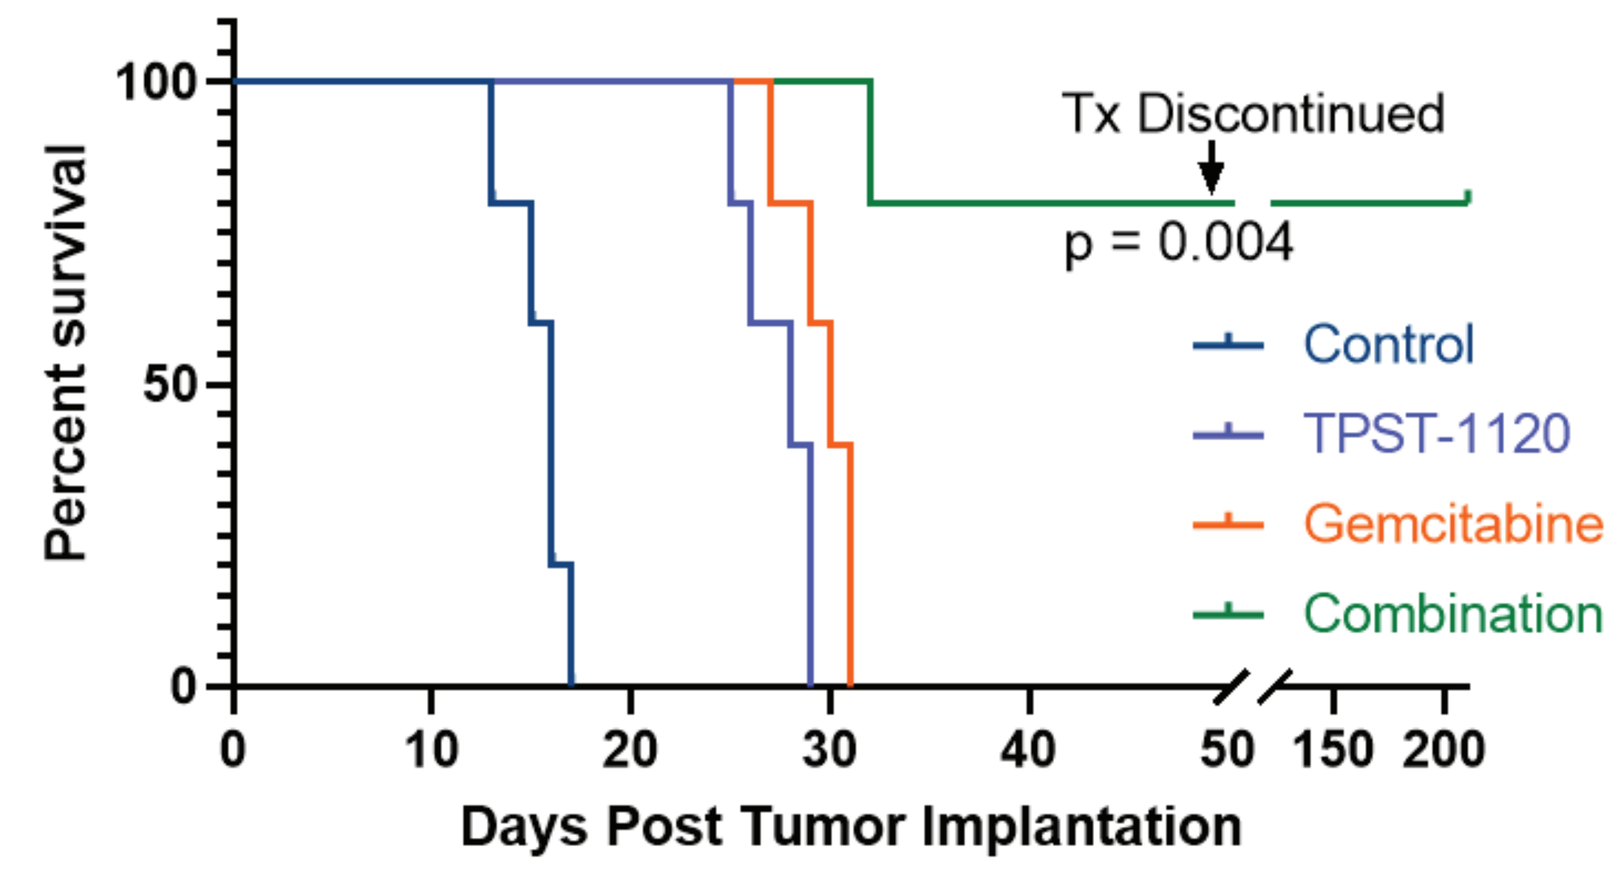
Tx Discontinued
p = 0.004
Percent survival
Control
TPST-1120
Gemcitabine
Combination
Days Post Tumor Implantation

Figure 7: TPST-1120 (30 mg/kg PO BID) response against PancOH7 tumors was improved by combination with gemcitabine 50 mg/kg IP q4d (N=5/group) in PancOH7. 4/5 mice in the combo group were alive at day 210 (treatment discontinued at day 48).

The anti-tumor effects of PPARα antagonism is dependent upon immune modulation

FIGURE 8: TPST-1120 Anti-Tumor Activity Requires Innate and Adaptive Immunity

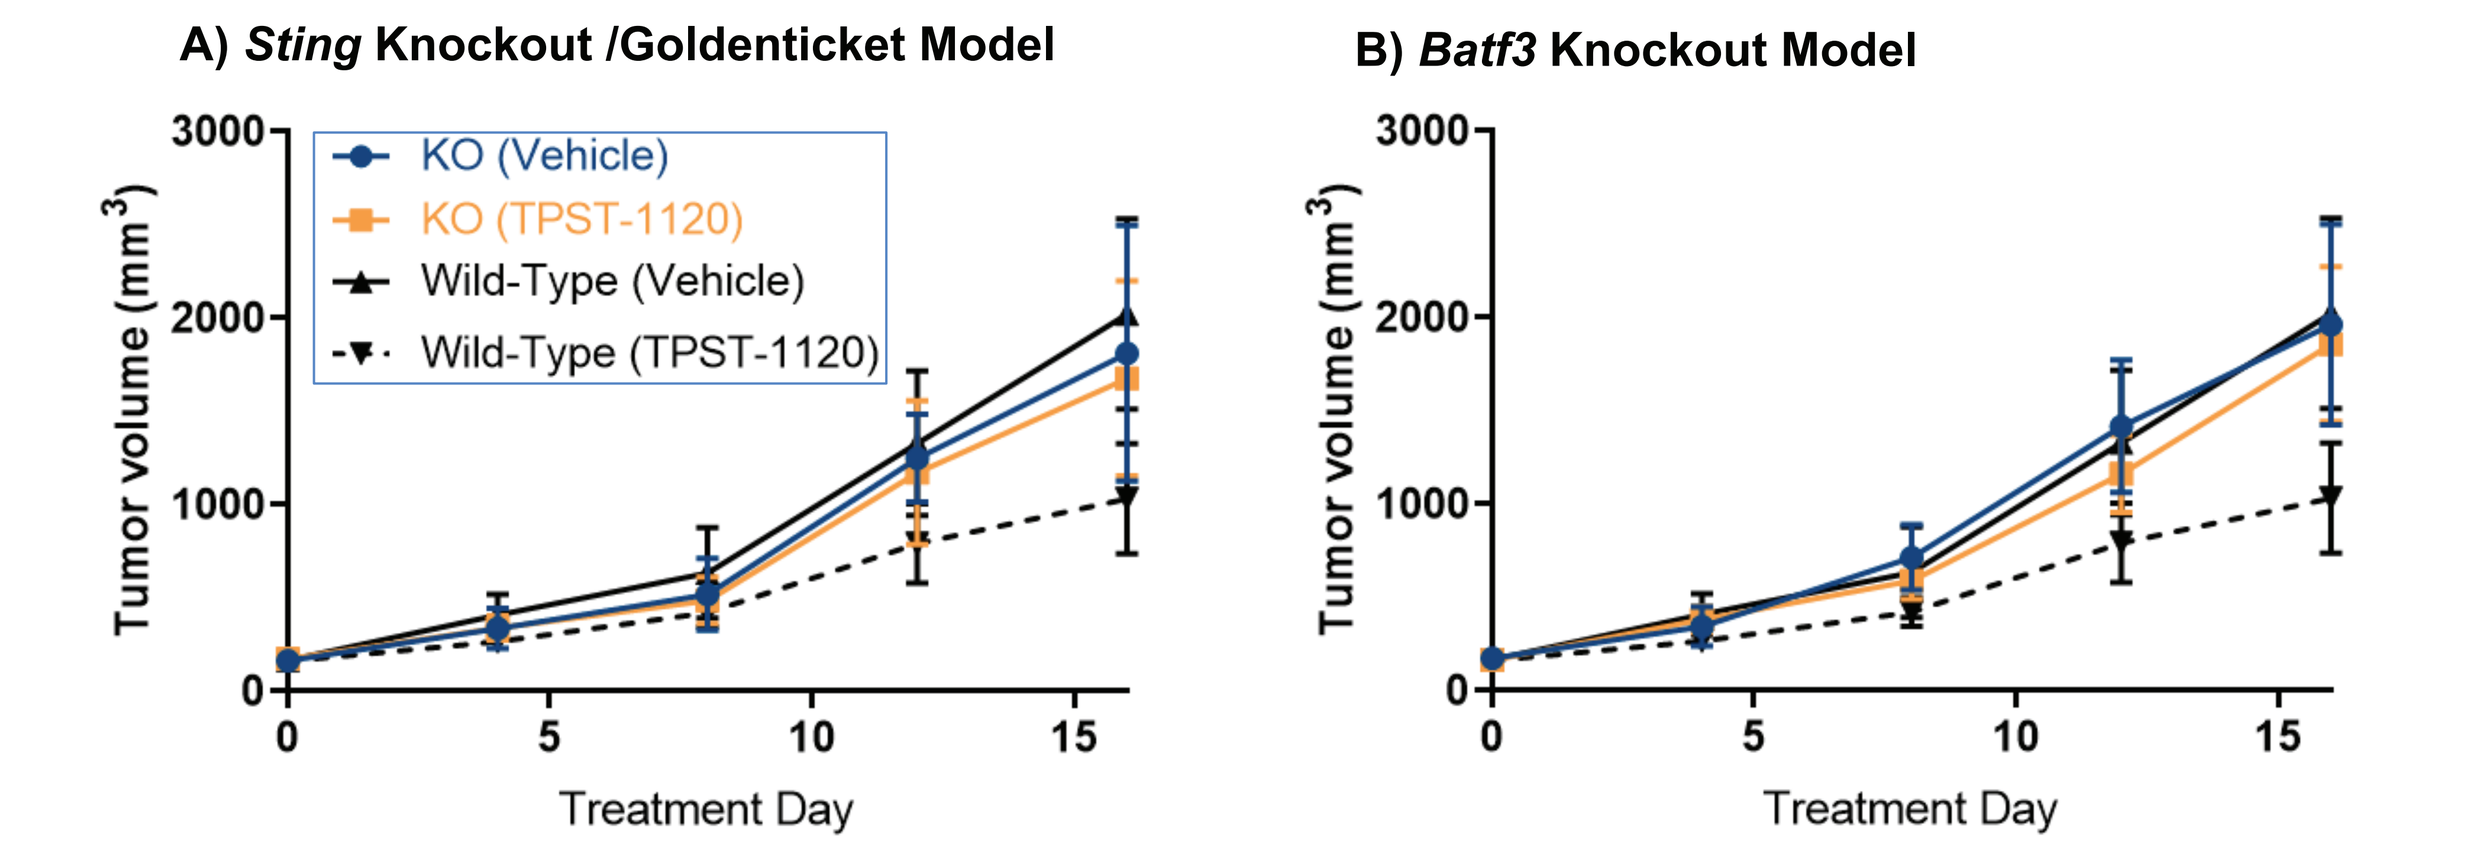
A) *Sting* Knockout /Goldenticket Model
B) *Batf3* Knockout Model
KO (Vehicle)
KO (TPST-1120)
Wild-Type (Vehicle)
Wild-Type (TPST-1120)
Tumor volume (mm3)
Treatment Day

Figure 8: A) TPST-1120 at 30 mg/kg PO BID had no effect in *Sting* or B) *Batf3* knockout mice (n=10) compared to wild type control (n=5).

FIGURE 9: TPST-1120 Anti-Tumor Efficacy is Dependent on TSP-1

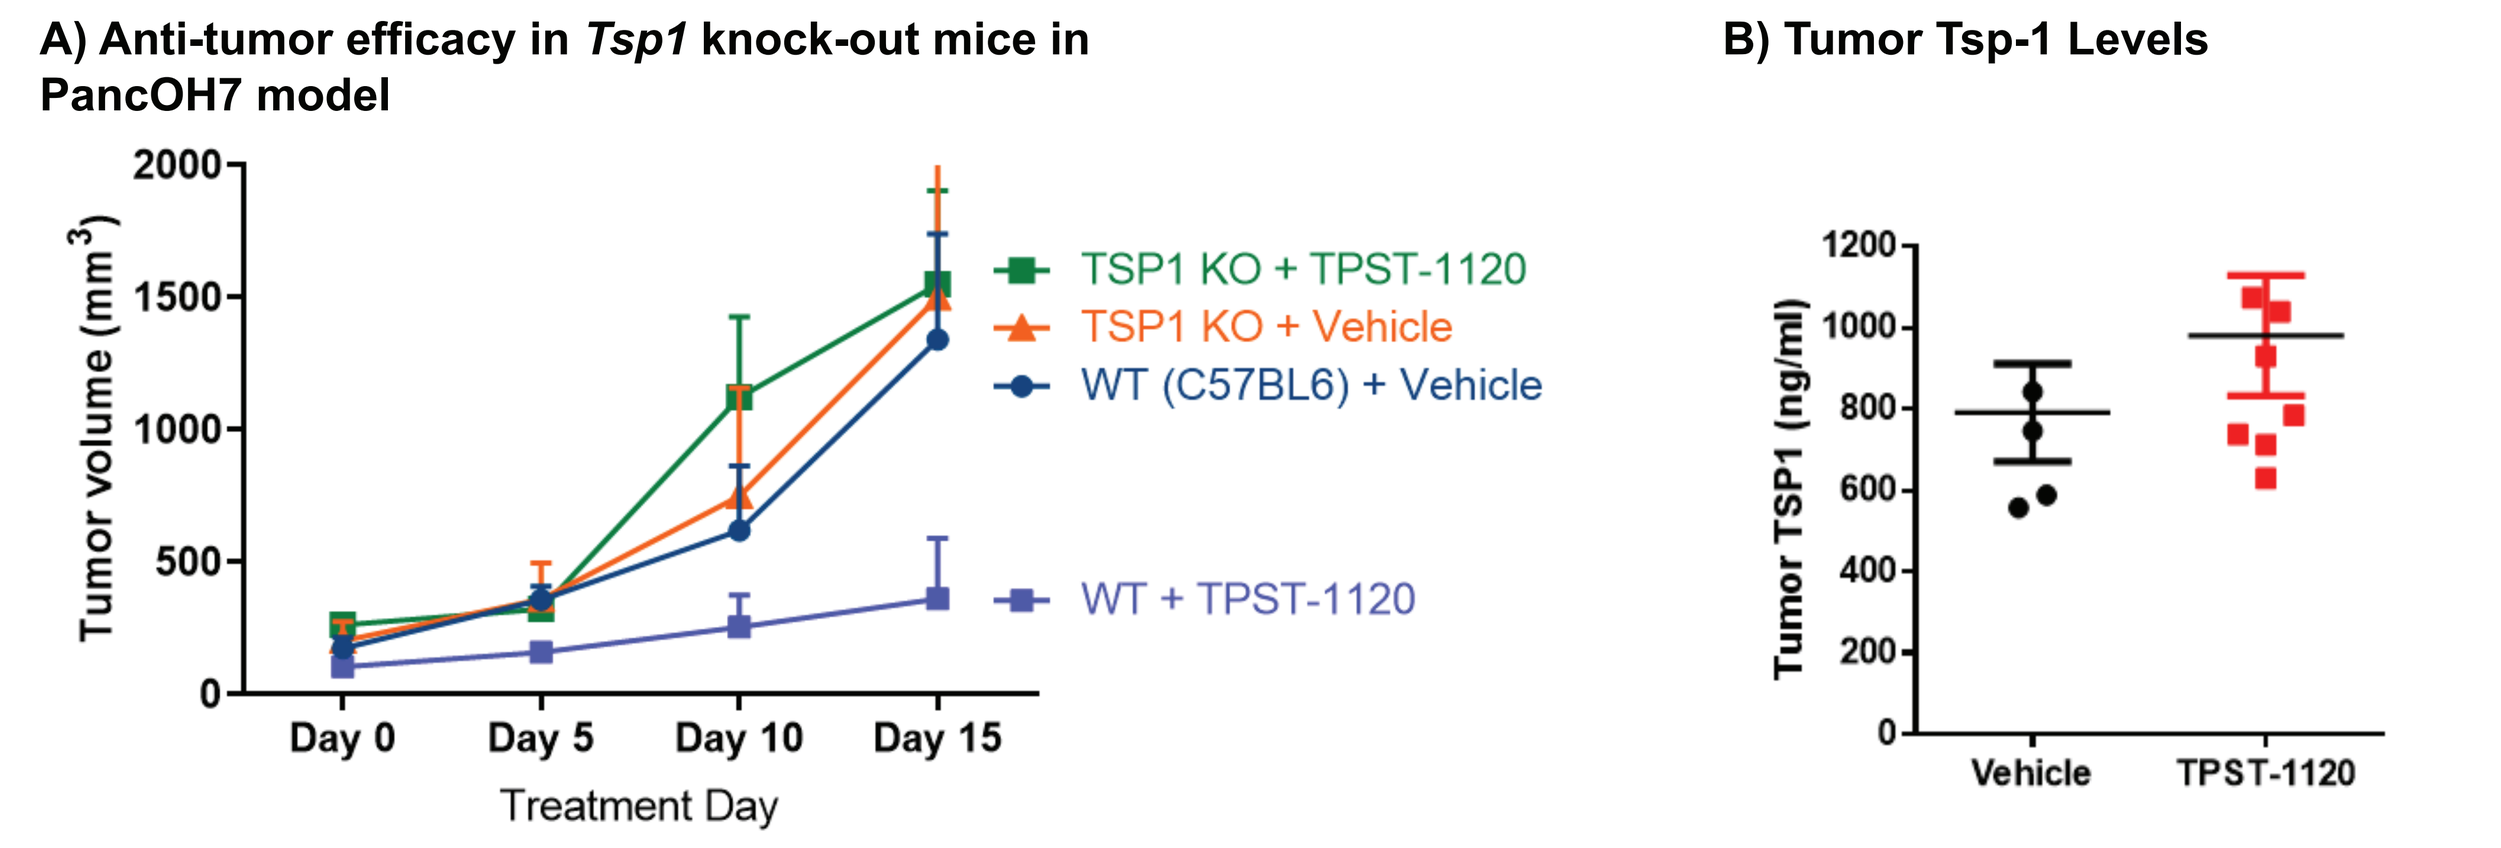
A) Anti-tumor efficacy in *Tsp1* knock-out mice in PancOH7 model
TSP1 KO + TPST-1120
TSP1 KO + Vehicle
WT (C57BL6) + Vehicle
WT + TPST-1120
Tumor volume (mm3)
Day 0
Day 5
Day 10
Day 15
Treatment Day
B) Tumor Tsp-1 Levels
Tumor TSP1 (ng/ml)
Vehicle
TPST-1120

Figure 9: A) TPST-1120 has potent anti-tumor monotherapy activity in WT mice but efficacy was lost when PancOH7 tumors were implanted in Tsp-1 knock-out mice. B) Anti-tumor efficacy is associated with increased Tsp-1 levels in the TME

FIGURE 10: TPST-1120 Combination with Anti-PD1 Induces Protective Immunity

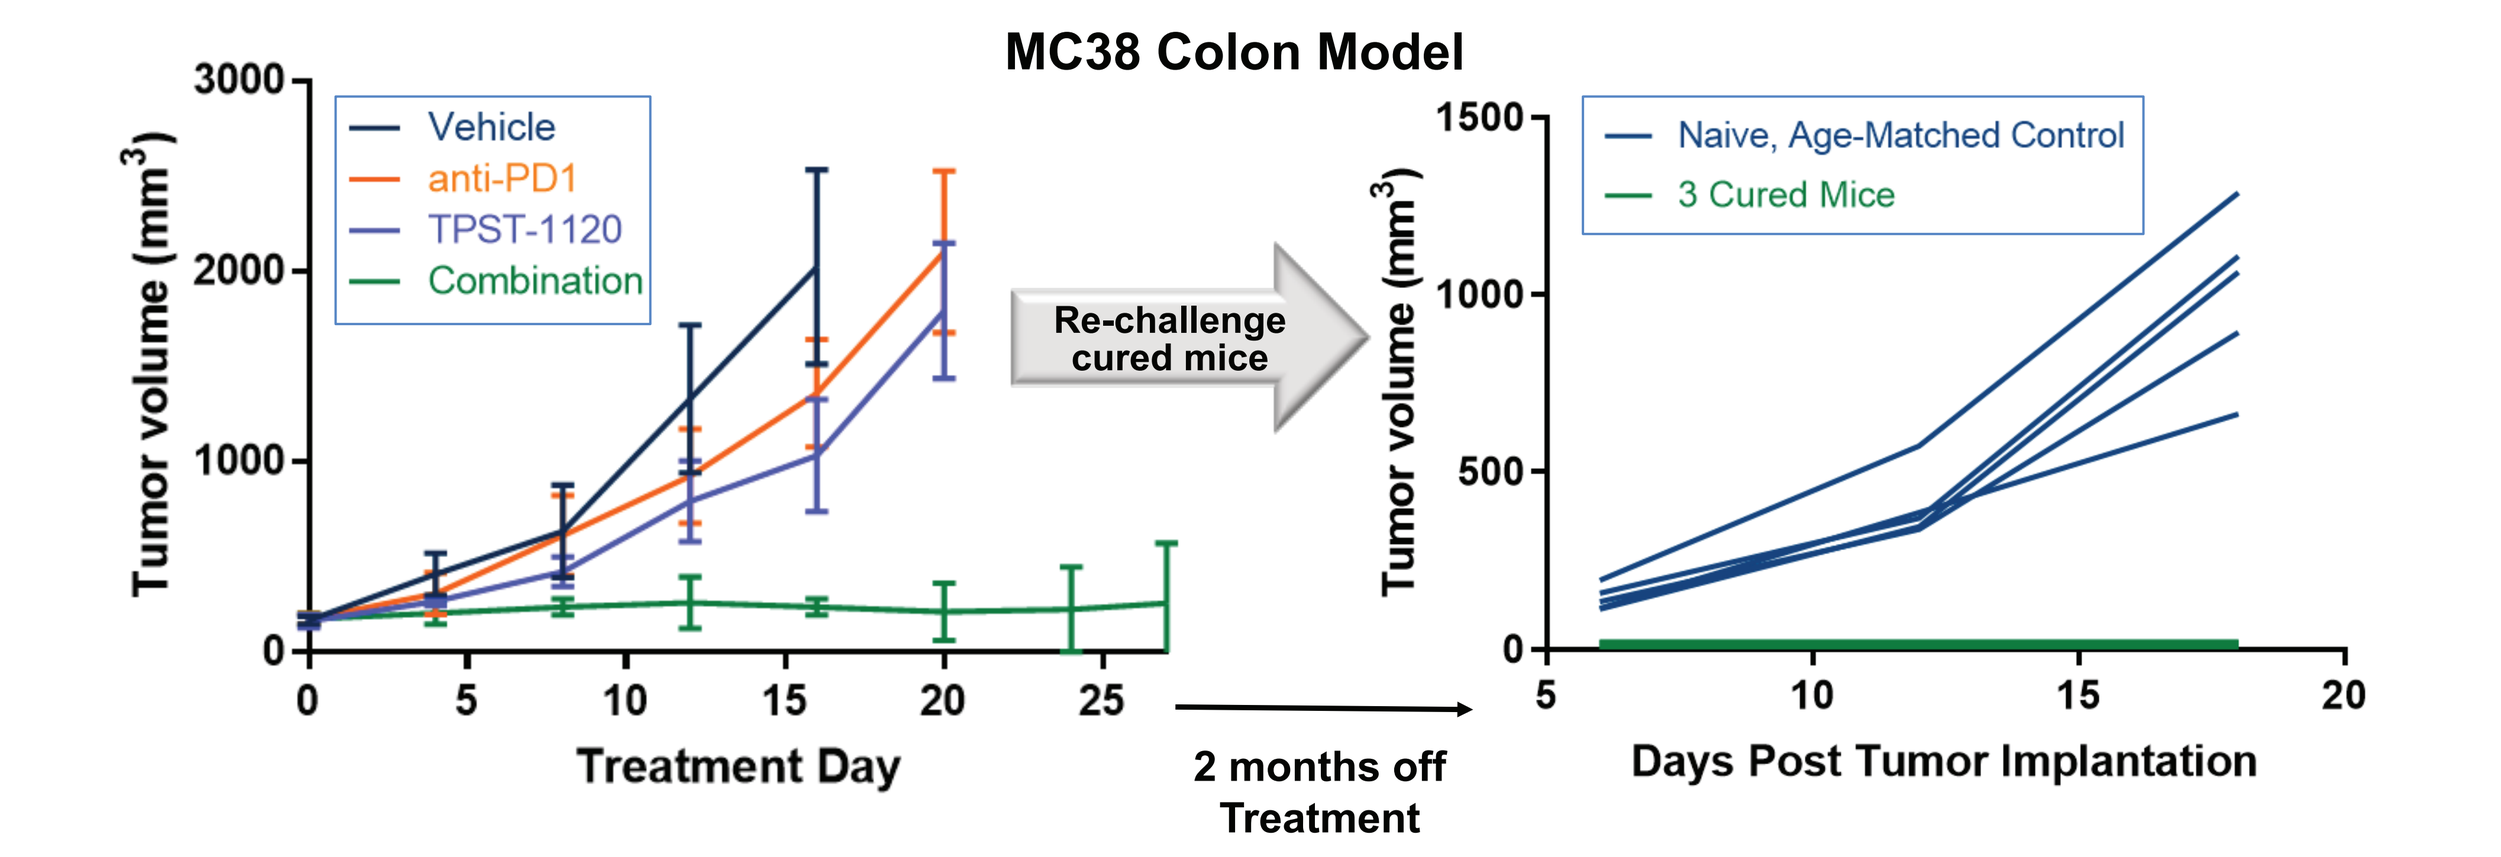
MC38 Colon Model
Vehicle
anti-PD1
TPST-1120
Combination
Tumor volume (mm3)
Treatment Day
Re-challenge cured mice
2 months off Treatment
Naive, Age-Matched Control
3 Cured Mice
Days Post Tumor Implantation

Figure 10: Established MC38 tumors (100-150mm^3) treated with vehicle (n=10), TPST-1120 (n=7), anti-PD1 alone (n=10), or TPST-1120 + anti-PD1 (n=15) were monitored for tumor growth. By day 24 only animals in the combo group remained alive. Three were tumor free at day 60 post removal of therapy and were tumor-free upon autologous tumor re-challenge while aggressive tumor growth occurred in naïve mice.

TPST-1120 CONCLUSIONS

- A first-in-class PPARα antagonist provides anti-tumor efficacy as a monotherapy or in combination with chemotherapy or anti-PD1
- Directly inhibits tumor proliferation
- Requires both innate and adaptive immunity for anti-tumor efficacy
- Anti-tumor efficacy is TSP-1 dependent
- A Phase 1/1b open-label, dose-escalation and dose-expansion study of TPST-1120 as a single agent or in combination with systemic anti-cancer therapies in subjects with advanced solid tumors is planned to initiate in early 2019

REFERENCES

1. Kaipainen A, Kieran MW, Huang S, Butterfield C, Bielenberg D, et al. (2007) PLOS ONE 2(2): e260
2. Luo X, Cheng C, Tan Z, Li N, Tang M, et al. (2017) Mol Cancer, 16(1): 76
3. Carracedo A, Cantley LC, Pandolfi PP. (2013) Nat Rev Cancer 13(4): 227-32
4. Biswas SK. (2015) Immunity, 43(3): 435-449
5. Camarda R, Zhou AY, Kohnz, RA, Balakrishnan, S, Mahieu, C, et al. (2016) Nat Med, 22(4): 427-432
6. Nath A, & Chan C. (2016) Sci Rep, 6: 1-13
7. Senni N, Savall M, Cabrerizo Granados D, Alves-Guerra MC, Sartor C, et al.(2018). *Gut, 0: 1-3*